Evaluation of Bone Formation Marker Changes in Summit, a Trial Assessing Bezuclastinib in Adults with Non-Advanced Systemic Mastocytosis : An Exploratory Analysis

Nathan A Boggs[1], Lindsay Rein[2], Thanh D. Hoang[1], Amanda Pilla[3], Jenna Zhang[3], Lei Sun[3], Nisha Shah[3], Priya Singh[3], Allison Harney[3], Bhaswati Sengupta[3], Raheela Soomro[3], Rachael Easton[3], Jessica Sachs[3], Frank Siebenhaar[4, 5]

[1]Department of Medicine, Uniformed Services University & Walter Reed National Military Medical Center, Bethesda, MD, USA; [2]Duke Cancer Institute, Durham, NC, USA; [3]Cogent Biosciences Inc., Waltham, MA, USA; [4]Institute of Allergology, Charité-Universitaetsmedizin Berlin, Berlin, Germany; [5]Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Immunology and Allergology IA, Berlin, Germany

cogent BIOSCIENCES

520

INTRODUCTION

Bezuclastinib (CGT9486) is an oral, potent, and selective type 1 TKI that reduces symptoms of nonadvanced systemic mastocytosis[1-3]

- Mast cells play a critical role in bone turnover, metabolism, and homeostasis[4]
- Nonadvanced SM (NonAdvSM) is characterized by mast cell accumulation and activation, which can lead to musculoskeletal impacts including[5-9]:
 - — Changes in bone remodeling markers
 - — Skeletal complications such as osteolysis, osteopenia, and osteoporosis
 - — Increased risk of fragility fractures
- The *KIT* c.2447 A>T (p.D816V) mutation is found in ~95% of patients with NonAdvSM[10]
- Bezuclastinib is highly active against KIT D816V, has minimal brain penetration, and spares closely related kinases, which minimizes off-target toxicities, such as bleeding, cognitive impairment, edema, and pleural effusion[1-3, 11]
- In the Summit trial, which evaluated bezuclastinib in patients with NonAdvSM, clinically meaningful symptom improvement and significant reductions in objective disease markers were demonstrated along with a favorable safety profile[3]
- Here we report the effect of bezuclastinib on bone formation markers, bone mineral density, and T-score

