Identification of CGT1263, a Potent KRAS Inhibitor With Selectivity for Mutant KRAS Over HRAS and NRAS

Leah Salituro, Keith Koch, Kim Alley, Karyn Bouhana, Richard Brizendine, Paul Carlson, Mark J Chicarelli, Michelle Crow, Brad Fell, John Fischer, Jennifer Fulton, Anna Guarnieri, Jackie Harrison, Ravi Jalluri, Amber Johnson, Cori Malinky, Matthew G. McDonald, Macedonio J. Mejia, Brad Newhouse, Scott Niman, Rob Rieger, John Robinson, Mareli Rodriguez, Vinny Scarato, Aaron Smith, Francis Sullivan, Yang Wang, Shannon Winski, Silas Wood, Yeyun Zhou

Cogent Biosciences, Inc., Boulder, CO and Waltham, MA

Poster #B024

BACKGROUND

Mutated KRAS Drives Uncontrolled Cell Proliferation, Survival, and Differentiation in Cancer

Activating mutations of KRAS occur in an estimated 175,000 new cancer cases per year in the U.S.

Currently approved agents target the KRAS G12C mutation which comprises only 14% of KRAS mutations[1,2]

Multi RAS inhibitors target KRAS, HRAS, and NRAS and can be associated with a range of toxicities, most commonly skin rash, diarrhea, nausea, vomiting, and stomatitis[3]

Highest Incidence of KRAS Mutations:[2]

Incidence of KRAS Mutations by Cancer Type

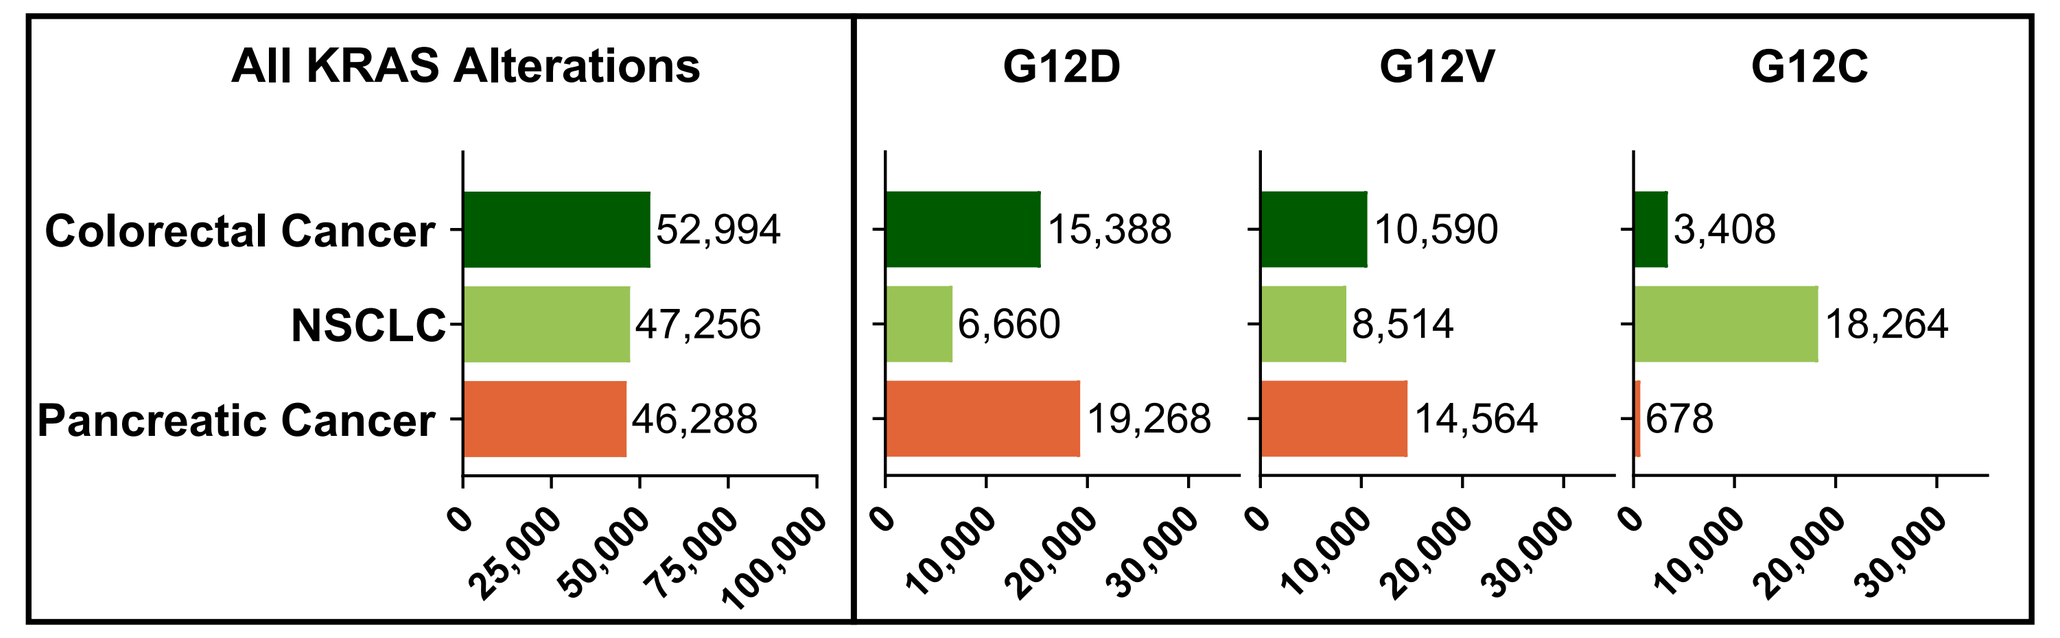
All KRAS Alterations
G12D
G12V
G12C
Colorectal Cancer
NSCLC
Pancreatic Cancer
52,994
47,256
46,288
15,388
6,660
19,268
10,590
8,514
14,564
3,408
18,264
678
0
25,000
50,000
75,000
100,000
10,000
20,000
30,000

Estimated New Patients per Year in U.S.

