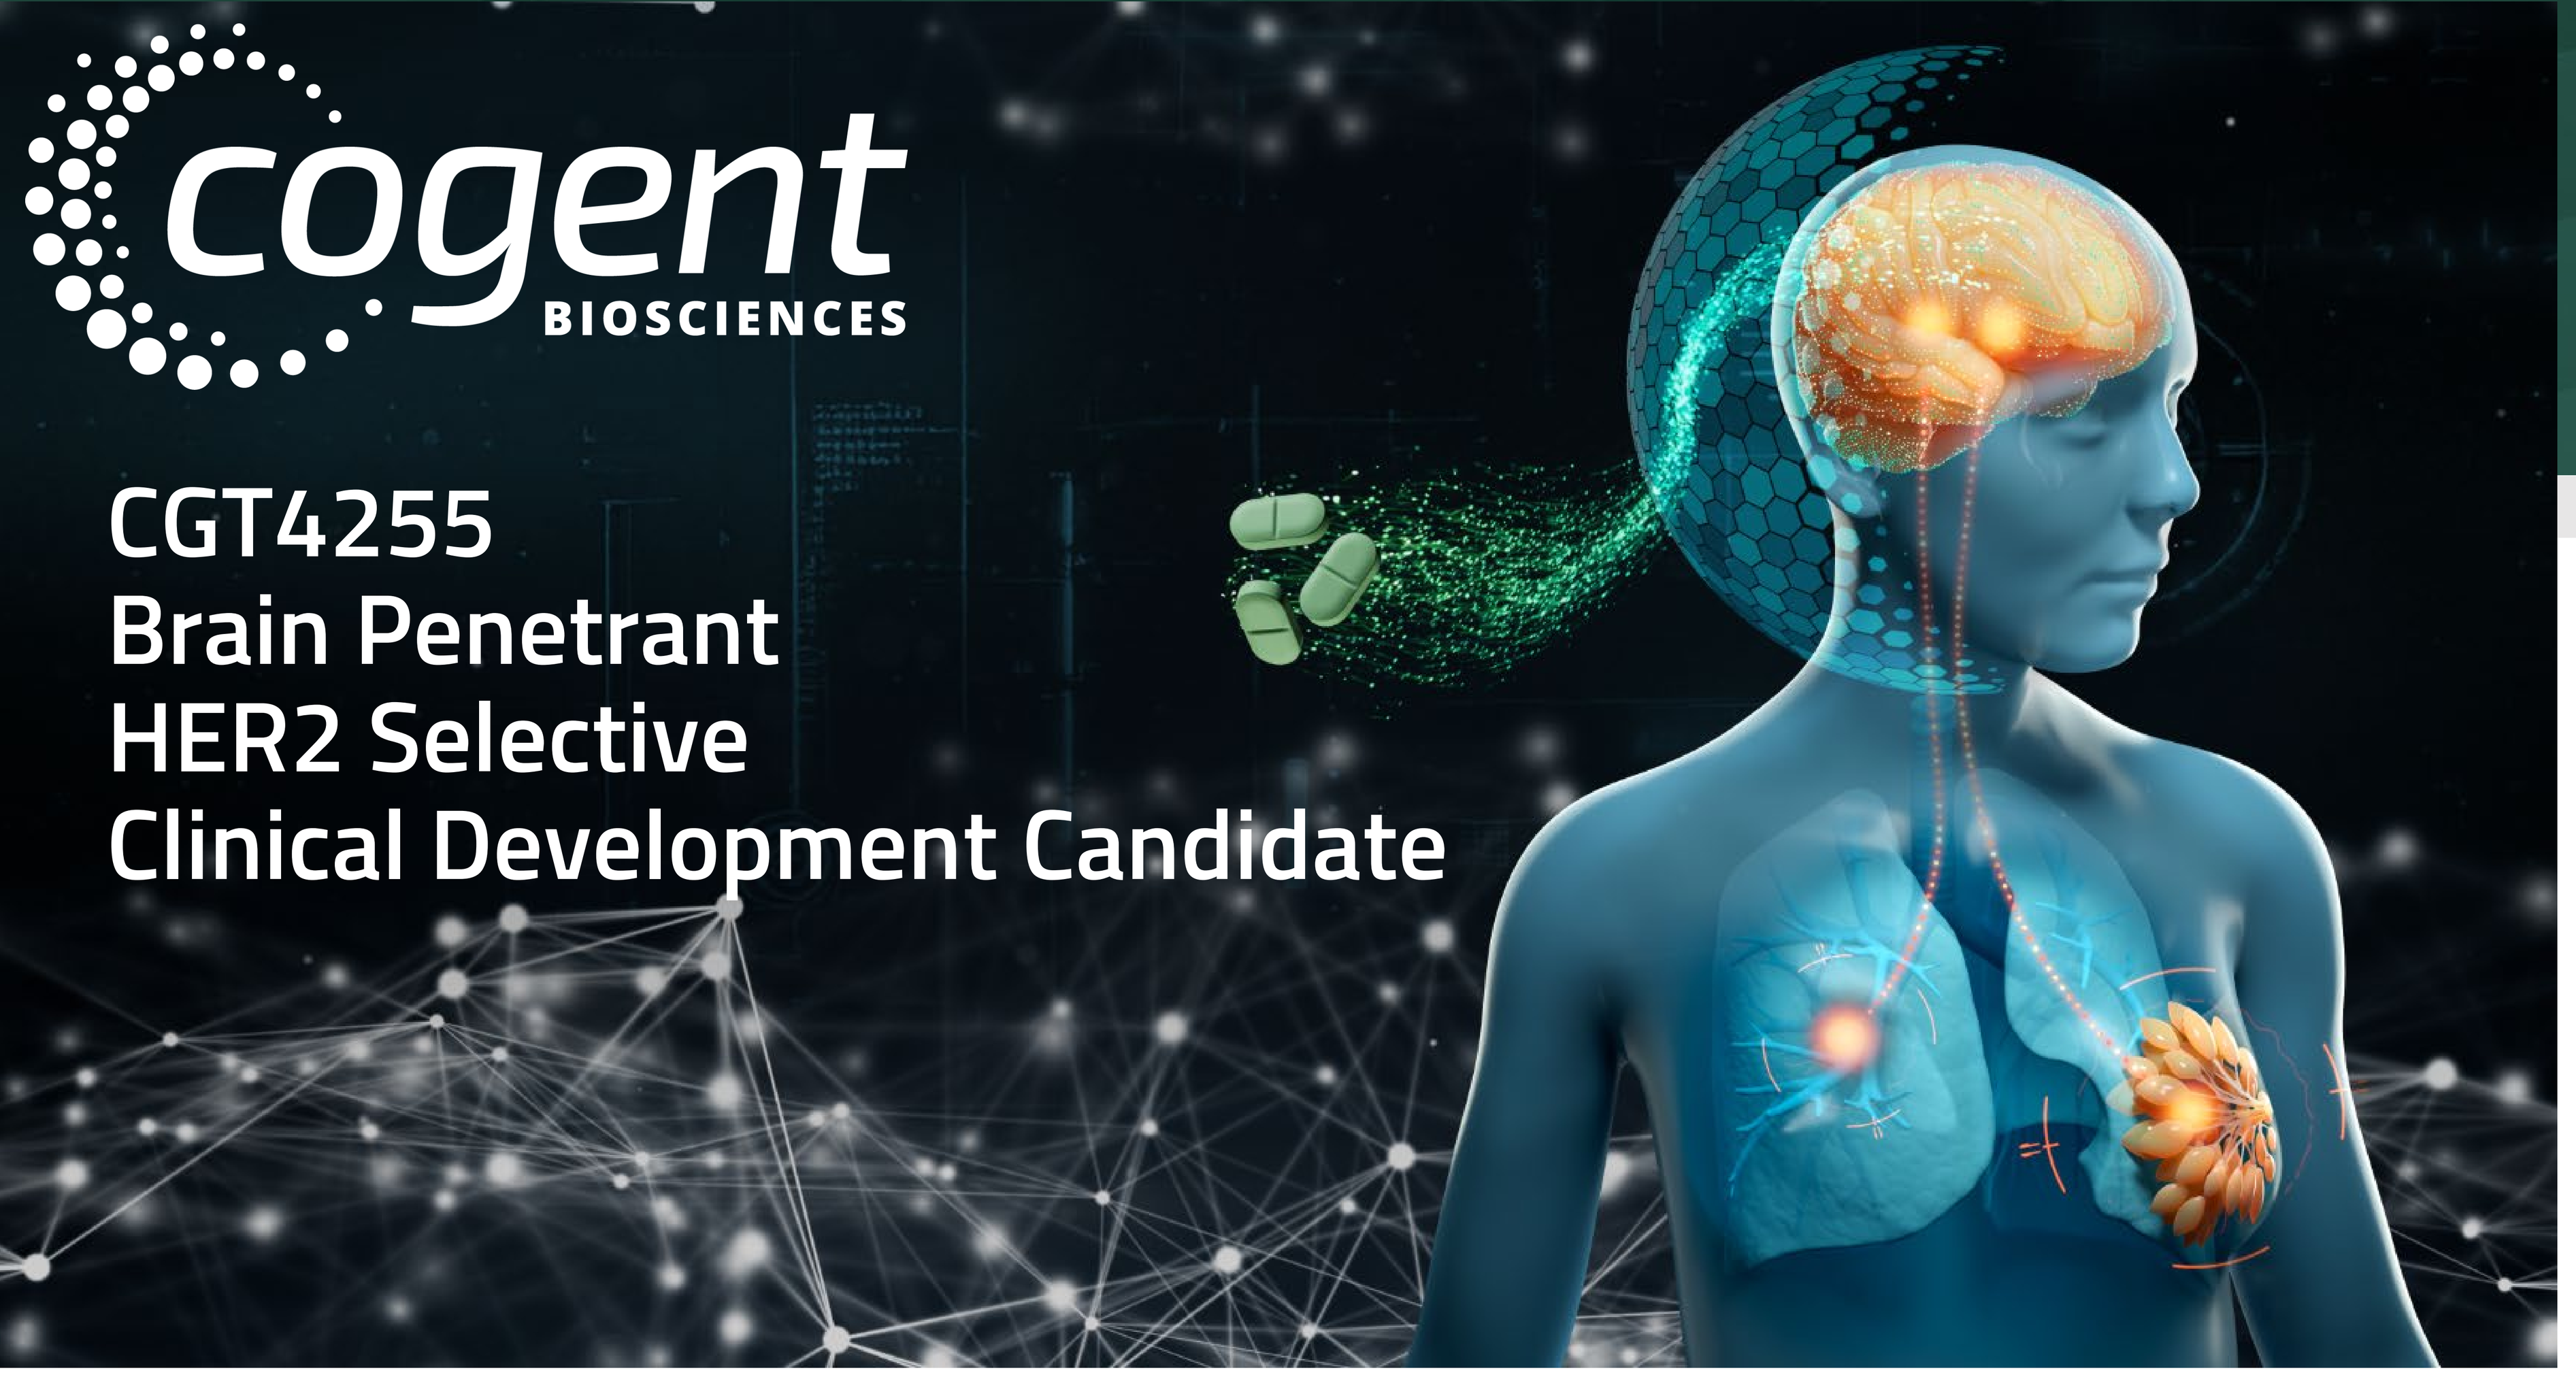
cogent
BIOSCIENCES
CGT4255
Brain Penetrant
HER2 Selective
Clinical Development Candidate