Bone remodeling cycle in NonAdvSM[4-9] and proposed effect of bezuclastinib

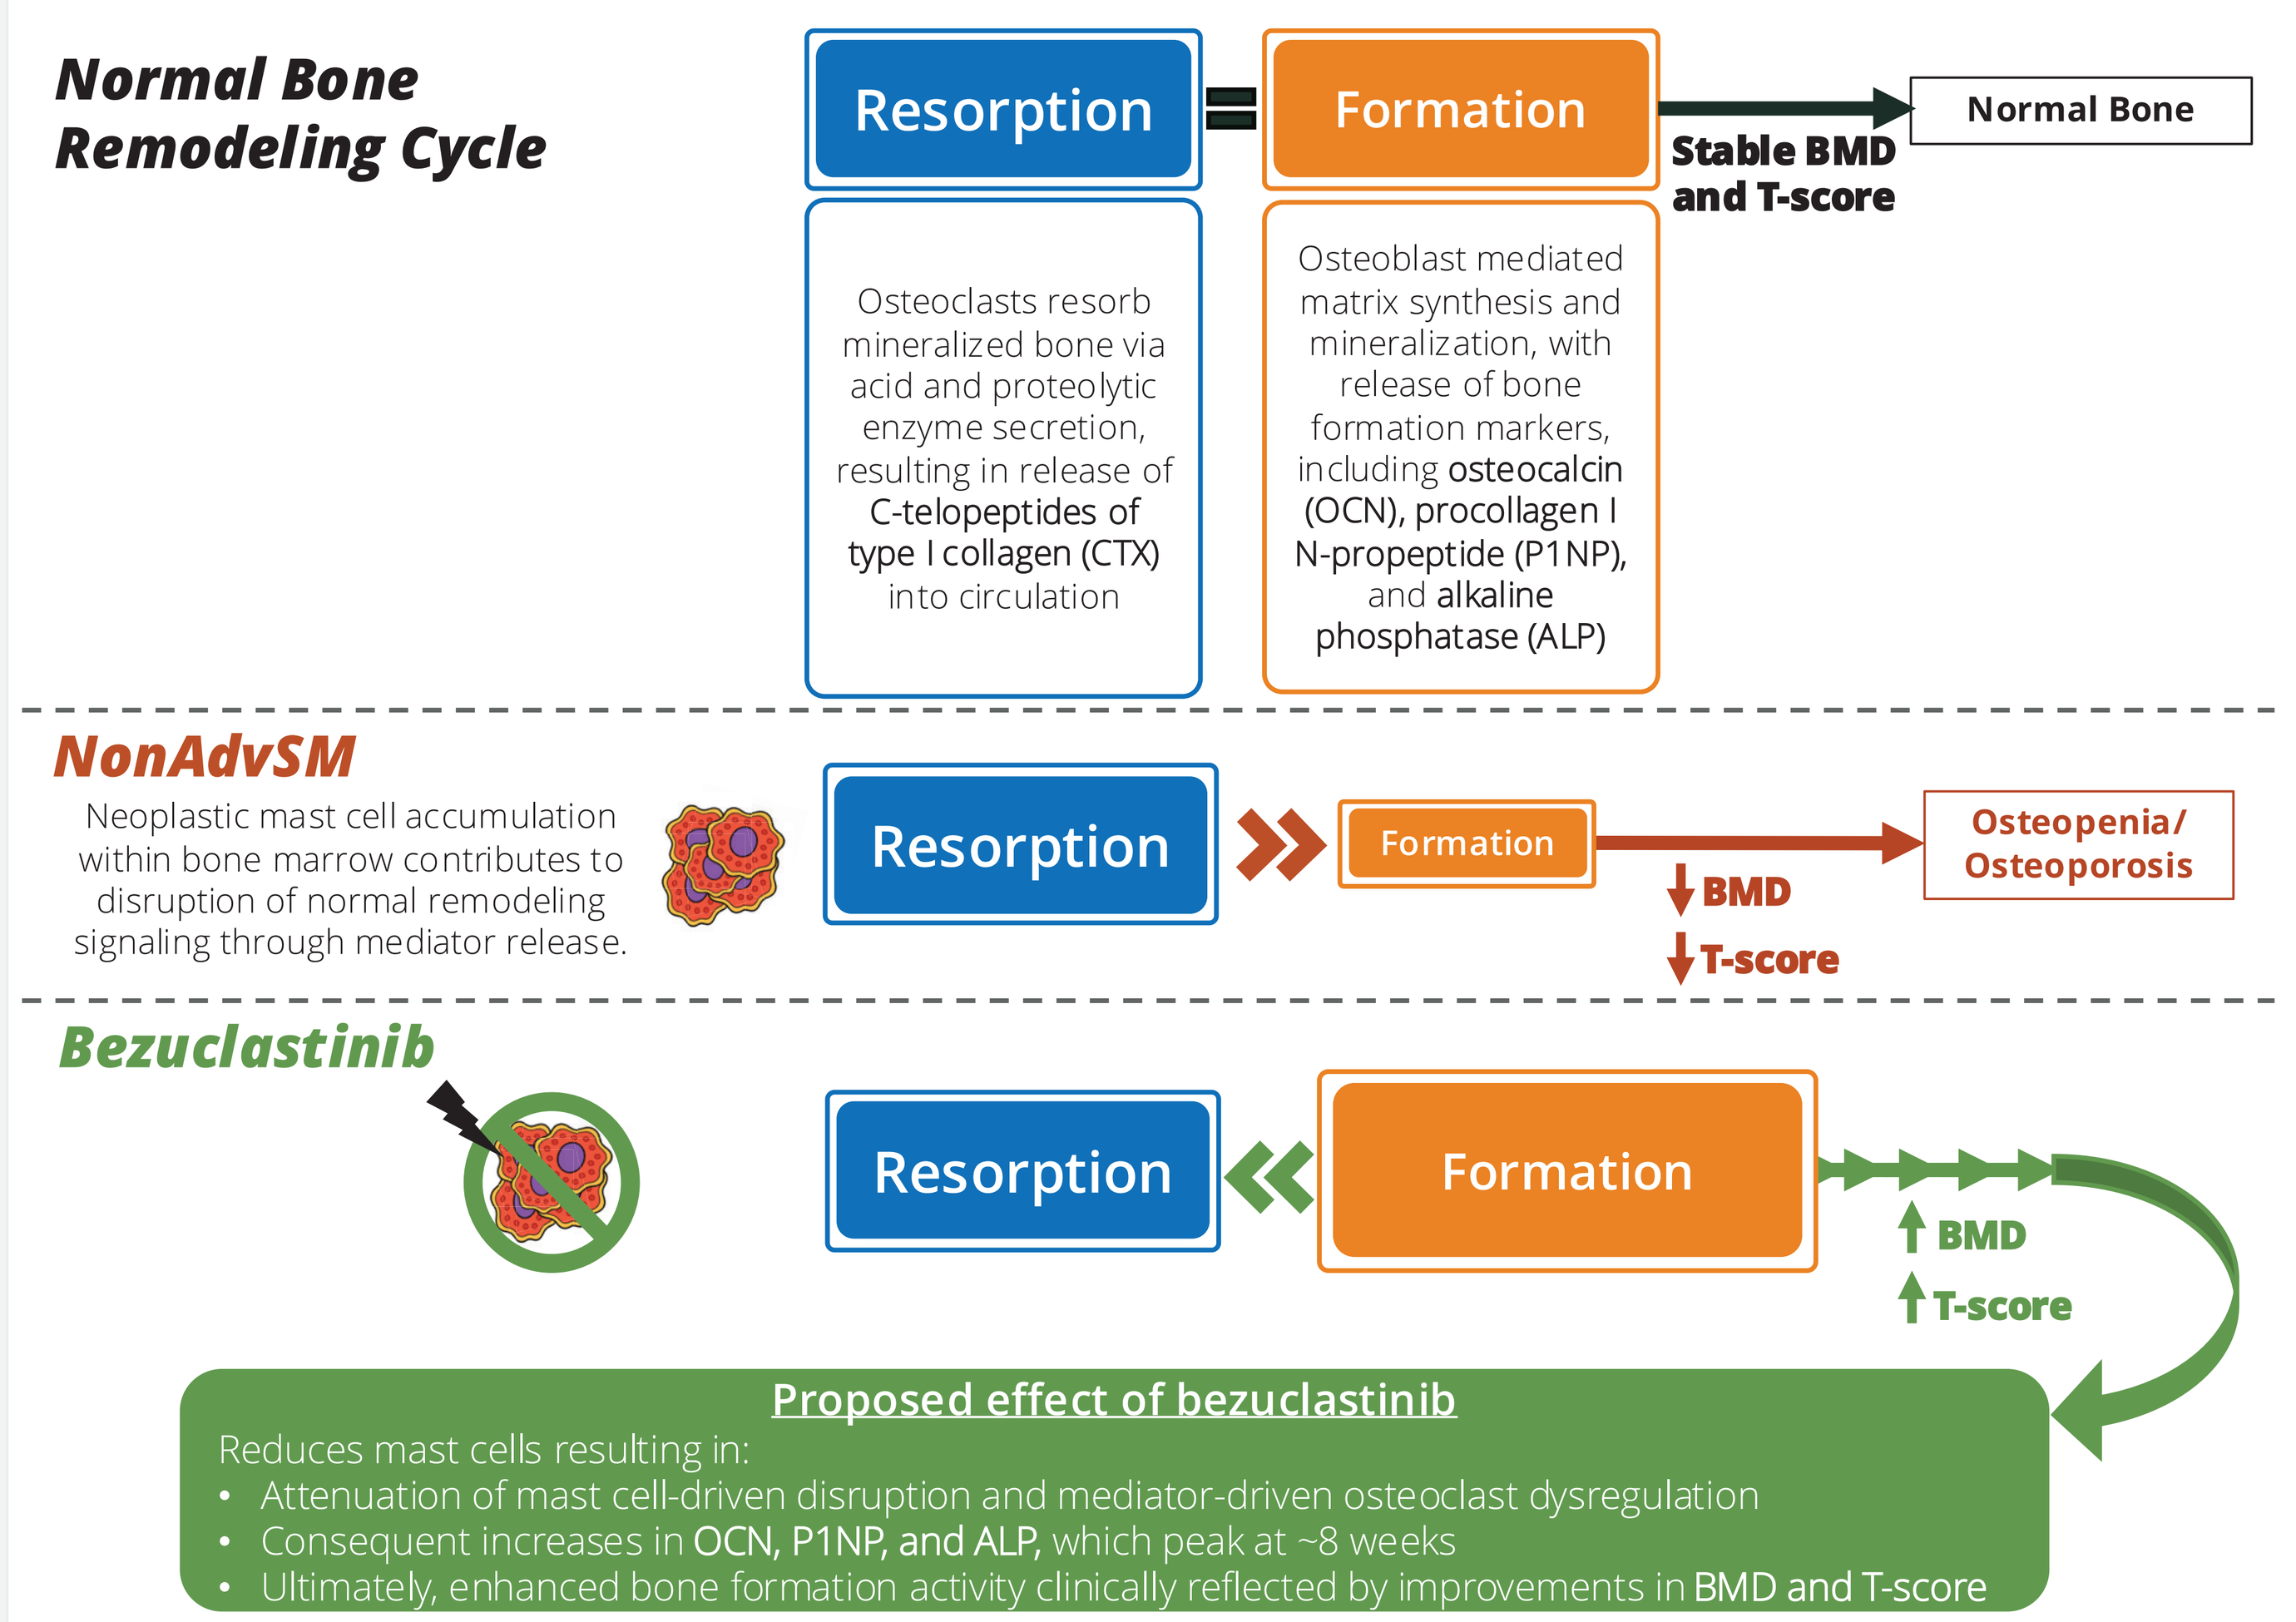
Normal Bone Remodeling Cycle
Resorption
Formation
Normal Bone
Stable BMD and T-score
Osteoclasts resorb mineralized bone via acid and proteolytic enzyme secretion, resulting in release of C-telopeptides of type I collagen (CTX) into circulation
Osteoblast mediated matrix synthesis and mineralization, with release of bone formation markers, including osteocalcin (OCN), procollagen I N-propeptide (P1NP), and alkaline phosphatase (ALP)
NonAdvSM
Neoplastic mast cell accumulation within bone marrow contributes to disruption of normal remodeling signaling through mediator release.
Resorption
Formation
↓BMD
↓T-score
Osteopenia/ Osteoporosis
Bezuclastinib
Resorption
Formation
↑BMD
↑T-score
Proposed effect of bezuclastinib
Reduces mast cells resulting in:
• Attenuation of mast cell-driven disruption and mediator-driven osteoclast dysregulation
• Consequent increases in OCN, P1NP, and ALP, which peak at ~8 weeks
• Ultimately, enhanced bone formation activity clinically reflected by improvements in BMD and T-score