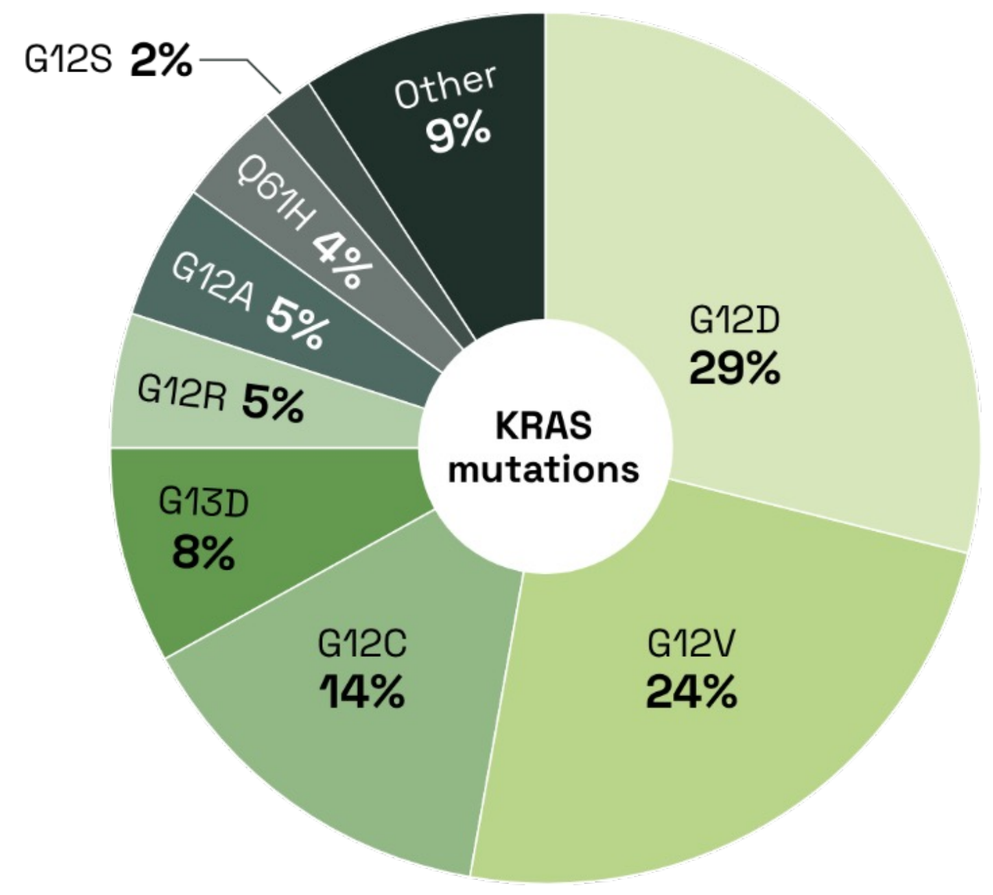
G12S 2%
Other 9%
Q61H 4%
G12A 5%
G12R 5%
G13D 8%
G12C 14%
G12V 24%
G12D 29%
KRAS mutations

There is a need for potent inhibitors that target multiple KRAS mutations and are selective for KRAS over HRAS and NRAS isoforms

CGT1263, A Selective PAN-KRAS Inhibitor, Potently Inhibits KRAS Mutants

RESULTS

CGT1263 Binds in the Switch II Pocket of GTP Bound KRAS(ON)