CGT4255 is a Brain Penetrant, HER2 Selective Clinical Development Candidate With Activity in CNS and Subcutaneous Tumors

Mark J Chicarelli, Vijay Kumar, Tanna Bettendorf, Abiezer Blandon, Karyn Bouhana, Richard K. Brizendine, LouAnn Cable, Michelle Crow, Brad Fell, John Fischer, Jennifer Fulton, Anna Guarnieri, Madison Hillman, Ravi Jalluri, Hailey Knox, Keith A. Koch, Cori A. Malinky, Rob Rieger, John Robinson, Lee M Stunkard, Francis Sullivan, John I Trujillo, Logan E Vine, Shannon Winski, and Yeyun Zhou; Cogent Biosciences, Inc., Boulder, CO and Waltham, MA

Presentation Number: PO3-26-02

Selective Across the Kinome and 100X Selective Over EGFR

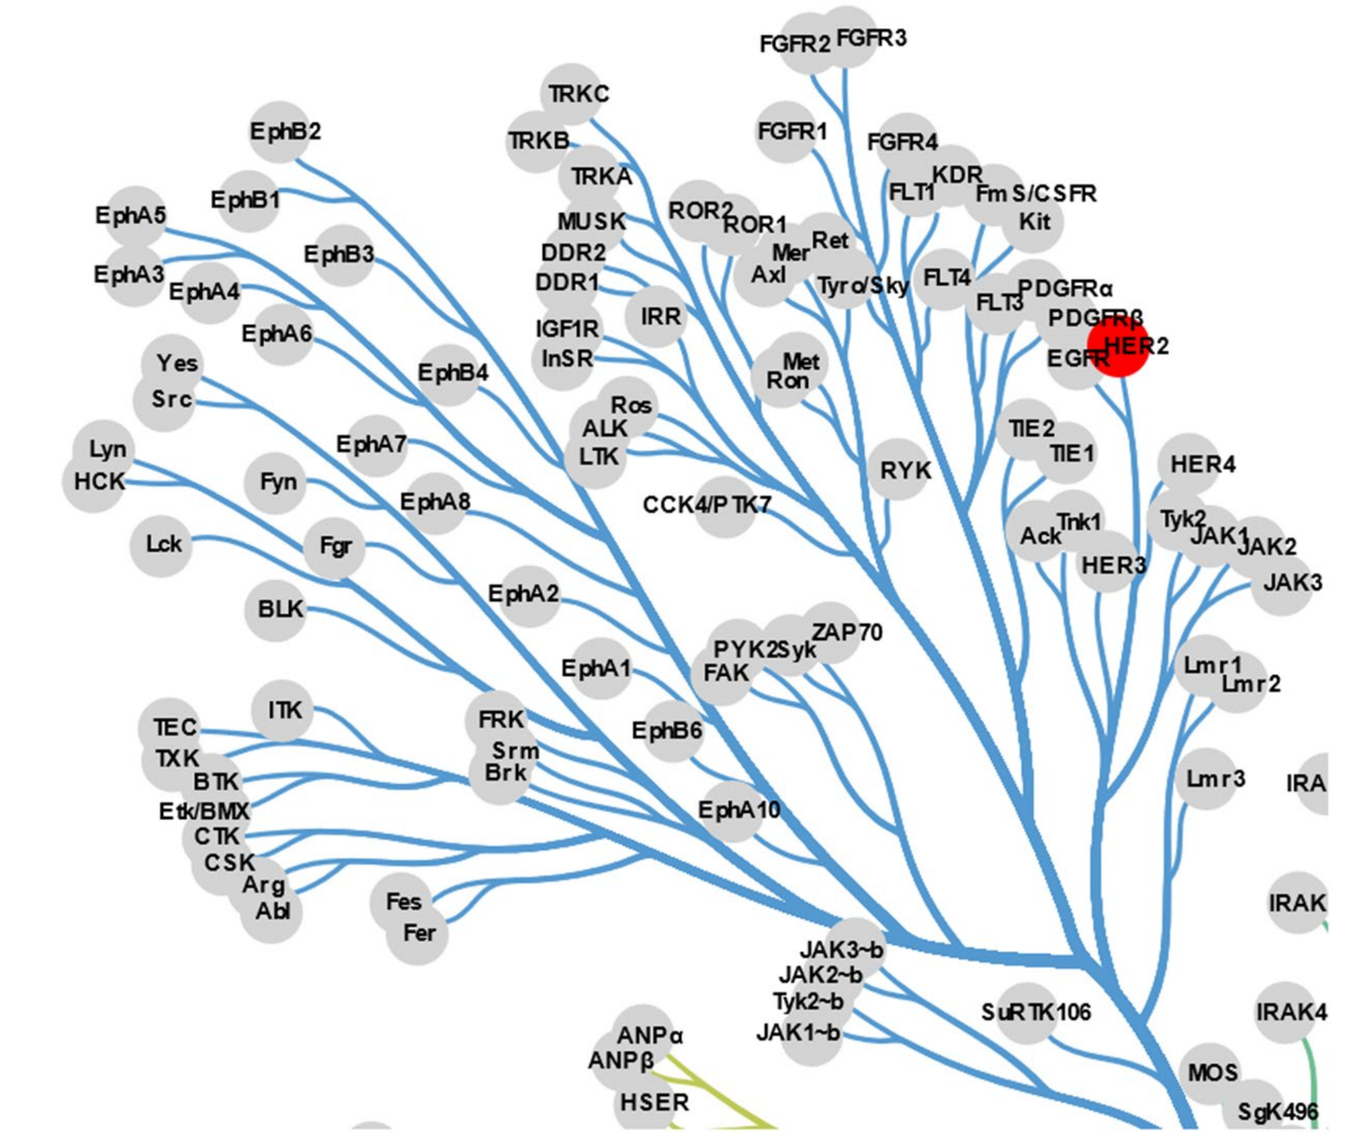

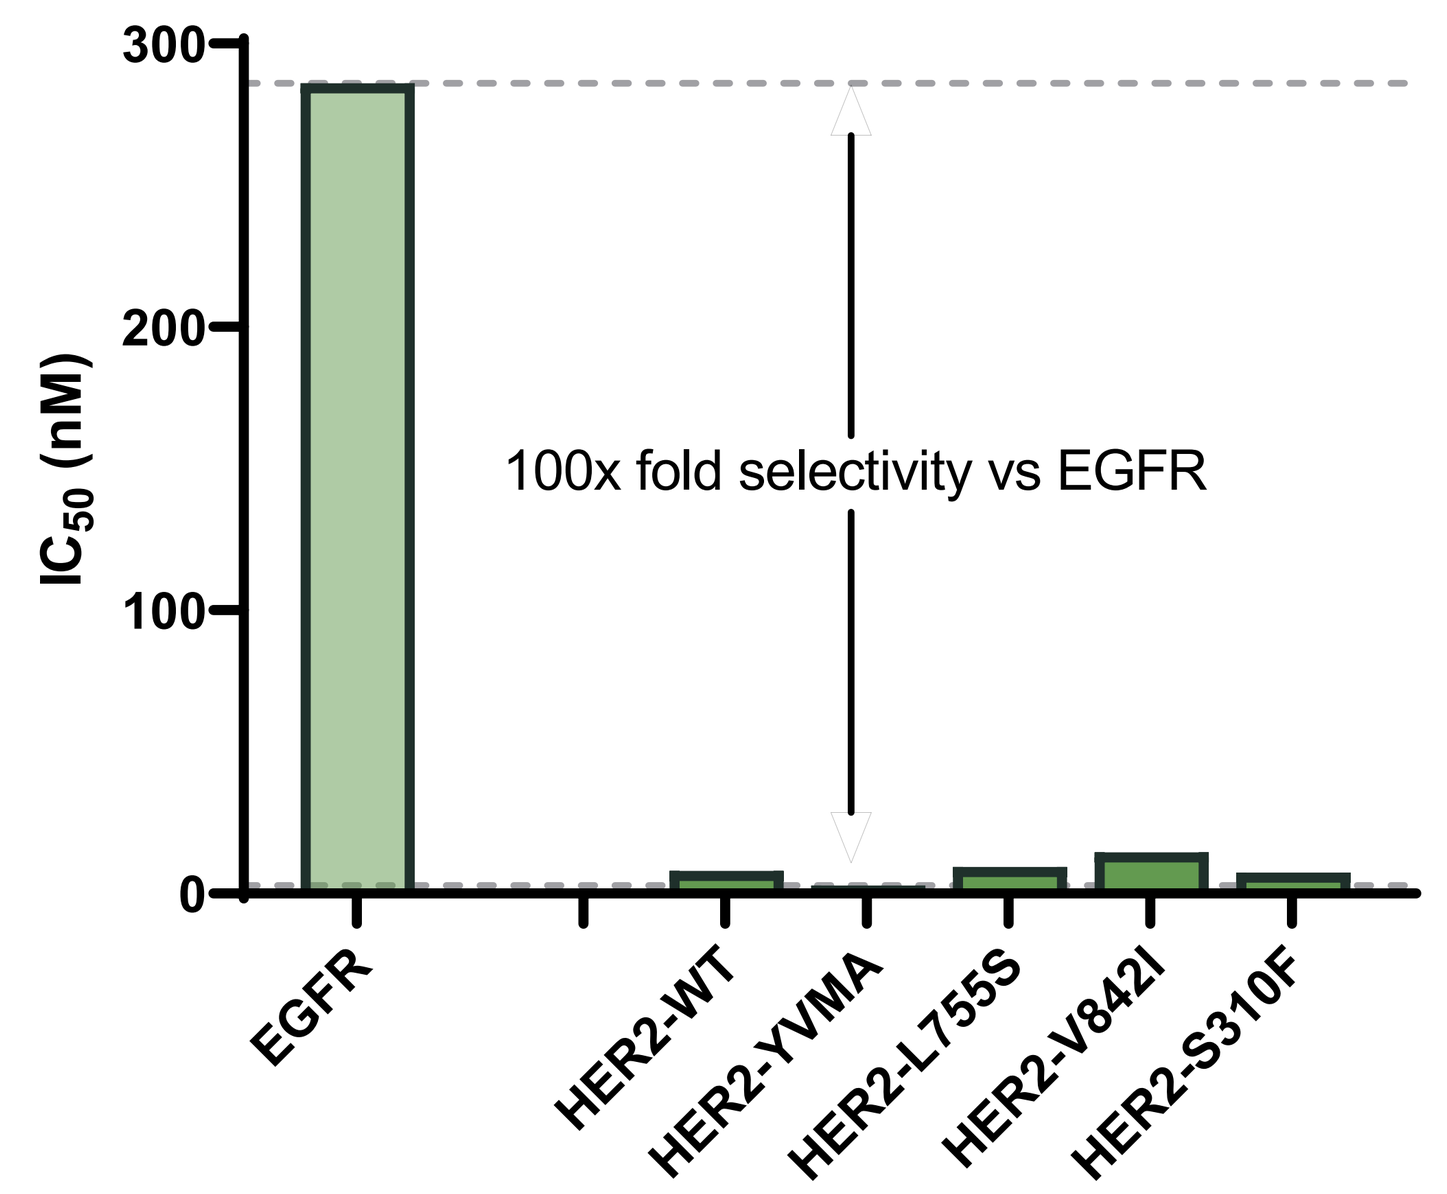
300
200
100
0
IC_{50} (nM)
100x fold selectivity vs EGFR
EGFR
HER2-WT
HER2-YVMA
HER2-L755S
HER2-V842I
HER2-S310F

- CGT4255 was profiled at 10x the enzyme IC_{50} for WT HER2 against a panel of 396 kinases, HER2 was the only kinase that showed >30% target inhibition
- Mechanistic cellular assays show CGT4255 is 100-fold selective for HER2 YVMA over WT-EGFR
- CGT4255 covers all HER2 mutations tested, including L755S, a prevalent activating mutation in breast cancer[1]