METHODS

Summit (NCT05186753): Pivotal phase 2, multicenter, randomized, double-blind, placebo-controlled study evaluating bezuclastinib in NonAdvSM

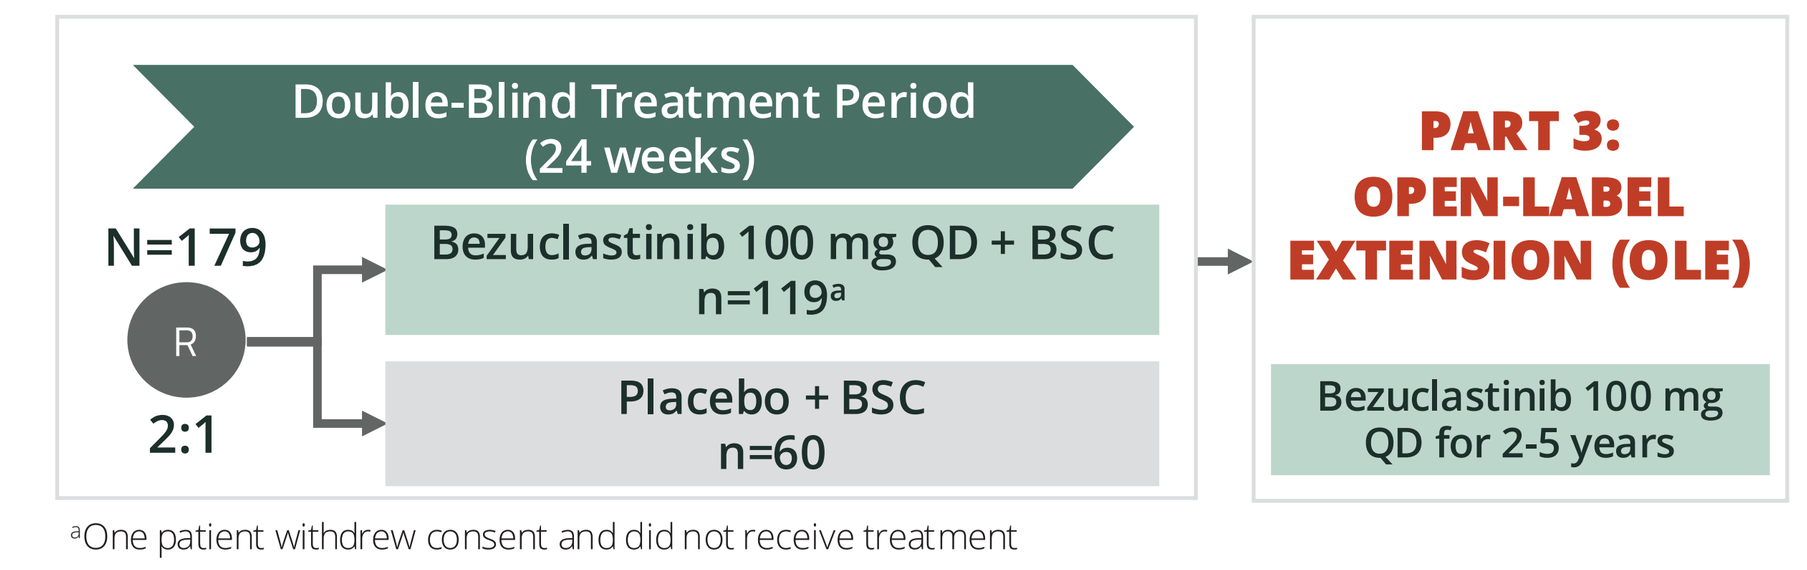
SUMMIT PART 2 | Primary Objective: Determine the efficacy of bezuclastinib
Double-Blind Treatment Period (24 weeks)
N=179
R
2:1
Bezuclastinib 100 mg QD + BSC n=119ᵃ
Placebo + BSC n=60
PART 3: OPEN-LABEL EXTENSION (OLE)
Bezuclastinib 100 mg QD for 2-5 years

ᵃOne patient withdrew consent and did not receive treatment

- Effect of bezuclastinib on bone was evaluated as exploratory assessments during Summit Part 2
 - — Bone formation markers, including serum P1NP and osteocalcin, were assessed
 - — Bone density was evaluated from optional dual-energy X-ray absorptiometry (DXA) scans

RESULTS

Bone health characteristics in patients with DXA scans

Parameter	Overall (N=179)	Bezuclastinib (N=119)	Placebo (N=60)
Patients with DXA scan, n (%)	79 (44.1%)	59 (49.6%)	20 (33.3%)
Normal BMD (T-score ≥ −1.0), n (%)	39 (49.4%)	27 (45.8%)	12 (60%)
Osteopenia, n (%)	31 (39.2%)	29 (49.2%)	2 (10%)
Osteoporosis, n (%)	9 (11.4%)	3 (5.1%)	6 (30%)

	Normal Bone Mineral Densityᵃ (N=39)	Osteopeniaᵃ (N=31)	Osteoporosisᵃ (N=9)
Age, years – median (range)	50 (24–73)	55 (29–71)	55 (36-66)
Female, n (%)	27 (69)	20 (65)	6 (67)
BMI, kg/m² – median (range)	31.0 (19.1–46.4)	27.7 (19.1–38.7)	27.3 (16.1-42.1)
Medical history of bone fracture, n (%)ᵇ	5 (13%)	7 (23%)	2 (22%)
Patients taking concomitant bone health medications, n (%)			
Calcium	10 (26%)	11 (35%)	4 (44)
Vitamin D	22 (56%)	19 (61%)	8 (89)
Bisphosphonates	0 (0)	4 (13%)	2 (22)
Denosumab	1 (3%)	1 (3%)	2 (22)
T-score on DXA scan, mean (SD) [n]			
Lumbar spine	−0.04 (0.87) [34]	−1.35 (0.93) [27]	-2.94 (0.29) [9]
Femoral neck	0.08 (0.83) [27]	−1.07 (0.71) [24]	-1.51 (0.67) [5]
Pelvis	0.47 (0.88) [19]	−0.72 (0.73) [17]	-0.57 (1.18) [3]