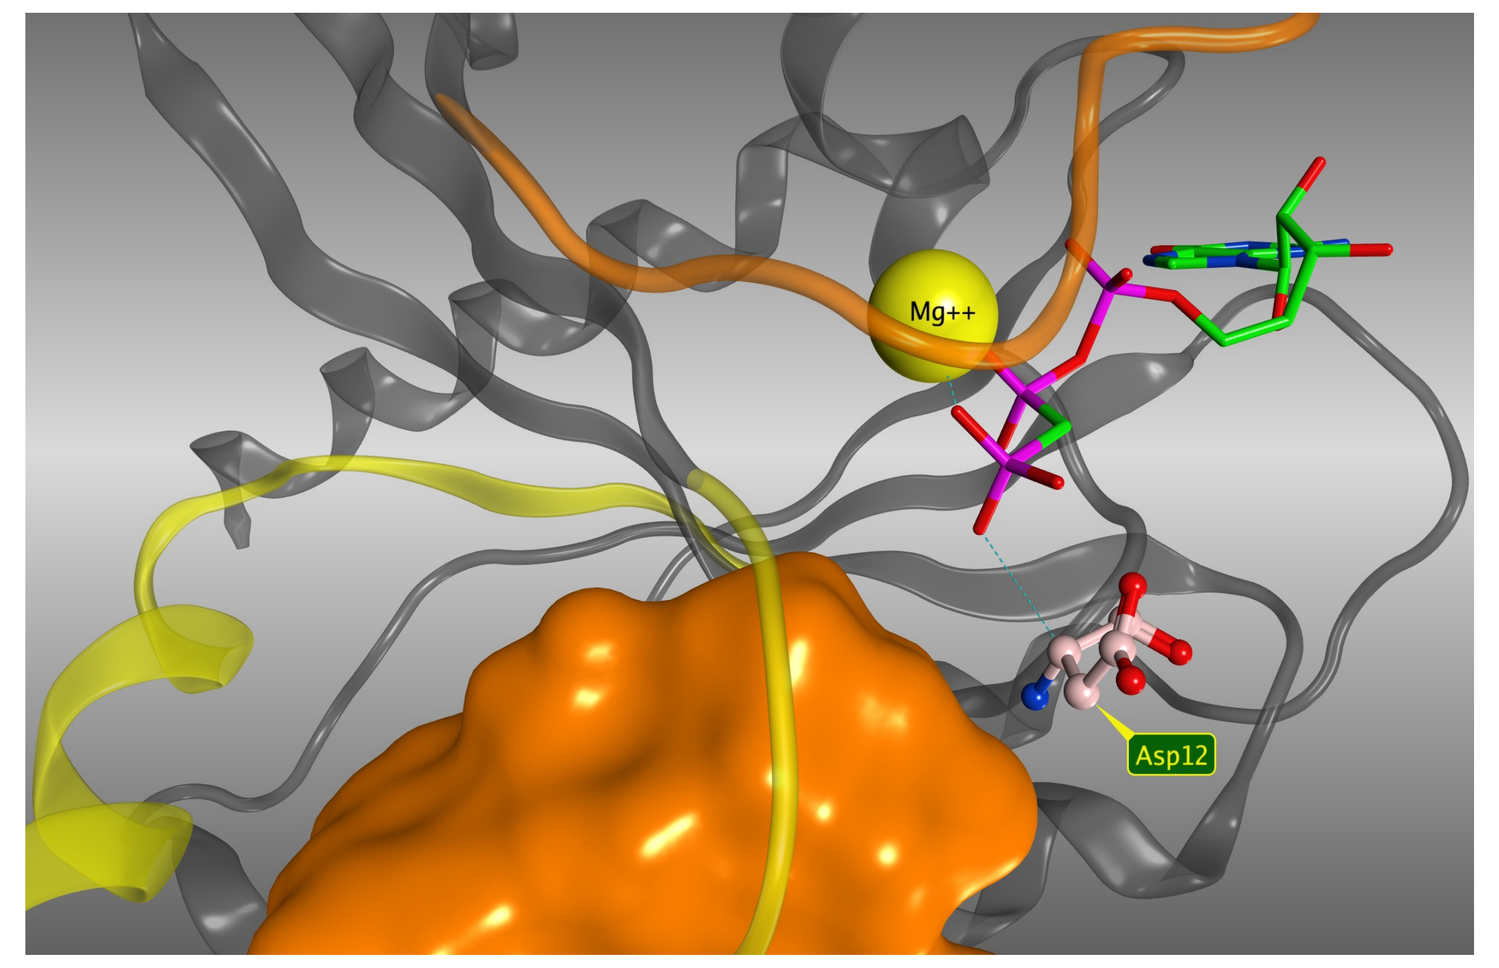
Mg++
Asp12

- 1.3Å resolution crystal structure of CGT1263 co-crystallized with GMPPCP loaded $KRAS^{G12D}$ protein was solved
- GMPPCP is clearly visible in the upper right of the image with a chelated Mg^{++}
- CGT1263 is an active GTP-KRAS(ON) Switch II binder

CGT1263 Is a Potent Pan-KRAS Binder With Selectivity Over HRAS/NRAS

RAS SPR Binding Assay K_D							Selectivity	
	KRAS						HRAS	NRAS
Nucleotide State	WT	G12V	G12D	G12C	G13D	Q61H	WT	WT
GTP	6 nM	17 nM	4 nM	4 nM	2 nM	4 nM	>250x Selective	>250x Selective
GDP	1 pM	5 pM	3 pM	8 pM	3 pM	5 pM	>500x Selective	>500x Selective

- Kinetic SPR binding of CGT1263 bound to KRAS, HRAS, and NRAS proteins each loaded with the GTP stable analog GMPPCP or GDP was determined
- pM/nM binding to KRAS WT and mutants with >250x selectivity over HRAS and NRAS was observed for CGT1263