Regressions Observed in HER2+ and Mutant HER2-L755S Models

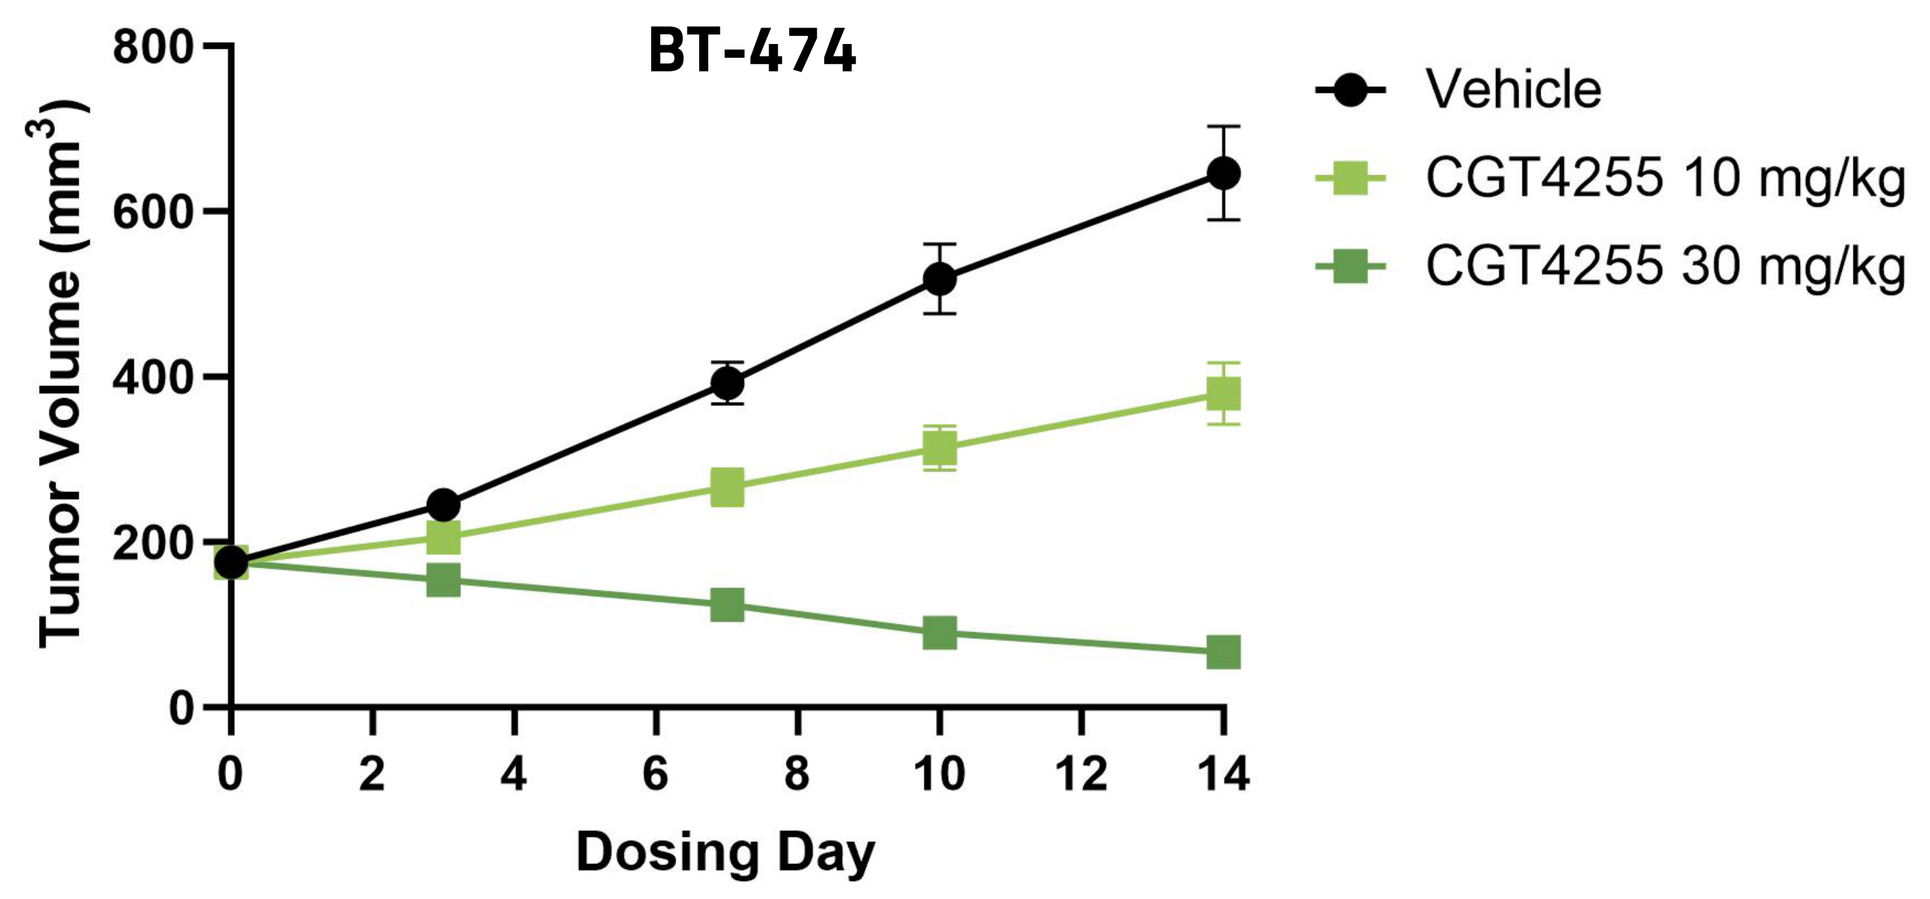
BT-474
Tumor Volume (mm^3)
Dosing Day
Vehicle
CGT4255 10 mg/kg
CGT4255 30 mg/kg

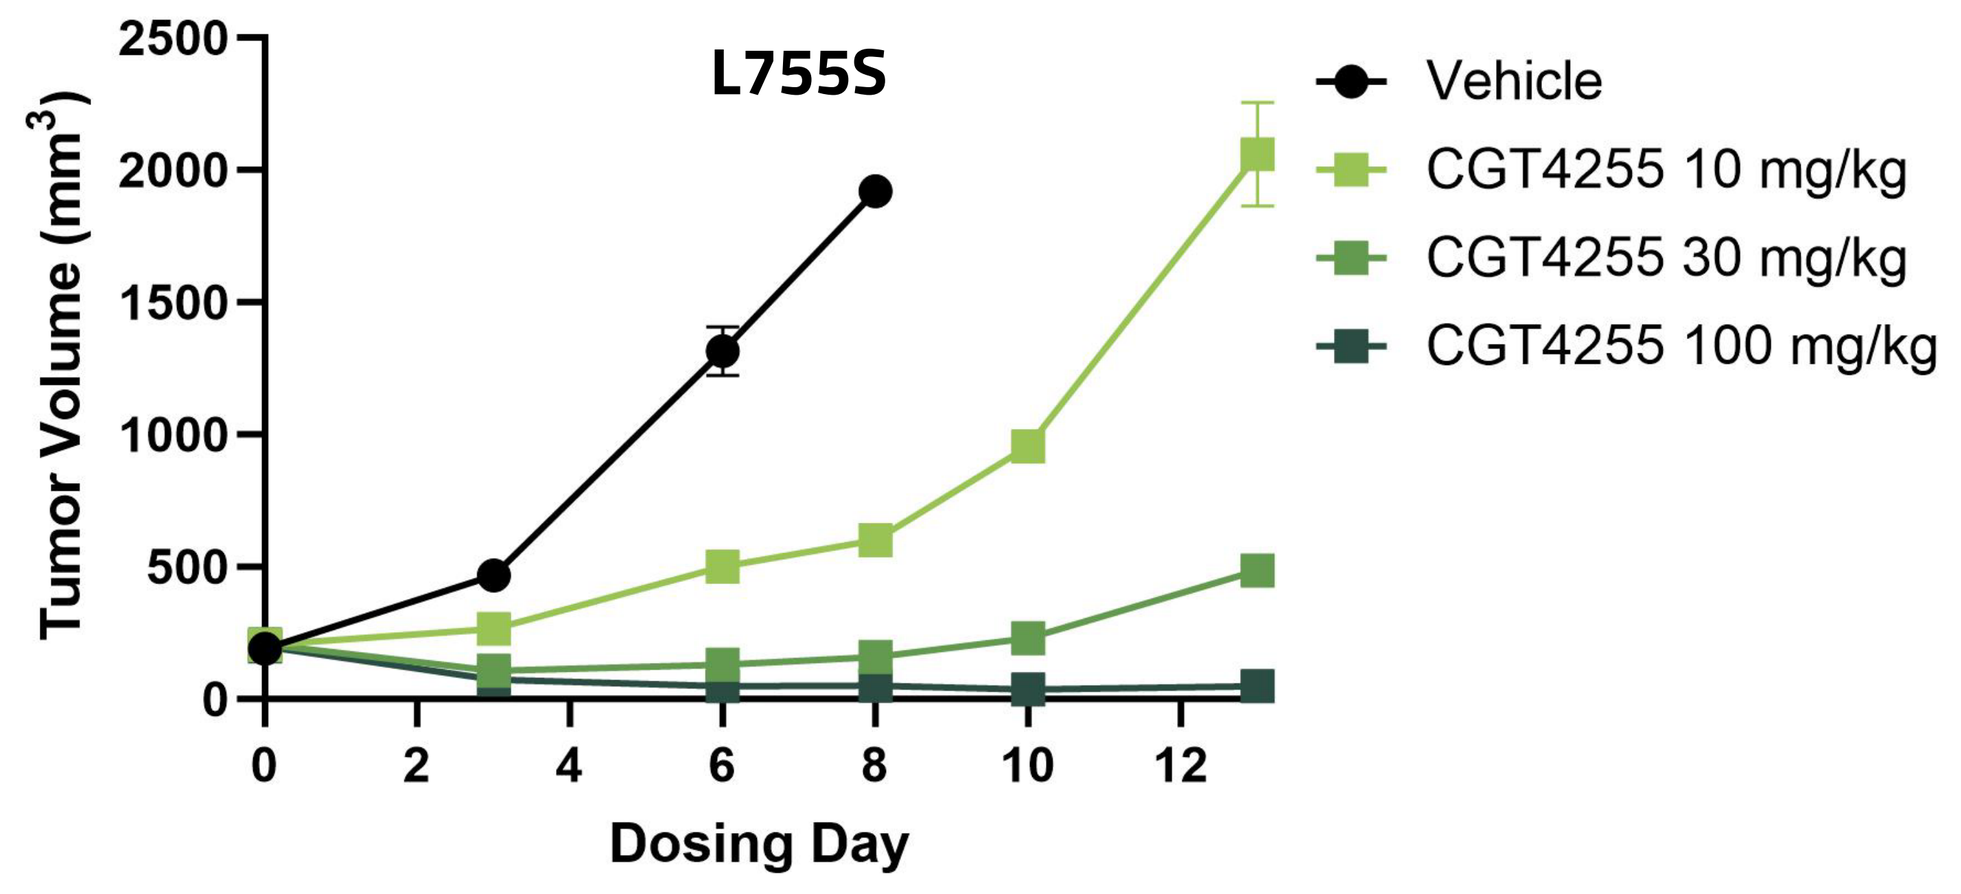
L755S
Tumor Volume (mm^3)
Dosing Day
Vehicle
CGT4255 10 mg/kg
CGT4255 30 mg/kg
CGT4255 100 mg/kg