- A significant proportion of patients enrolled in Summit with DXA scans had compromised bone health, including 39% with osteopenia and 11% with osteoporosis
- Characteristics were generally balanced between placebo and bezuclastinib arms

ᵃBone mineral density categories are based on lowest T-score (Normal ≥ −1.0; Osteopenia −2.5 to < −1.0; Osteoporosis < −2.5).
ᵇMedical history depends on what is reported and may be incomplete regarding fracture history.

Patient cases demonstrate improvement in T-score as early as 24 weeks after bezuclastinib treatment

Patient Case 1

- 43-year-old male with ISM and low BMD, including lumbar spine T-score in the Grade 1 osteoporotic range and Z-score below age-expected norms
- No prior ISM-directed therapy; received concomitant vitamin D3 and calcium citrate throughout study
- Randomized to bezuclastinib 100 mg on 7 Oct 2024

	BMD		T-score		Z-score	
	BL	Wk 24	BL	Wk 24	BL	Wk 24
Neck	0.93	0.96	0	0.2	0.6	0.8
Spine	0.83	0.90	-2.4	-1.7	-2.2	-1.5
Pelvis	0.98	1.0	-0.3	-0.2	-0.1	0

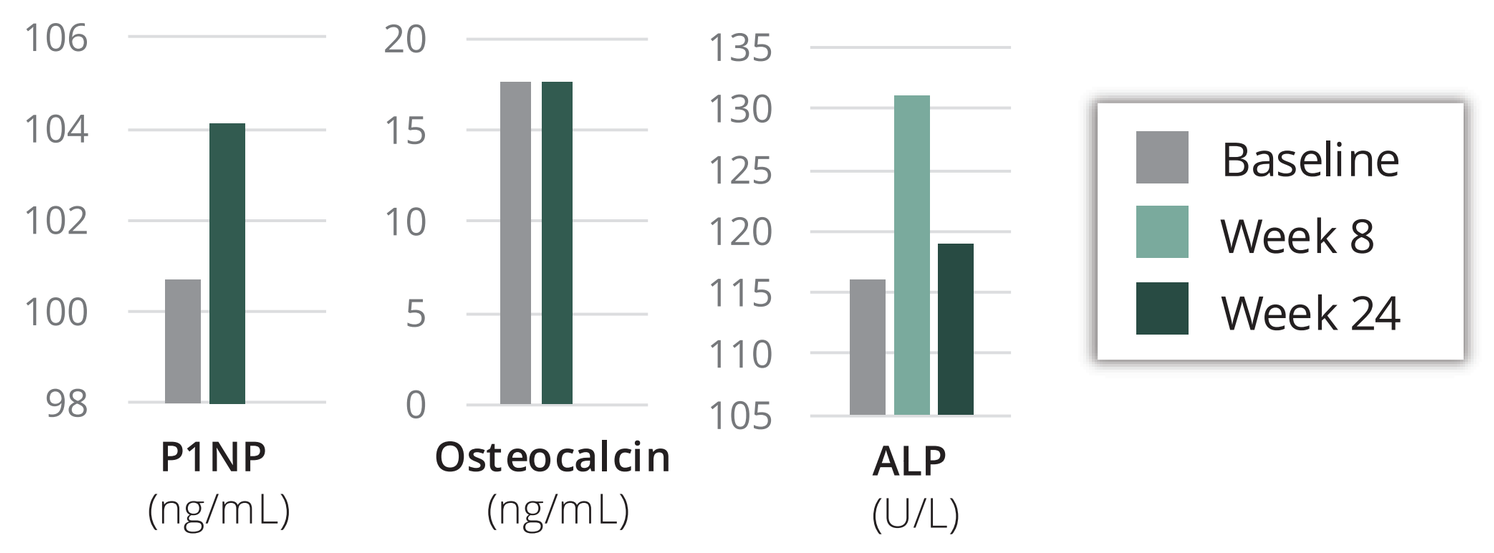
P1NP (ng/mL)
Osteocalcin (ng/mL)
ALP (U/L)
Baseline
Week 8
Week 24

Biomarkers	BL	Wk 24	Wk 24 %CFB
MC Burden	10	2	-80
KIT	0.07	0	-100
Tryptase	22.5	3.6	-84

Early rise or stabilization of bone formation markers, coupled with measurable BMD gains by Week 24 of bezuclastinib treatment, collectively suggest an emerging bone-anabolic treatment effect in this patient.