KEY FINDINGS

- **Development candidate:** CGT1815, the prodrug of CGT1263, is progressing toward IND enabling studies
- **Binds in the Switch II pocket:** An X-ray structure of CGT1263 bound to $KRAS^{G12D}$ loaded with GTP analog GMPPCP shows CGT1263 bound in the switch II pocket, the gamma phosphate of GMPPCP is clearly visible
- **SPR confirms active KRAS binding and selectivity:** pM binding was observed by SPR to GDP loaded wild type and mutant KRAS with >500x selectivity for HRAS and NRAS
- **Sub nM potency in a panel of KRAS cell lines:** In a diverse panel of KRAS cell lines CGT1263 showed sub nM inhibition of pERK on mutant and wild type amplified lines
- **24-hr IC_{90} $KRAS^{G12D}$ target coverage at 10 mg/kg PO in dogs:** The prodrug CGT1815 showed 1.7-fold increase in AUC in mouse and dog compared to direct administration of CGT1263 with 24 hr target coverage exceeding ff_{adj} $KRAS^{G12D}$ IC_{90} in dogs when administered at 10 mg/kg
- **Superior efficacy in $KRAS^{G12D}$ TGI:** Nearly complete tumor regressions were observed with CGT1815 administered PO at 20 mg/kg equivalent dose BID in an ASPC-1 $KRAS^{G12D}$ tumor-bearing mouse model surpassing efficacy of RMC-6236 at the standard 25 mg/kg dose
- **Regressions at 10 mg/kg in $KRAS^{G12V}$ model:** Administered at 10 mg/kg BID, CGT1815 showed >90% tumor regressions, similar to RMC-6236 at 25 mg/kg
- **Selectivity**: CGT1263 has potential to alleviate the toxicological issues associated with multi-RAS inhibitors

RESULTS (cont'd)

CGT1263 Shows ≤ 1n M Potency Across a Panel of KRAS Cell Lines

Cell Line	Tumor Type	KRAS Mutation	CGT1263 Cellular IC_{50}	RMC-6236 Cellular IC_{50}	AMG-410 Cellular IC_{50}
MKN1	Gastric cancer	Wild Type Amplified	0.65 nM	8.5 nM	1.8 nM
NCI-H2009	Non-small cell lung cancer	G12A	0.11 nM	0.89 nM	0.68 nM
NCI-H358	Non-small cell lung cancer	G12C	0.22 nM	0.30 nM	1.9 nM
AsPC-1	Pancreatic cancer	G12D	0.24 nM	1.28 nM	1.1 nM
A549	Non-small cell lung cancer	G12S	0.35 nM	0.65 nM	3.9 nM
SW480	Colorectal cancer	G12V	0.37 nM	0.29 nM	1.4 nM
HCT116	Colorectal cancer	G13D	1.0 nM	0.96 nM	10.4 nM
NCI-H460	Non-small cell lung cancer	Q61H	0.30 nM	0.50 nM	2.0 nM

- Inhibition of pERK was determined for CGT1263, RMC-6236, and AMG-410 in a variety of cell lines and tumor types to interrogate activity across a range of KRAS mutations
- CGT1263 showed pM/nM pERK inhibition across the spectrum of KRAS cell lines shown

RESULTS (cont'd)

CGT1815 Is a Prodrug of CGT1263 That Has Been Optimized for Human Performance

Mouse Plasma PK

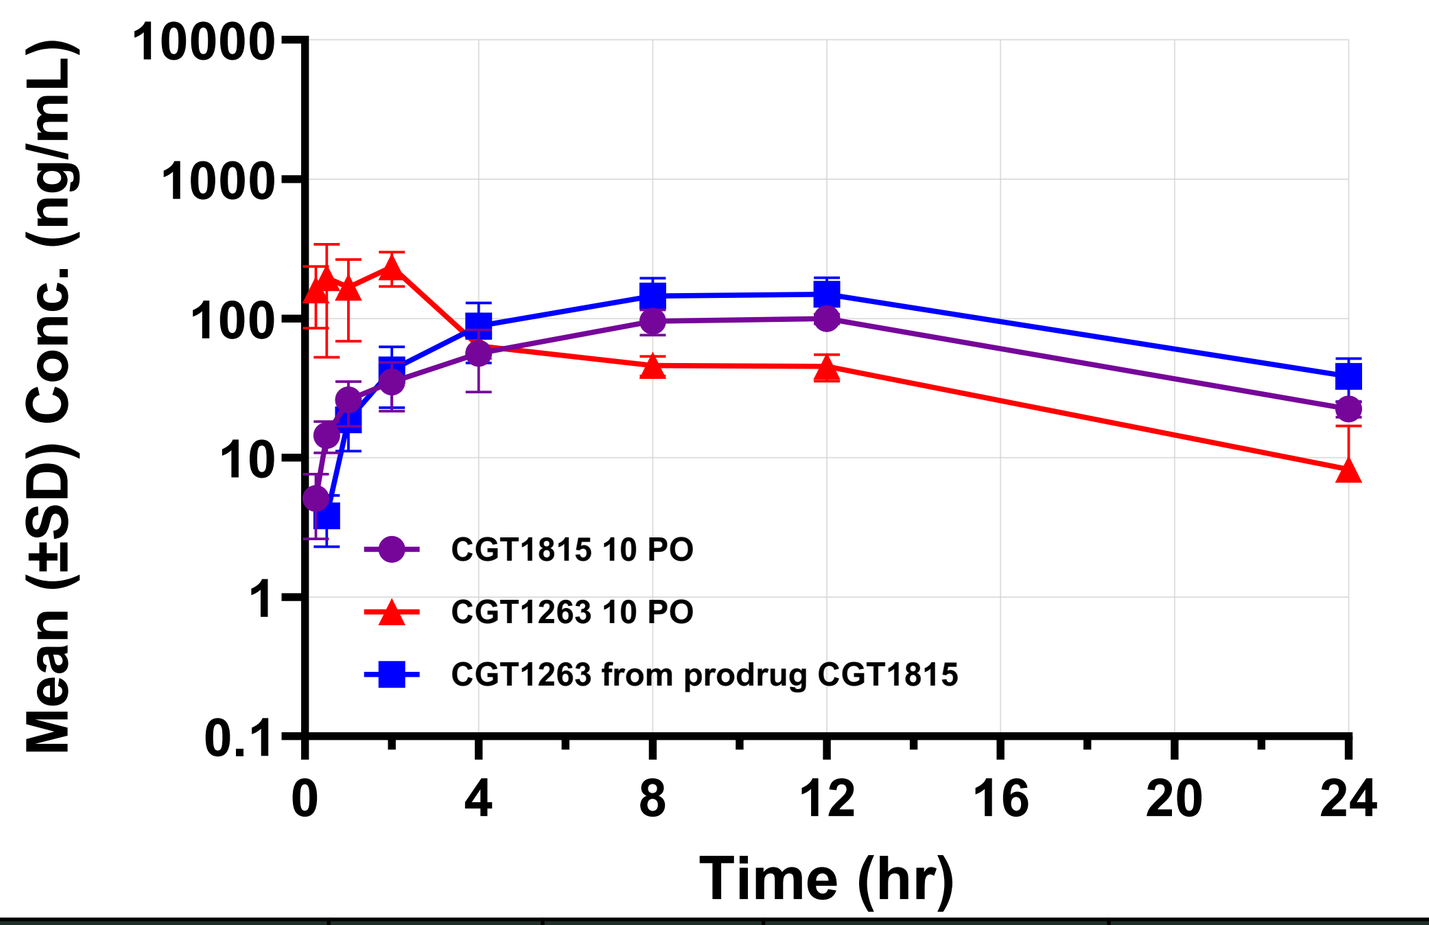
Mean (±SD) Conc. (ng/mL)
10000
1000
100
10
1
0.1
0
4
8
12
16
20
24
Time (hr)
CGT1815 10 PO
CGT1263 10 PO
CGT1263 from prodrug CGT1815