- CGT4255 was orally administered at 10 or 30 mg/kg QD in a subcutaneous BT-474 TGI model
- CGT4255 demonstrated dose responsive TGI with tumor regression observed at 30 mg/kg

- CGT4255 was orally administered at 10, 30 or 100 mg/kg BID in a NIH3T3-Her2-L755S TGI model
- CGT4255 demonstrated dose responsive TGI with complete responses observed at 100 mg/kg

Best-in-Class Fully Brain Penetrant HER2 Inhibitor

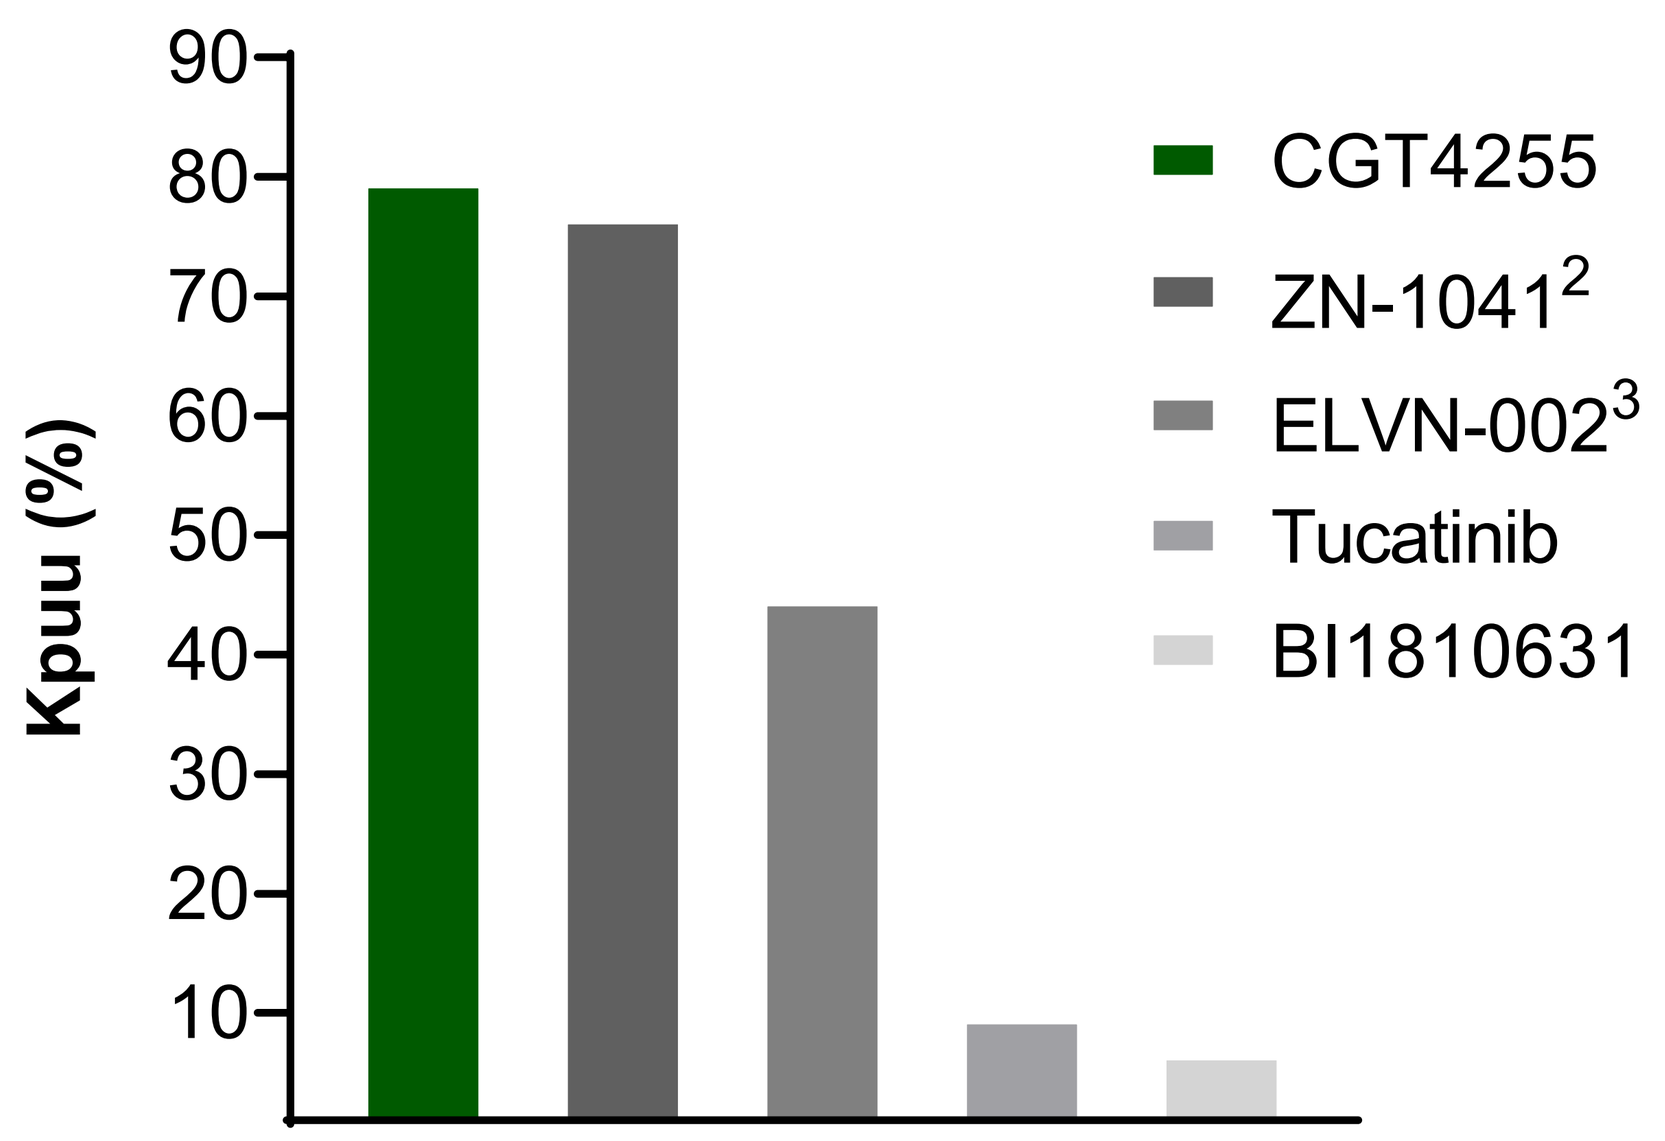
Kpuu (%)
CGT4255
ZN-1041[2]
ELVN-002[3]
Tucatinib
BI1810631

