Results from the Summit pivotal trial: Symptom improvement correlates with reductions in objective measures of disease in adults with non-advanced systemic mastocytosis

Tracy I. George[1,2], Lindsay Rein[3], Daniel J. DeAngelo[4], Kishore Polireddy[5], Akhilesh Atluri[5], Peter Chang[5], Adekemi Taylor[5], Amanda Pilla[6], Jenna Zhang[6], Lei Sun[6], Nisha Shah[6], Priya Singh[6], Raheela Soomro[6], Jessica Sachs[6], Nathan A Boggs[7]

[1]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT, USA; [2]ARUP Laboratories, Salt Lake City, UT, USA; [3]Duke Cancer Institute, Durham, NC, USA; [4]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, MA, USA; [5]Certara Drug Development Solutions, [6]Cogent Biosciences Inc., Waltham, MA, USA; [7]Department of Medicine, Uniformed Services University & Walter Reed National Military Medical Center, Bethesda, MD, USA;

cogent BIOSCIENCES

532

INTRODUCTION

Nonadvanced Systemic Mastocytosis (NonAdvSM) Assessment of Patient-Reported Outcome Measures and Objective Markers of Disease

- NonAdvSM is driven by the *KIT* p.D816V mutation in ~95% of patients, leading to neoplastic mast cell (MC) infiltration, mediator release, and a range of debilitating symptoms[1]
- Primary evaluation is through patient-reported outcome measures that assess symptom severity[2]
- Objective markers of disease, such as serum tryptase, provide quantitative and disease-defining measures of pharmacodynamic activity that complement symptom-based endpoints[3]
 - Serum tryptase of >20 ng/mL is recognized by the World Health Organization (WHO) as a minor diagnostic criterion for SM[4]

Response in Objective Measures of Disease and Corresponding Symptom Improvement

- Bezuclastinib is an oral, potent, selective type 1 tyrosine kinase inhibitor with activity against KIT D816V[5,6]
- The Summit trial of bezuclastinib in NonAdvSM showed clinically meaningful symptom improvement and significant reductions in objective disease markers along with a favorable safety profile[7]

Bezuclastinib treatment resulted in consistent reductions across all objective markers of disease[7], including serum tryptase

	Bezuclastinib	Placebo
Median serum tryptase at 24 weeks, ng/mL (range)	6.1 (2.1-97.7)	42.5 (6.4-298.0)
Patients reaching ≥50% reduction in serum tryptase at 24 weeks [ITT population[a]], % (n/N)	87.4 (104/119)	0 (0/60)
Patients reaching ≥50% reduction in serum tryptase at 24 weeks [patients with 24w data], % (n/N)	95.4 (104/109)	0 (0/57)
Patients achieving <20 ng/mL[b], % (n/N)	83.1 (74/89)	0 (0/47)
Patients achieving <11.4 ng/mL[b], % (n/N)	77.9 (81/104)	0 (0/55)

Symptom improvement significantly correlated with serum tryptase reduction[7]