Compound	$t_{1/2}$ (hr)	T_{max} (hr)	C_{max} (ng/mL)	AUC_{0-24} (ng*hr/mL)
CGT1815*	5.9	9.3	103	1449
CGT1263	6.4	1.5	302	1261
CGT1263 from prodrug CGT1815	8.2	9.3	151	2202

Dog Plasma PK

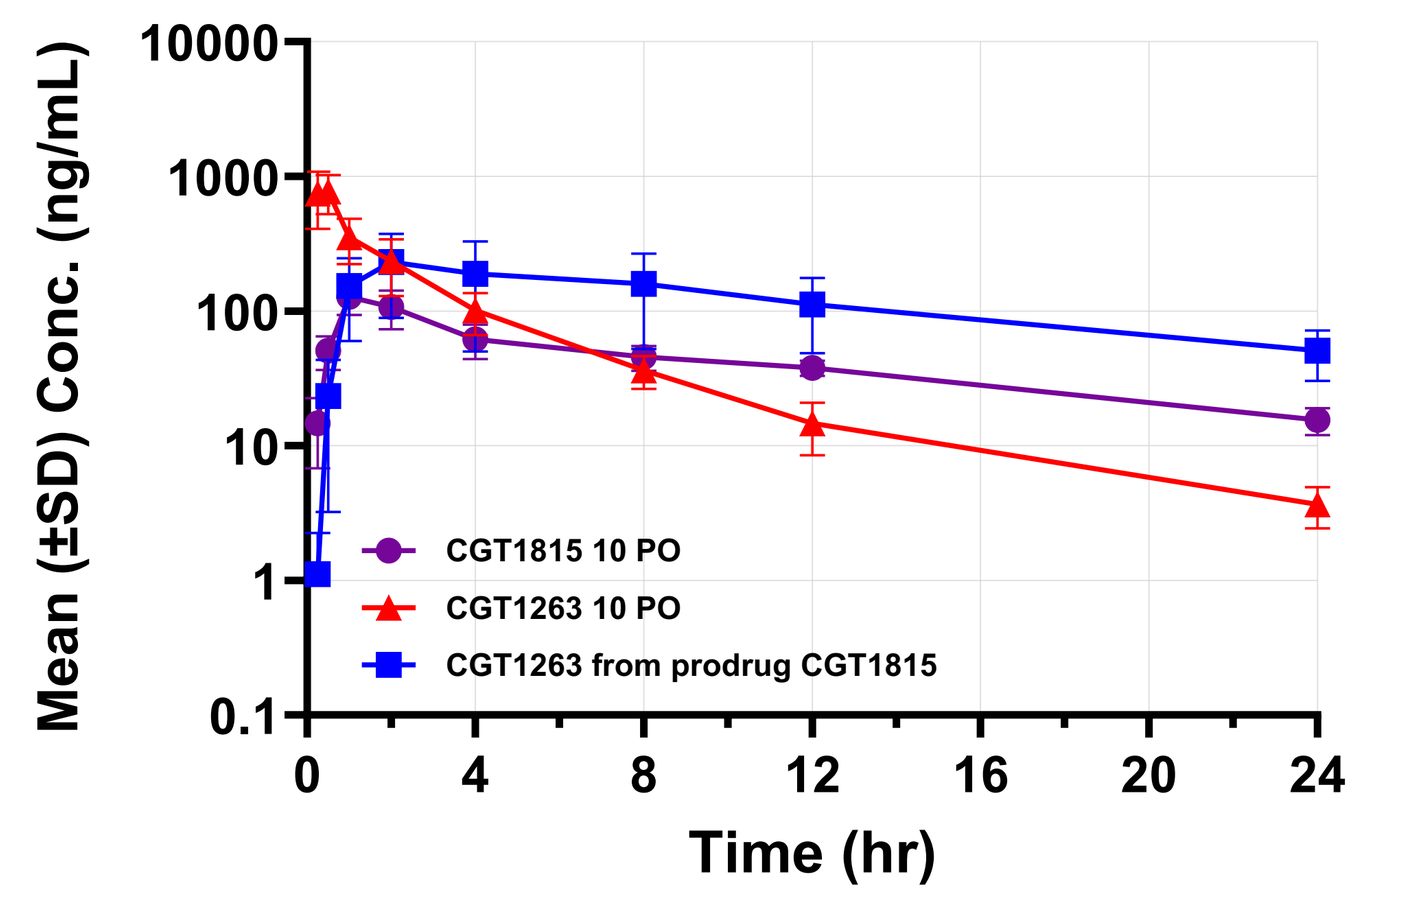
Mean (±SD) Conc. (ng/mL)
10000
1000
100
10
1
0.1
0
4
8
12
16
20
24
Time (hr)
CGT1815 10 PO
CGT1263 10 PO
CGT1263 from prodrug CGT1815