Patient Case 2

- 63-year-old male with ISM and baseline Grade 1 osteopenia, with mild neck and low back pain
- No prior ISM-directed or bone-targeted therapies
- Randomized to bezuclastinib 100 mg on 4 Nov 2024

	BMD		T-score		Z-score	
	BL	Wk 24	BL	Wk 24	BL	Wk 24
Neck	1.05	ND	-0.2	1.2	1.1	0
Spine	0.96	ND	-2.2	0.1	-1.4	-0.7

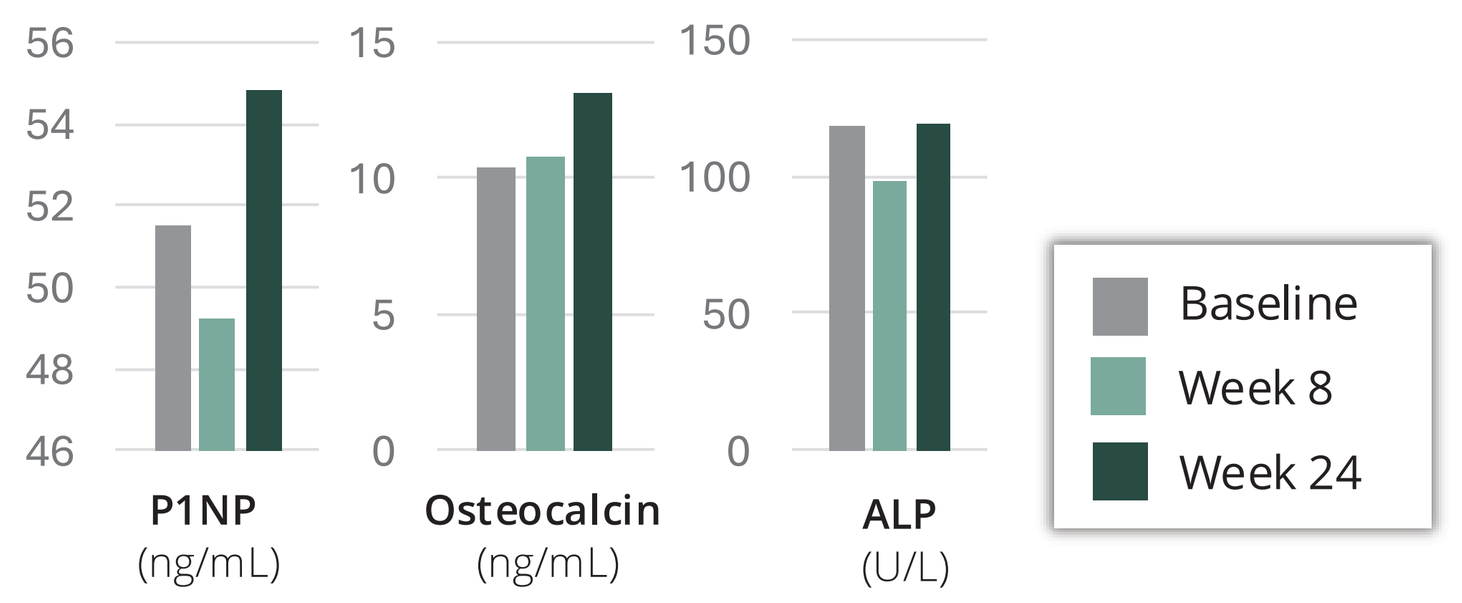
P1NP (ng/mL)
Osteocalcin (ng/mL)
ALP (U/L)
Baseline
Week 8
Week 24

Biomarkers	BL	Wk 24	Wk 24 %CFB
MC Burden	10	2	-80
KIT	0.04	0	-100
Tryptase	20.6	5.7	-72.33

Marked improvements in both lumbar spine and femoral neck T-scores, and supportive improvement in spine Z-score, by Week 24 of bezuclastinib treatment suggest an early, favorable effect on bone density, particularly at the lumbar spine, which is a clinically relevant site for fracture risk in patients with ISM.

Bezuclastinib improves bone formation markers, bone mineral density, and T-scores by Week 24, demonstrating evidence of early bone-anabolic response

Bone formation markers increase 8 weeks after initiating bezuclastinib, indicating that treatment has activated the bone remodeling process

- Osteocalcin, P1NP and ALP all show increases at 8 weeks in bezu patients compared to no increase in placebo patients
 - — This increase at 8 weeks corresponds to the bone formation phase, which should occur 6-12 weeks following activation of the bone remodeling process