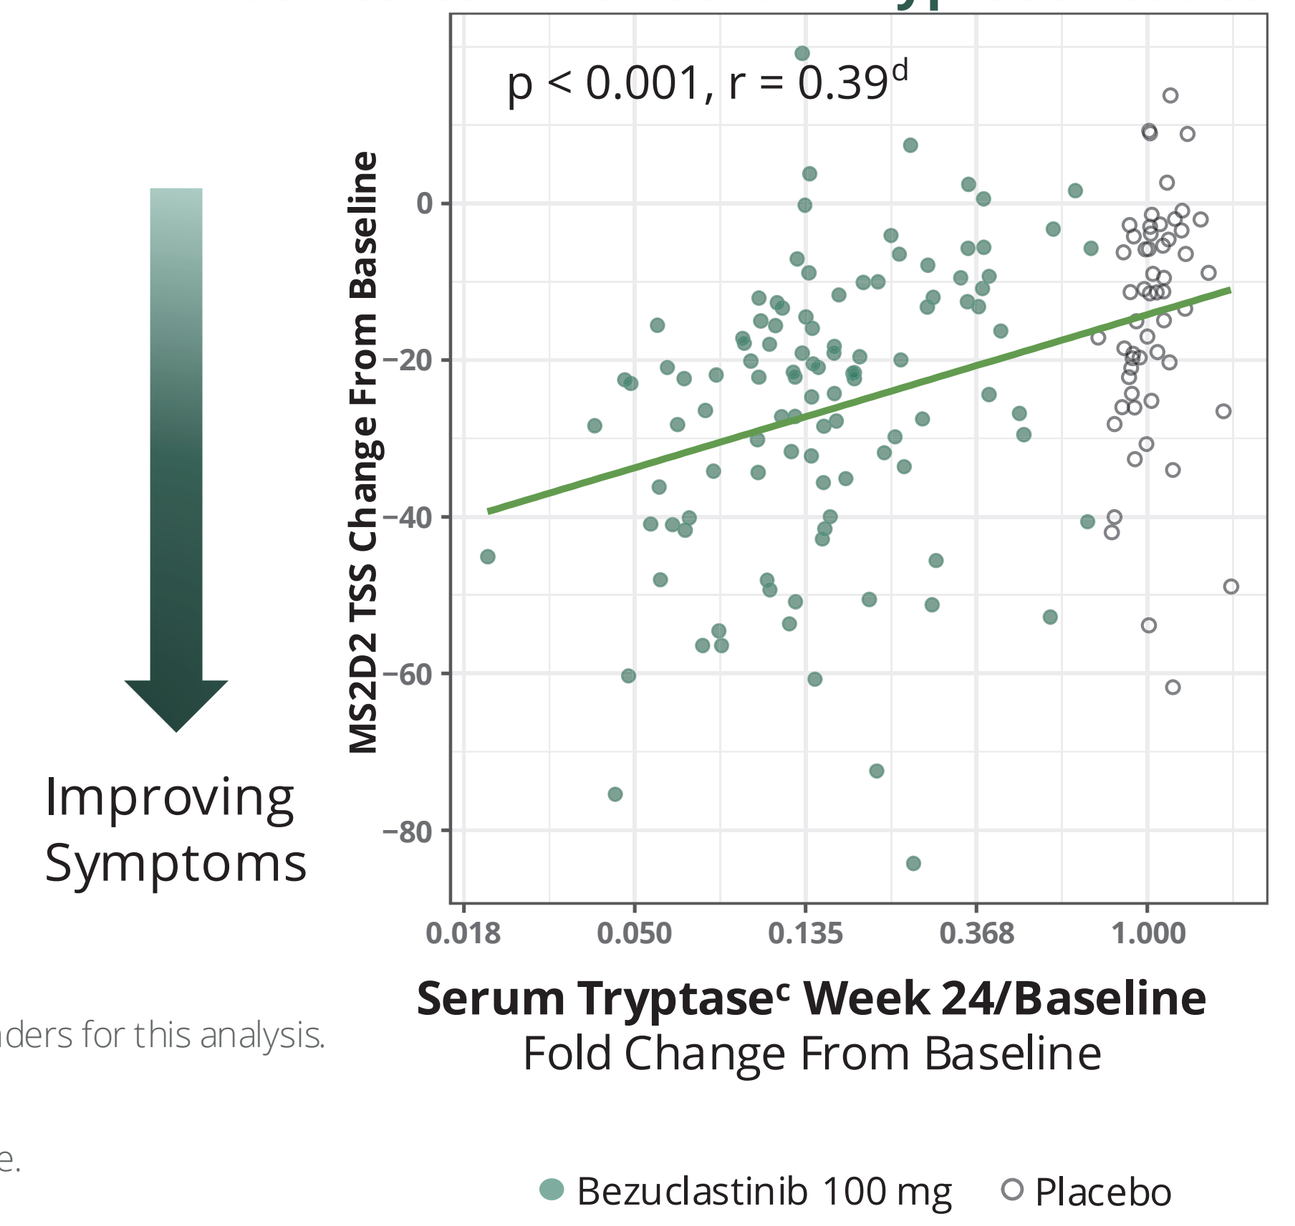
p < 0.001, r = 0.39[d]
Improving Symptoms
MS2D2 TSS Change From Baseline
0
−20
−40
−60
−80
0.018
0.050
0.135
0.368
1.000
Serum Tryptase[c] Week 24/Baseline
Fold Change From Baseline
Bezuclastinib 100 mg
Placebo