Compound	$t_{1/2}$ (hr)	T_{max} (hr)	C_{max} (ng/mL)	AUC_{0-24} (ng*hr/mL)
CGT1815*	13	1.3	129	1014
CGT1263	4.8	0.3	895	1600
CGT1263 from prodrug CGT1815	14	2.0	232	2820

- CGT1815 is an optimized prodrug of CGT1263: the pharmacokinetic performance of CGT1815, CGT1263, and CGT1263 from prodrug CGT1815 was compared in mouse and dog administered at 10 mg/kg PO
- In both species, $T_{1/2}$ and T_{max} of CGT1263 increased with prodrug administration, notably dog $T_{1/2}$ increased from 4.8 hr to 14 hr, providing sustained drug plasma concentrations
- CGT1263 AUC increased 1.7-fold in both species when administered as prodrug
- CGT1263 plasma concentrations surpassed the ff_{adj} cellular $KRAS^{G12D}$ IC_{90} of 35 ng/mL for 24 hr in dog after a single oral dose of CGT1815 prodrug at 10 mg/kg equivalent

*CGT1815 dose normalized to active.

CGT1815 Showed Regressions at 10 mg/kg in ASPC-1 $KRAS^{G12D}$ and Panc038-Y062 $KRAS^{G12V}$ Models