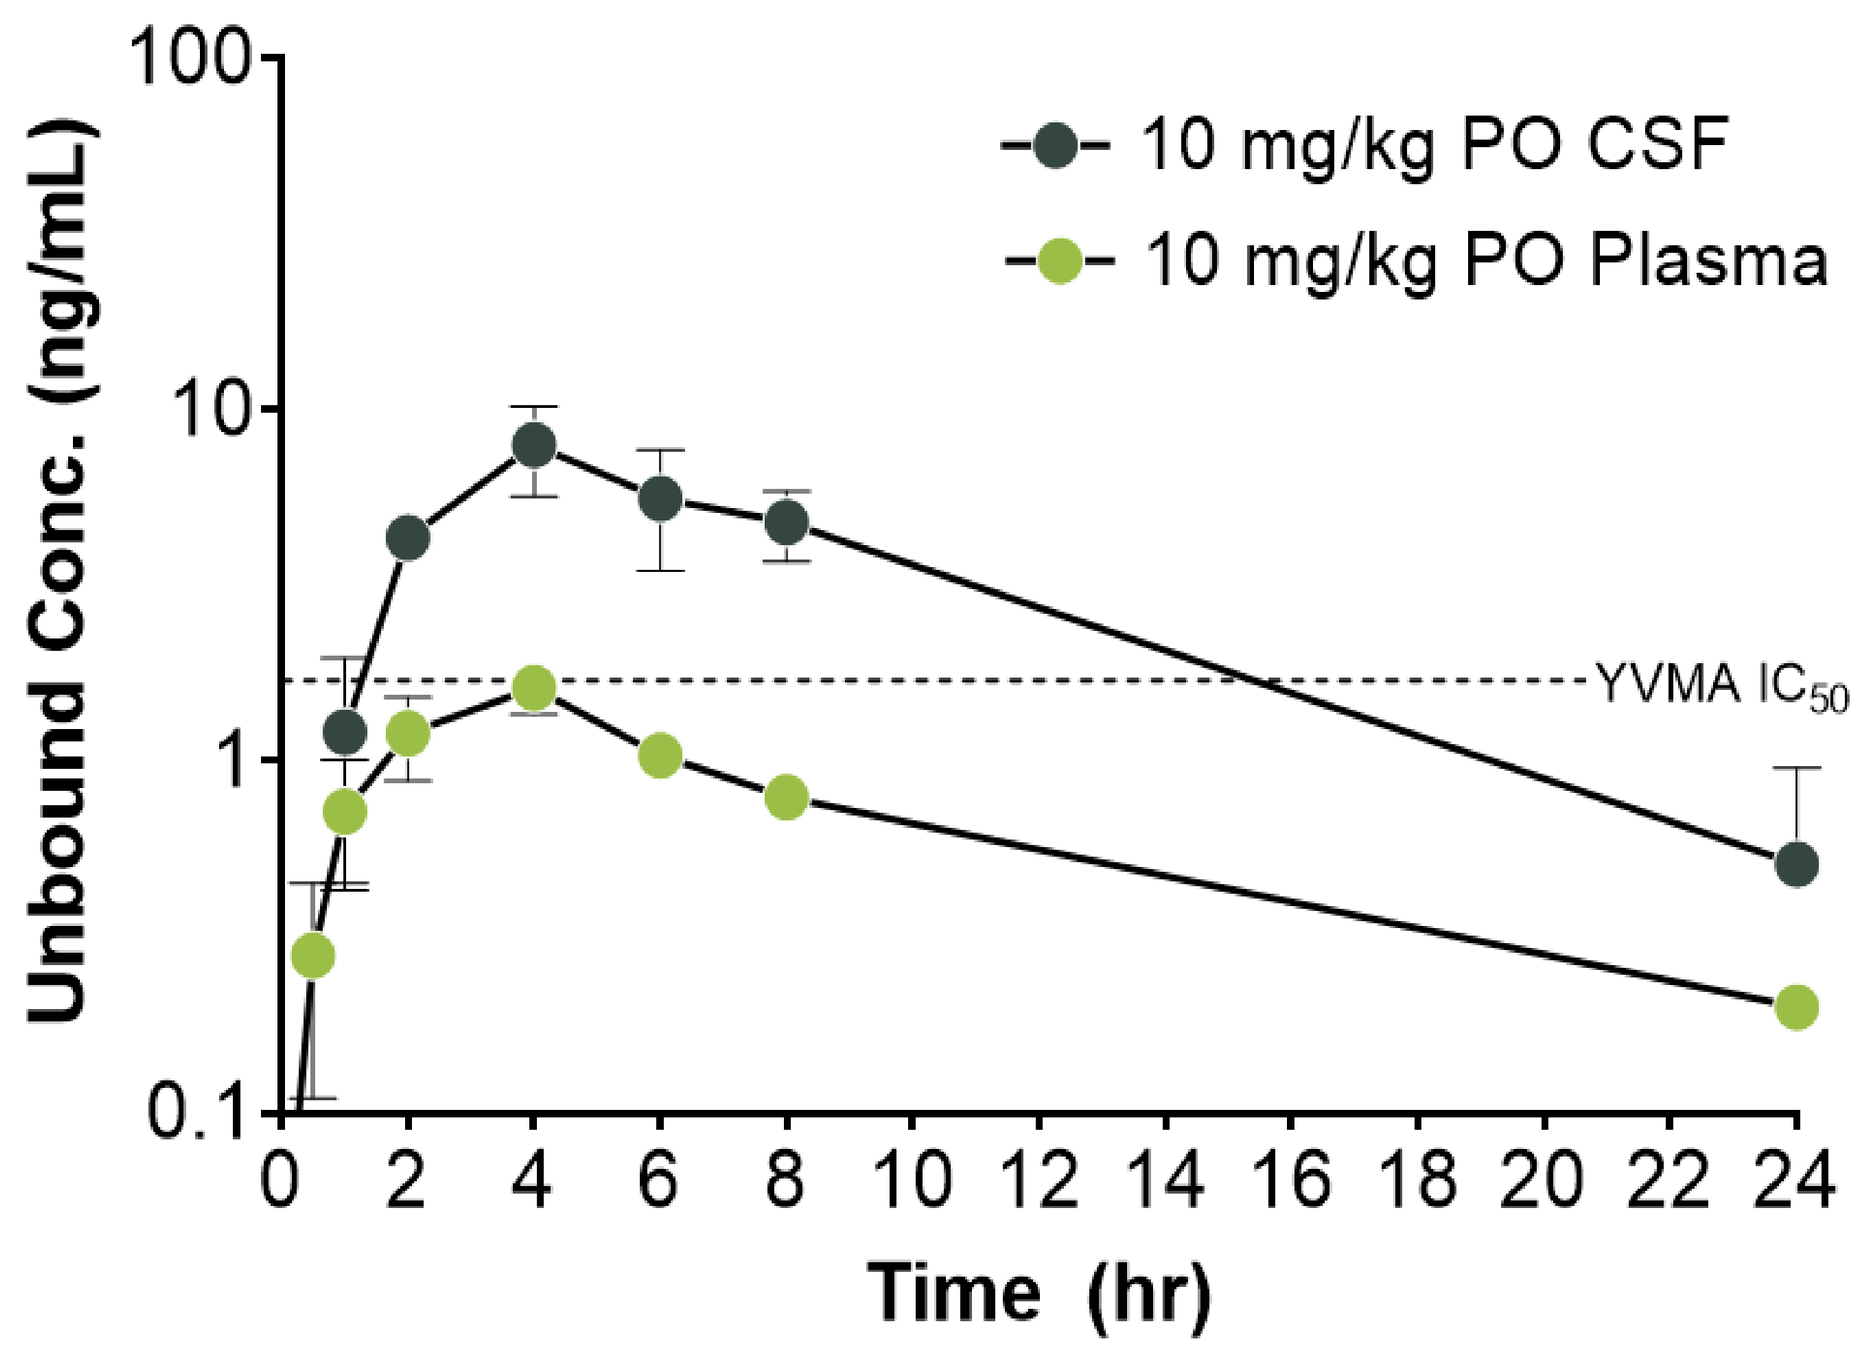
Unbound Conc. (ng/mL)
10 mg/kg PO CSF
10 mg/kg PO Plasma
YVMA IC_{50}
Time (hr)