[a]ITT population: patients missing data at baseline and/or week 24 are treated as nonresponders for this analysis.
[b]Of patients with baseline serum tryptase above threshold.
[c]Bezuclastinib (n=107) and placebo (n=56).
[d]Pearson correlation coefficient for r value. Pearson correlation testing for two-sided p value.
ITT, intention to treat.

Here, we further characterize the relationship between reaching <20 ng/mL serum tryptase and symptom severity in NonAdvSM

RESULTS

Treatment-Driven Reduction of Serum Tryptase to the WHO SM Diagnostic Threshold (<20 ng/mL) at Week 24 Correlates with Greater Improvement in MS2D2 Total Symptom Score, Symptom Domains, and Individual Symptoms

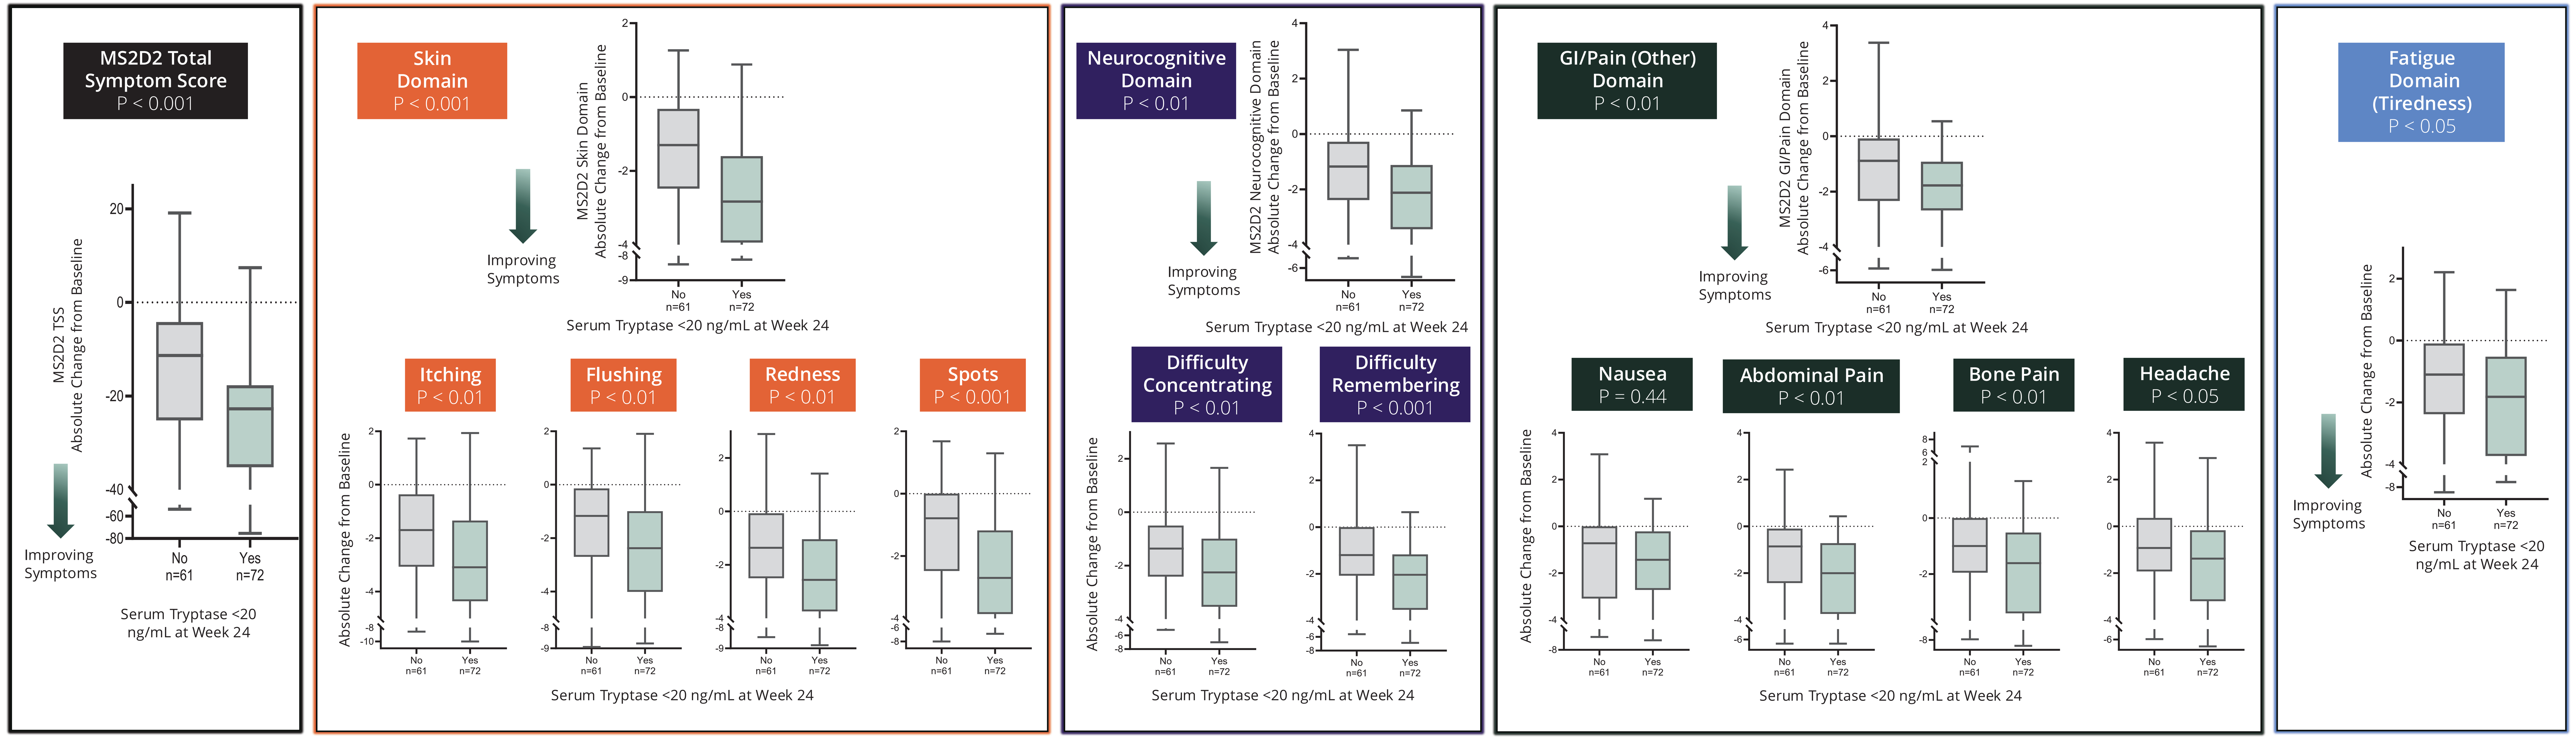
MS2D2 Total Symptom Score
P < 0.001
MS2D2 TSS Absolute Change from Baseline
Improving Symptoms
No n=61
Yes n=72
Serum Tryptase <20 ng/mL at Week 24
Skin Domain
P < 0.001
MS2D2 Skin Domain Absolute Change from Baseline
Itching
P < 0.01
Flushing
P < 0.01
Redness
P < 0.01
Spots
P < 0.001
Absolute Change from Baseline
Neurocognitive Domain
P < 0.01
MS2D2 Neurocognitive Domain Absolute Change from Baseline
Difficulty Concentrating
P < 0.01
Difficulty Remembering
P < 0.001
GI/Pain (Other) Domain
P < 0.01
MS2D2 GI/Pain Domain Absolute Change from Baseline
Nausea
P = 0.44
Abdominal Pain
P < 0.01
Bone Pain
P < 0.01
Headache
P < 0.05
Fatigue Domain (Tiredness)
P < 0.05

Significance was tested using the Wilcoxon rank-sum test.