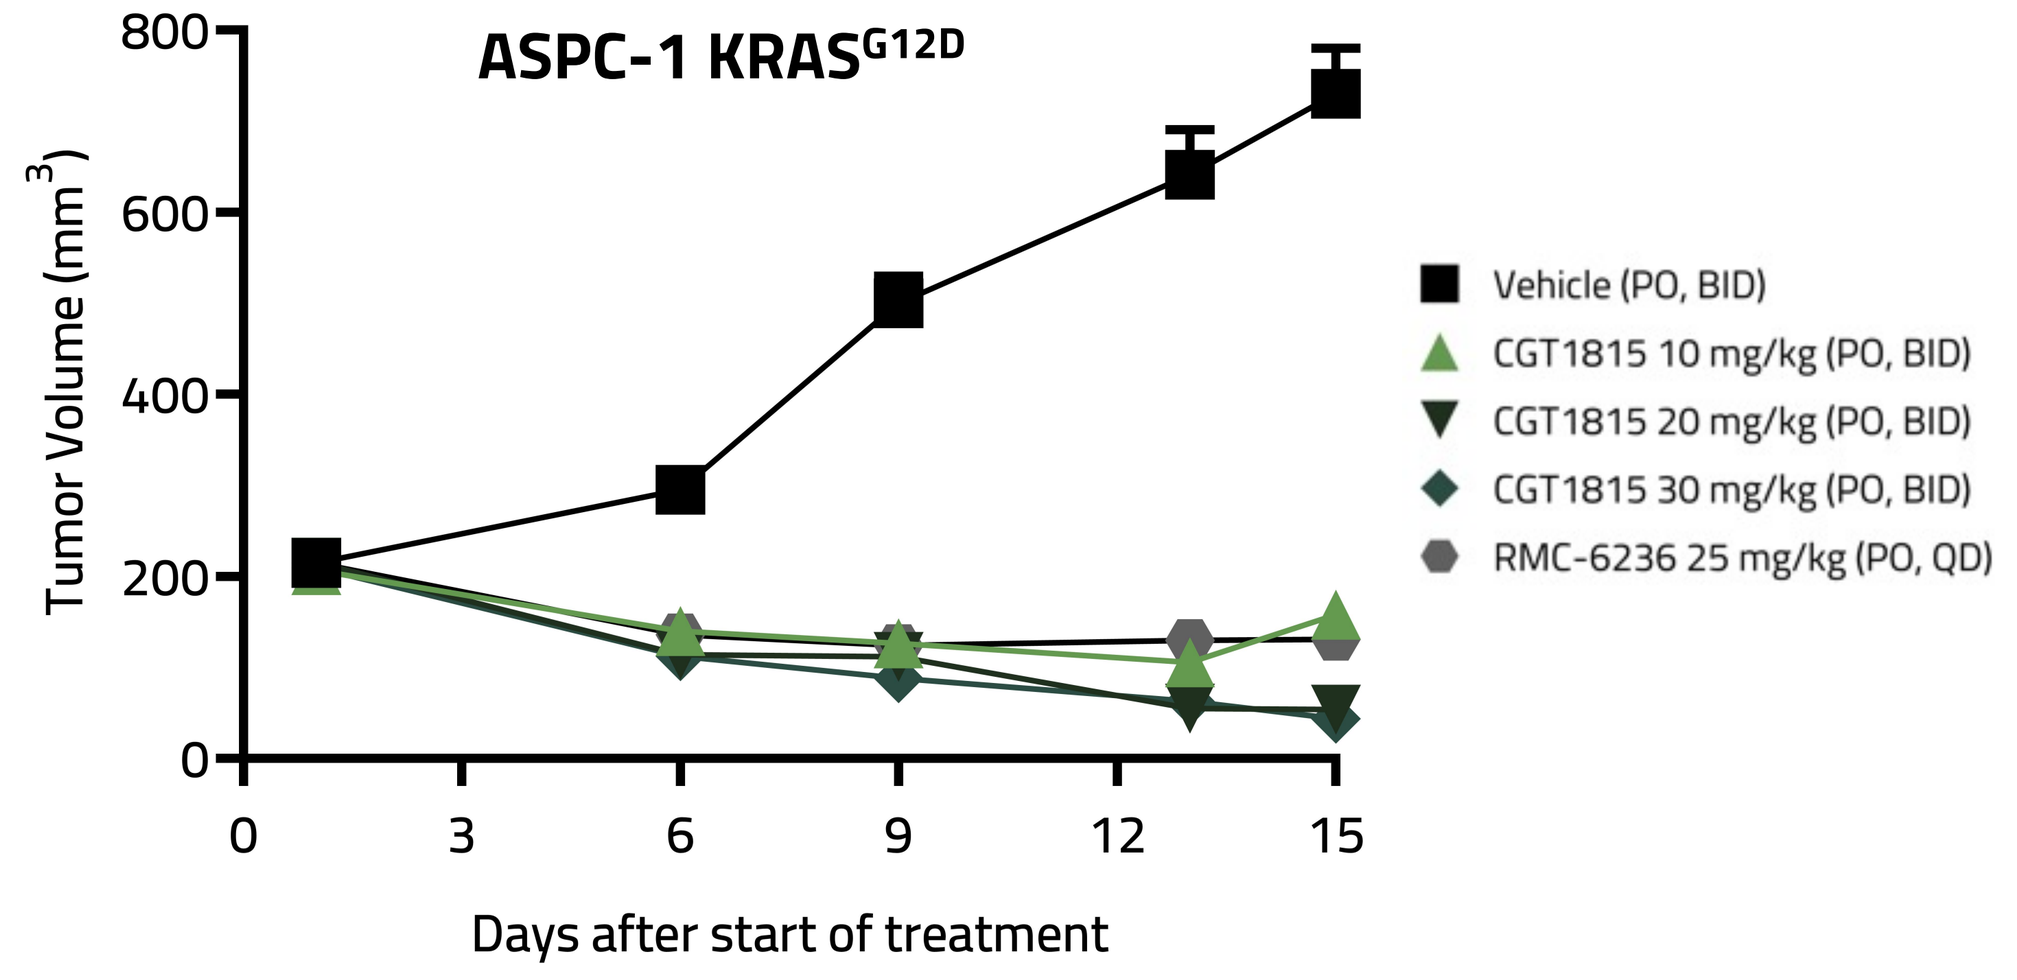
ASPC-1 KRAS G12D
Tumor Volume (mm3)
800
600
400
200
0
0
3
6
9
12
15
Days after start of treatment
Vehicle (PO, BID)
CGT1815 10 mg/kg (PO, BID)
CGT1815 20 mg/kg (PO, BID)
CGT1815 30 mg/kg (PO, BID)
RMC-6236 25 mg/kg (PO, QD)

Treatment	% TGI Day 15	Max % Regression
CGT1815 10 mg/kg (PO, BIDx14)	78	40 (D13)
CGT1815 20 mg/kg (PO, BIDx14)	93	75 (D13)
CGT1815 30 mg/kg (PO, BIDx14)	94	79 (D15)
RMC-6236 25 mg/kg (PO, QDx14)	82	39 (D15)

- CGT1815 and RMC-6236 were administered to mice bearing staged 200 mm^3 $KRAS^{G12D}$ ASPC-1 tumors for 15 d
- CGT1815 administered at 10 mg/kg BID afforded equivalent tumor regressions to RMC-6236 25 mg/kg QD
- CGT1815 showed >90% tumor growth inhibition at the 20 and 30 mg/kg BID PO doses