- CGT4255 has a high unbound brain to plasma partition coefficient (Kpuu) and is considered fully brain penetrant[4]
- Equivalent to reported Kpuu of Zion/Roche ZN-1041
- Two-fold higher Kpuu than ELVN-002 reported values
- Eight to ten times higher brain to plasma ratio compared to tucatinib and BI1810631

- CGT4255 was orally administered to cynomolgus monkeys at 10 mg/kg and drug concentrations were measured in both plasma and cerebral spinal fluid (CSF)
- CSF/Plasma ratio of 4.8 confirms full brain penetration; literature precedence of similar data correlates with high brain levels in human[5]

Added Efficacy in Combination with T-DXd in an Intracranial Model

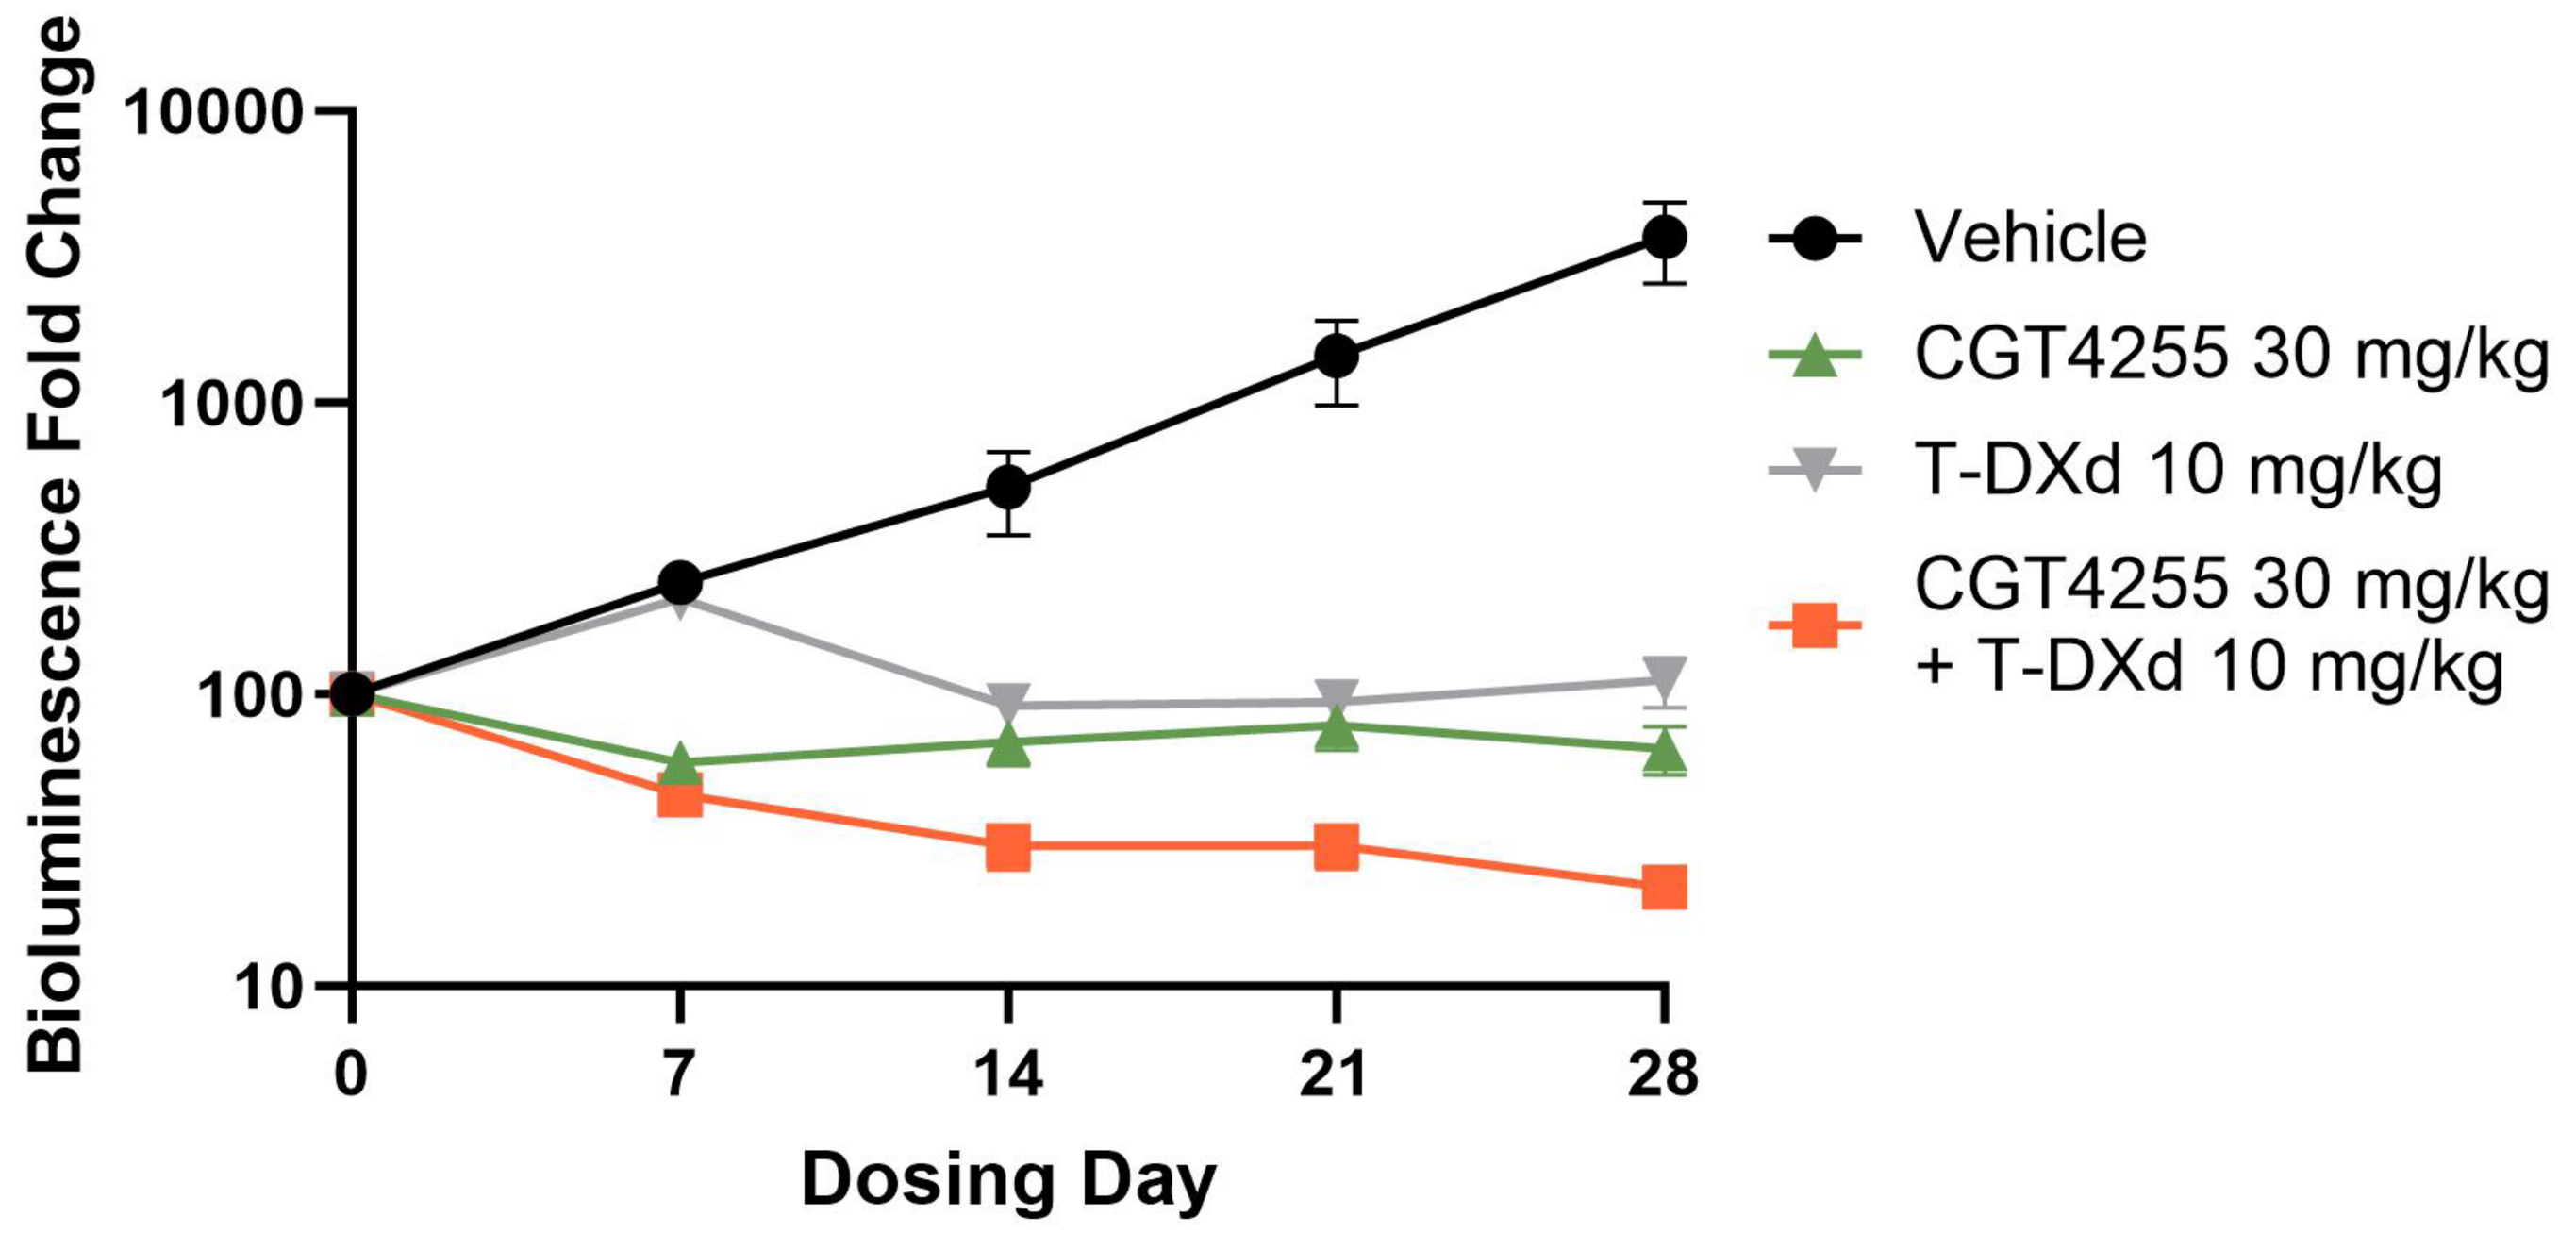
Bioluminescence Fold Change
Dosing Day
Vehicle
CGT4255 30 mg/kg
T-DXd 10 mg/kg
CGT4255 30 mg/kg
+ T-DXd 10 mg/kg