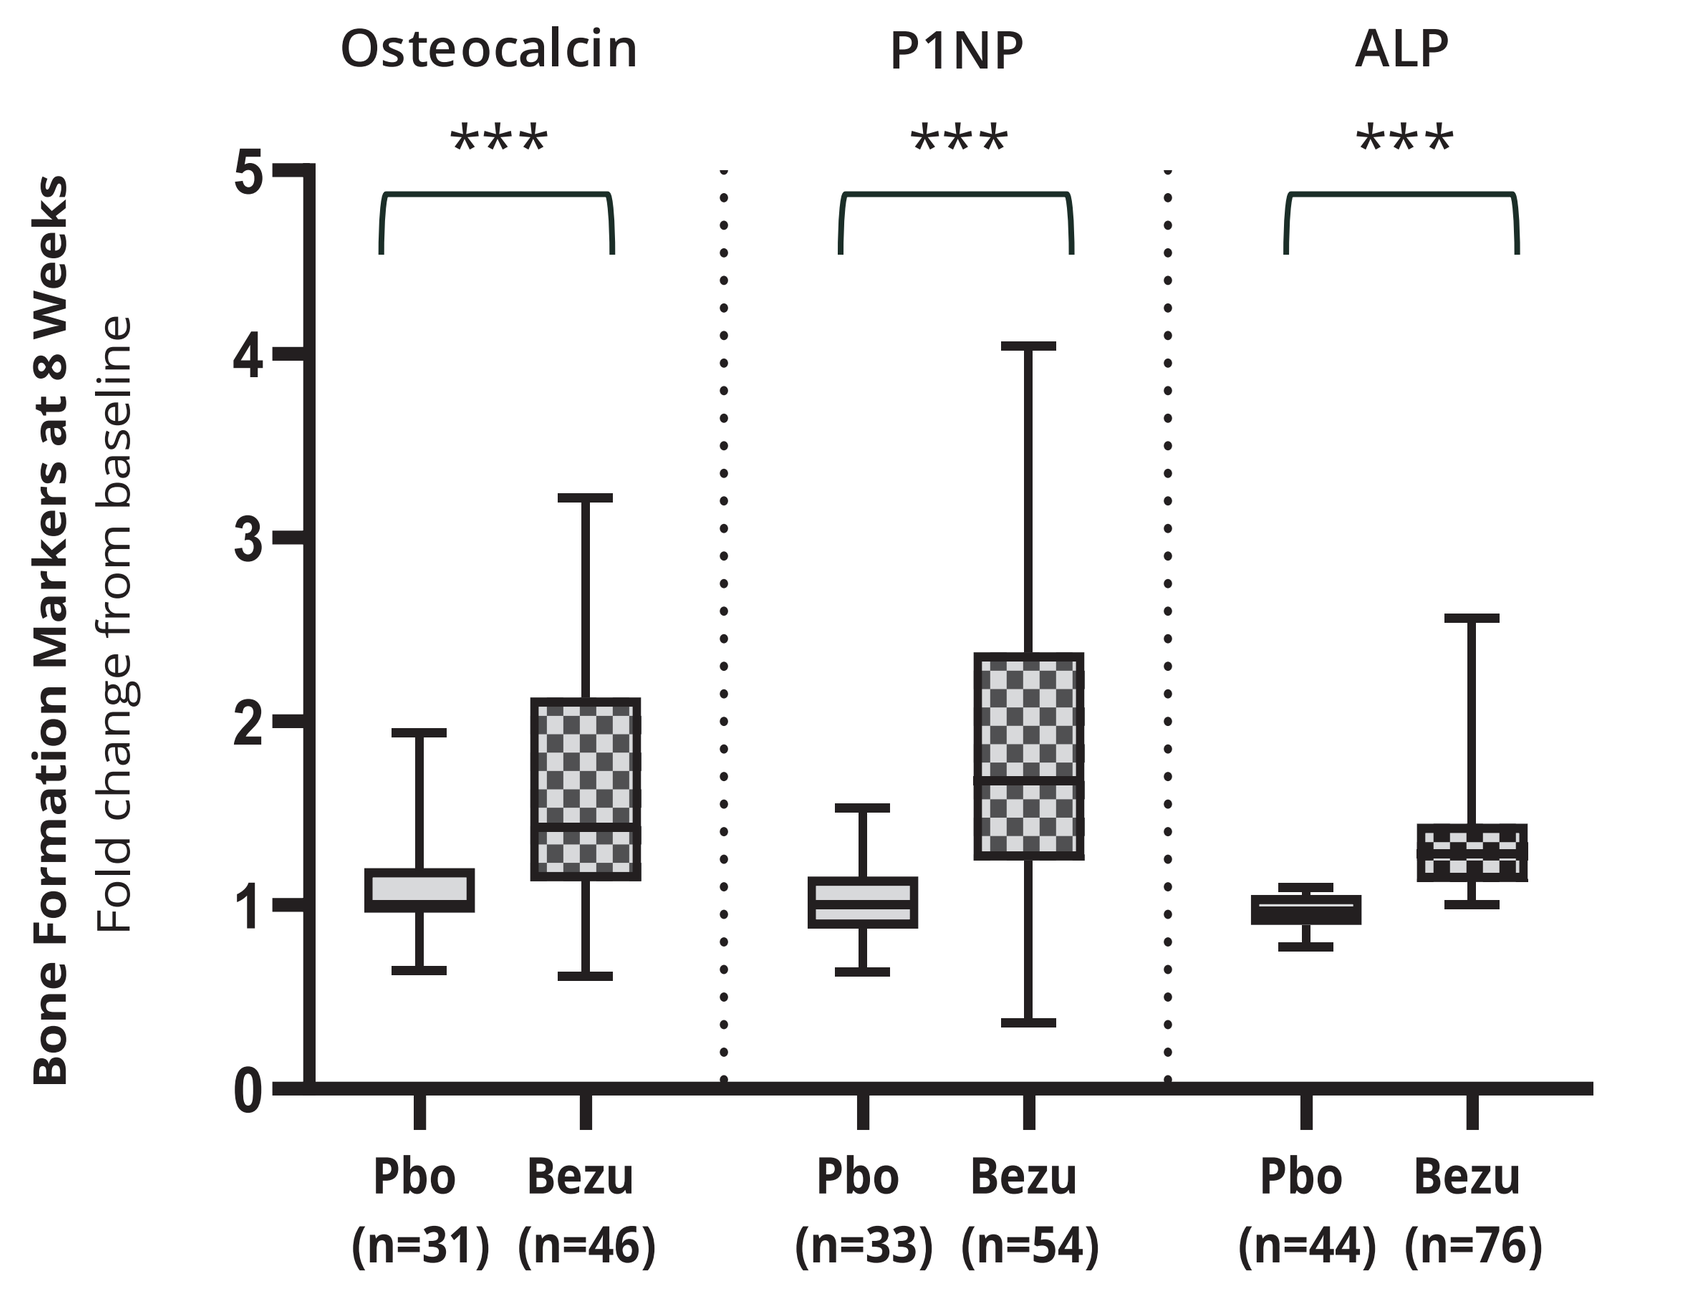
Bone Formation Markers at 8 Weeks
Fold change from baseline
Osteocalcin
P1NP
ALP