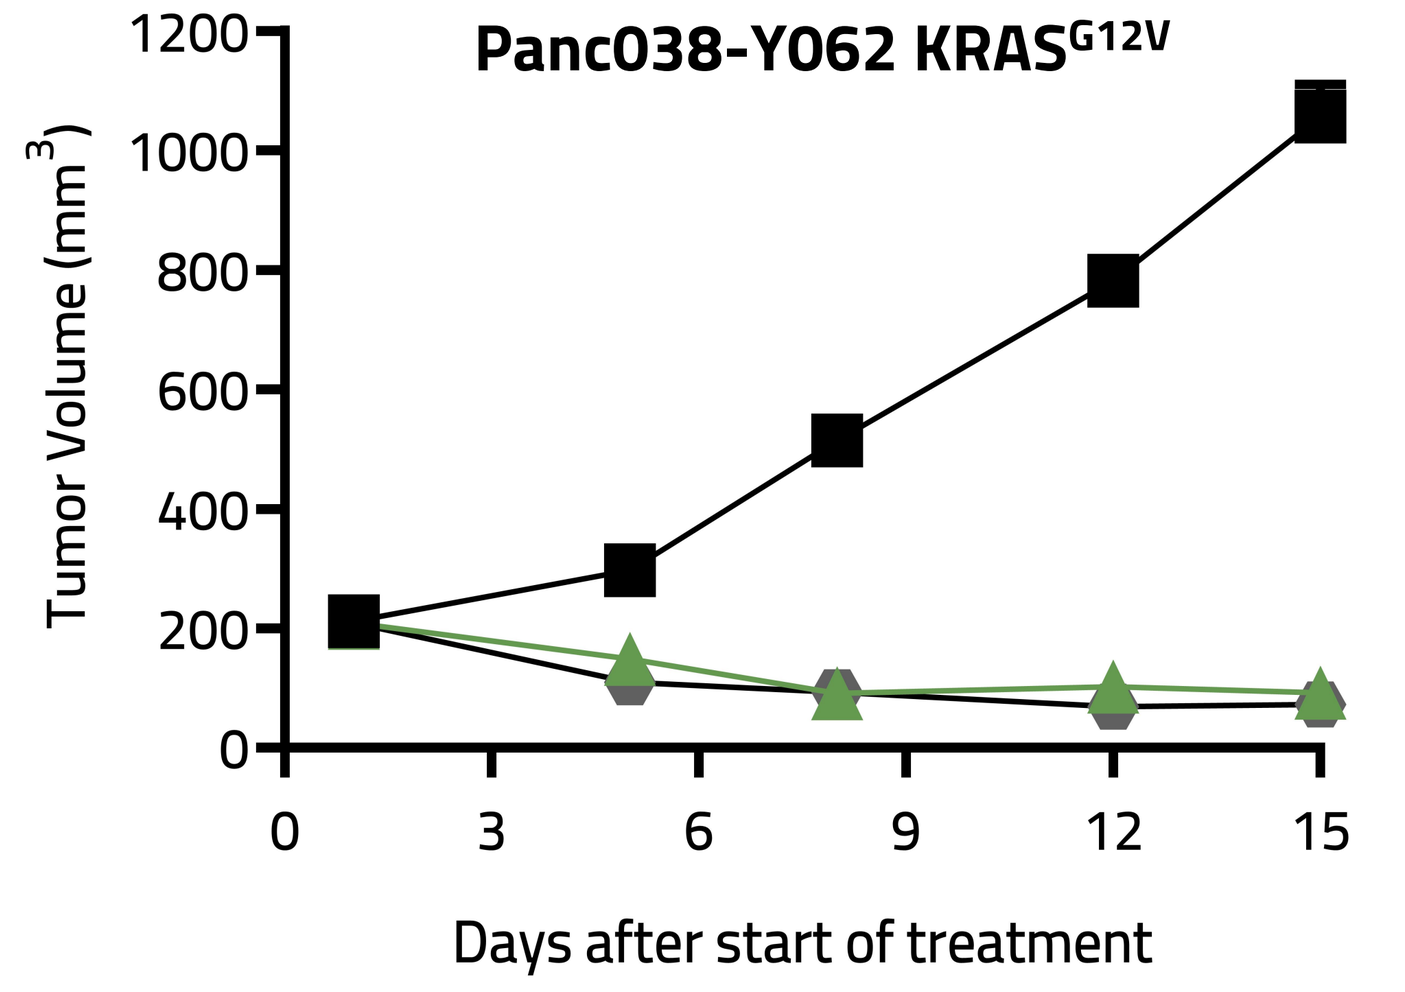
Panc038-Y062 KRAS G12V
Tumor Volume (mm3)
1200
1000
800
600
400
200
0
0
3
6
9
12
15
Days after start of treatment

- CGT1815 and RMC-6236 were administered to mice bearing staged 250 mm^3 $KRAS^{G12V}$ Panc038-Y062 PDX tumors for 15 days
- CGT1815 administered at 10 mg/kg BID demonstrated >90% tumor regressions equivalent to RMC-6236 administered at 25 mg/kg QD

CGT1815 is progressing toward IND enabling studies

References: 1. Moore A et al. *Nat. Rev. Drug Discov*. 2020;19(8):533–52. **2.** The AACR Project GENIE Consortium. AACR Project GENIE: Powering Precision Medicine Through An International Consortium, Cancer Discovery 2017. **3.** Skoulidis et al. WCLC 2025.

Acknowledgments/Disclosures This study was funded by Cogent Biosciences. All authors are employed by Cogent Biosciences.

AACR-NCI-EORTC International Conference | October 22-26, 2025, Boston, MA