- CGT4255 was orally administered at 30 mg/kg QD in an NCI-N87-luc intracranial model as a single agent and in combination with T-DXd at 10 mg/kg IV, Q3W
- CGT4255 and T-DXd demonstrate stable disease as monotherapies
- Added efficacy was observed in combination with T-DXd with decreased luminescence indicative of tumor regression
- All regimens were well tolerated with no body weight decreases or other safety signals observed

CGT4255 Profile

- **Fully brain penetrant in mouse and monkey; high brain concentrations predicted for human**
- **Added efficacy in an intracranial model in combination with T-DXd**
- **Regressions in HER2-overexpressed subcutaneous mouse tumor model**
- **Complete regressions in mutant HER2 L755S subcutaneous mouse TGI**
- **Well-tolerated at efficacious doses in TGI models**
- **Low DDI risk based on in vitro safety panel**
- **Low nM potency across HER2 mutations**
- **>100-fold selectivity over EGFR**

CGT4255 is currently in IND-enabling studies with clinical trials planned in 2025

In Vitro Safety Assessment Predicts Low DDI Risk

Assay	CGT4255
Kinome Screen	Clean
Safety Receptor Panel	Clean
Ion Channel Panel	Clean
P-gp Efflux Transporter Substrate	No
BCRP Efflux Transporter Substrate	No
CYP Inhibition	Favorable CYP inhibition profile
CYP450 TDI	Clean
CYP Induction	3A4 – Moderate Risk
Transporter Inhibition	Low Risk
Non GLP AMES	Negative
hERG	No Risk

- High kinome selectivity for HER2, no inhibition >30% of 396 kinases tested
- Clean on safety receptor and ion channel panels
- Not a P-gp or BCRP efflux transporter substrate, efflux ratio < 1.5 for both transporters
- Favorable CYP inhibition profile, IC_{50} > 20 μM across CYPs
- No CYP450 time-dependent inhibition risk
- Moderate risk for CYP 3A4 induction, no induction risk for 1A2 and 2B6
- Low risk for OATP1B1 inhibition
- > 400X safety window on hERG

San Antonio Breast Cancer Symposium®
December 10th, 2024

REFERENCES: (1) Subramanian *et al.* Oncologist (2019) 12:e1303-e1314. (2) Fei Ma *et al.* ASCO 2023 Poster #1040. (3) Monette Aujay *et al.* AACR 2023 Poster # 4019. (4) Irena Loryan *et al.* Pharmaceutical Research (2022) 39:1321-1341. (5) (a) J Med Chem (2013), 56, 2-12. (b) The AAPS Journal (2021) 23: 81, 1-12. (c) Zhang *et al.* Breast Cancer Research (2023), 25:81, 1-14. (d) CGT4255 is not a P-gp or BCRP efflux transporter substrate in Monkey

For questions, please contact mark.chicarelli@cogentbio.com.

DISCLOSURES: All authors are employees of Cogent Biosciences
ACKNOWLEDGEMENTS: Zhang Peichuan, Qiaoqiao Xu, Huijuan Lv, Ning Han, and the Enzymology & *In Vitro* Novel Drug Screening Team, WuXi Biology. Dr. Binglin Wang, Dr. Le Chang, Dr. Ke Zhang and their WuXi AppTec Chemistry team.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster

**This presentation is the intellectual property of Cogent Biosciences, Inc.
Contact evan.kearns@cogentbio.com for permission to reprint and/or distribute.**