METHODS

Summit (NCT05186753): Pivotal Phase 2, Multicenter, Randomized, Double-Blind, Placebo-Controlled Study Evaluating Bezuclastinib in NonAdvSM

PART 2: EXPANSION

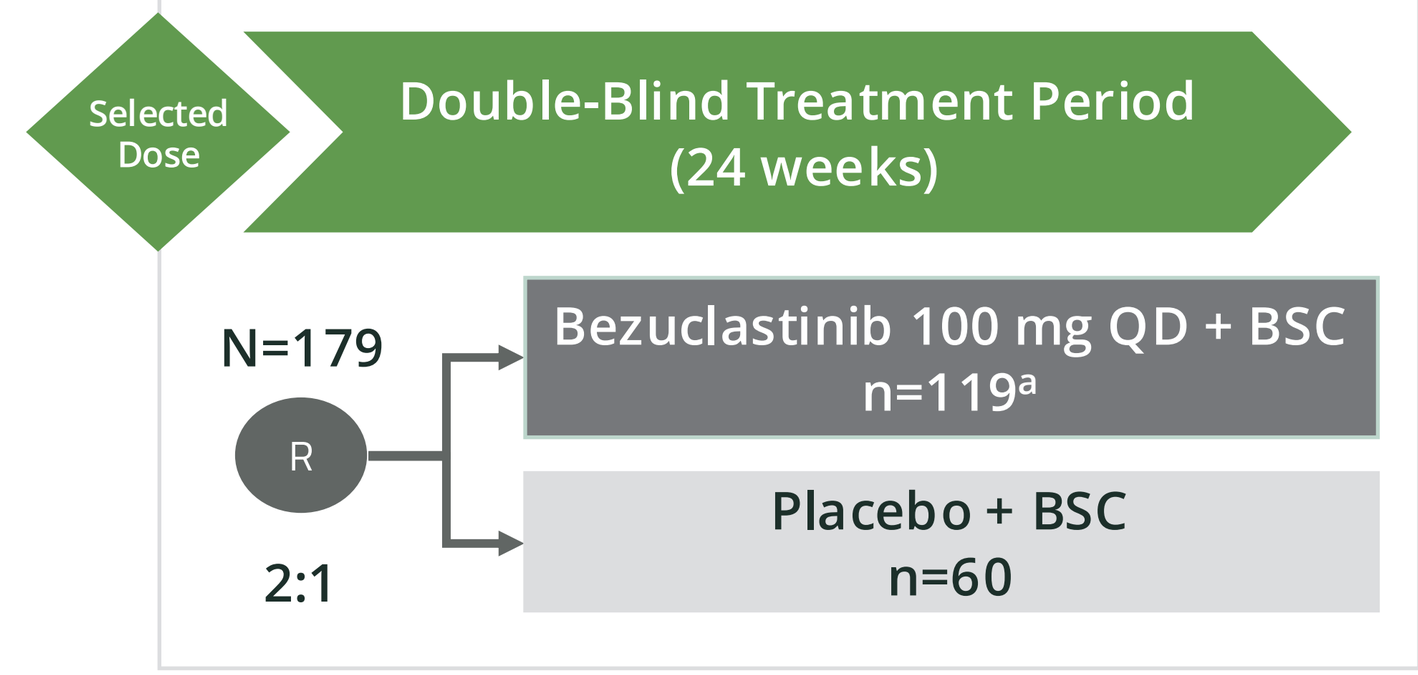
Selected Dose
Double-Blind Treatment Period (24 weeks)
N=179
R
2:1
Bezuclastinib 100 mg QD + BSC n=119[a]
Placebo + BSC n=60

- The relationship between improvement in symptom severity (measured by the MS2D2) and reduction in serum tryptase, an objective indicator of MC burden, was assessed in the Summit trial at week 24
 - Symptom severity scores included in this analysis were MS2D2 TSS, 4 symptom domains (skin, neurological, fatigue, and other [GI/pain]), and 11 individual symptoms

[a]One patient withdrew consent and did not receive treatment.

BM, bone marrow; BMM, bone marrow mastocytosis; BSC, best supportive care; H1RA, histamine receptor type 1 antagonist; HR2A, histamine receptor type 2 antagonist; ISM, indolent SM; LTRA, leukotriene receptor antagonists; MC, mast cell; MS2D2, mastocytosis symptom severity daily diary; NonAdvSM, non-advanced systemic mastocytosis; PPI, proton pump inhibitors; QD, once daily; R, randomized; SSM, smoldering SM; TSS, total symptom score; VAF, variant allele frequency; WHO, World Health Organization.

MS2D2: A Comprehensive Patient-Reported Outcome Measure of Symptom Severity in Patients Across the Spectrum of NonAdvSM

- The MS2D2[a] is a 17-item measure addressing signs and symptoms of NonAdvSM
- Eleven symptoms within 4 domains are included in MS2D2 TSS
- Severity of each symptom is assessed daily from 0 (none) to 10 (worst possible)
- TSS is analyzed as a 14-day average
- Data from Summit support MS2D2 as a reliable, valid and "fit for purpose" PRO measure to assess treatment efficacy[a]

MS2D2 Total Symptom Score (TSS)

Domain	Symptom
Skin	• Itching • Flushing • Skin redness • Spots
Neurocognitive	• Difficulty concentrating • Difficulty remembering
Fatigue	• Tiredness
GI/Pain (Other)	Gastrointestinal • Nausea • Abdominal pain Pain • Headache • Bone pain

[a]MS2D2 developed according to FDA Guidance for Industry PRO measures and regulatory agency feedback and reached alignment with FDA for use of MS2D2 TSS in Part 2 of the registration-directed Summit trial.
FDA, US Food and Drug Administration; MS2D2, mastocytosis symptom severity daily diary; NonAdvSM, non-advanced systemic mastocytosis; PRO, patient-reported outcome.

CONCLUSIONS

Summit Part 2 data demonstrated that treatment-driven reduction of serum tryptase to the WHO SM diagnostic threshold (<20 ng/mL) at week 24 significantly correlates with greater symptom improvement as measured by MS2D2

- Treatment-driven reduction of serum tryptase to the WHO SM diagnostic threshold (<20 ng/mL) at week 24 was significantly correlated with:
 - All symptom domains: skin, neurocognitive, fatigue, and GI/pain (other)
 - Ten of the eleven individual symptoms
- Correlation between reduction in serum tryptase and improvement in symptom severity is consistent with the pathogenetic link between neoplastic mast cells and clinical symptomatology
- Response in objective measures of disease, such as serum tryptase, and corresponding symptom improvement indicate modification of underlying disease

American Academy of Allergy, Asthma & Immunology (AAAAI) Annual Meeting
February 27 – March 2, 2026 | Philadelphia, PA
For questions, please contact MedicalAffairs@cogentbio.com

1. Farmer I, Radia DH. Curr Hematol Malig Rep. 2024;19(5):197-207. 2. Akin C, et al. J Allergy Clin Immunol. 2024;153(2):AB223. 3. Pardanani A. Am J Hematol. 2023;98(7):1097-1116. 4. Khoury JD, et al. Leukemia 2022; 36:1703-19. 5. DeAngelo DJ, et al. Hemasphere. 2022;6(suppl). 6. Guarnieri A, et al. Poster presented at: AACR Annual Meeting; April 8-13, 2022; New Orleans, LA. 7. Rein LAM, et al. Oral presentation at: ASH Annual Meeting; December 6-9, 2025; Orlando, FL.

The authors would like to thank the patients participating in this study, as well as their families, the investigators, patient advocates, research and operations staff. This study is sponsored by Cogent Biosciences.

To obtain a digital copy of this poster, please scan the Quick Response (QR) code. Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from AAAAI or the authors of this poster.