Pbo (n=31)
Bezu (n=46)
Pbo (n=33)
Bezu (n=54)
Pbo (n=44)
Bezu (n=76)

Significant improvement in bone mineral density at 24 weeks, demonstrating clinical impact of bone remodeling after treatment

Comparison to placeboᵃ

- Mean lumbar spine BMD increased at Week 24 with bezuclastinib, whereas patients on placebo experienced a decline over the same period

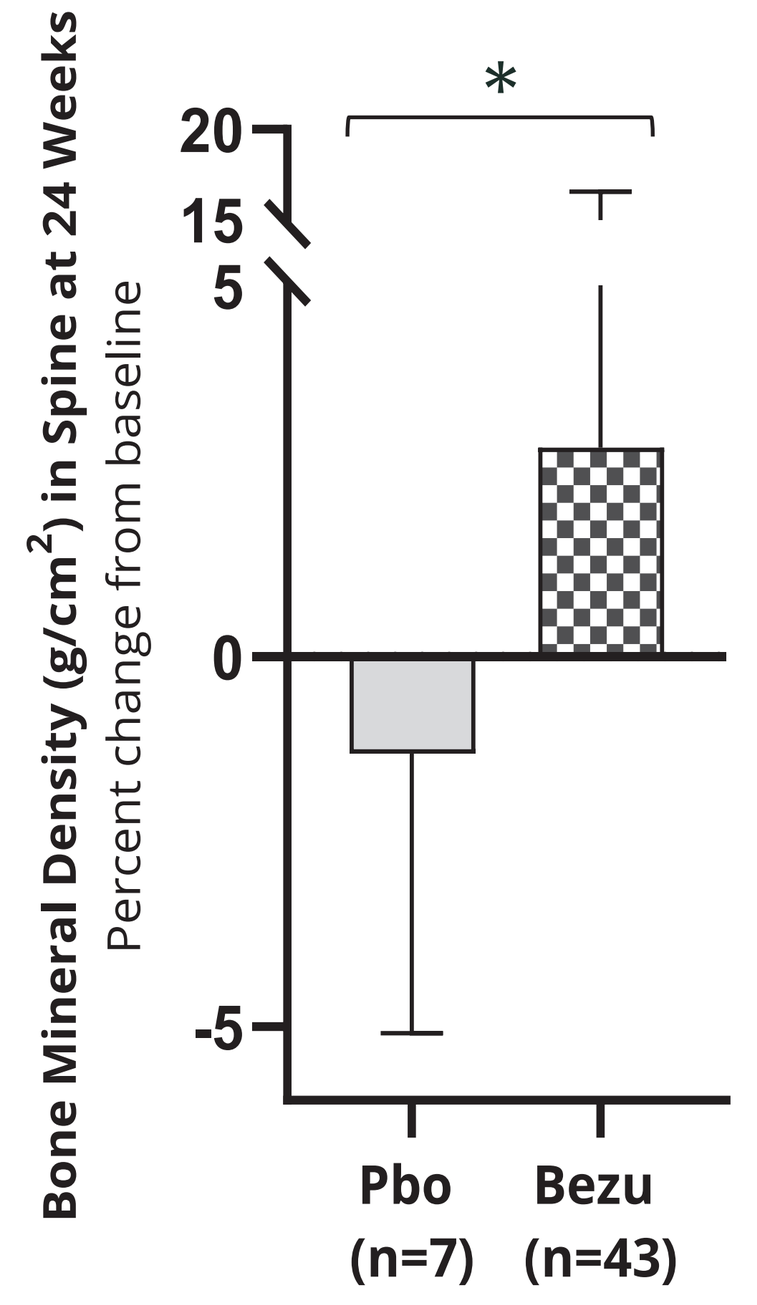
Bone Mineral Density (g/cm²) in Spine at 24 Weeks
Percent change from baseline
*
Pbo (n=7)
Bezu (n=43)

Comparison to baseline in patients receiving bezuclastinibᵃ

- Improvement in mean BMD were observed across baseline bone health categories in bezuclastinib-treated patients, indicating a treatment-related gain in bone density

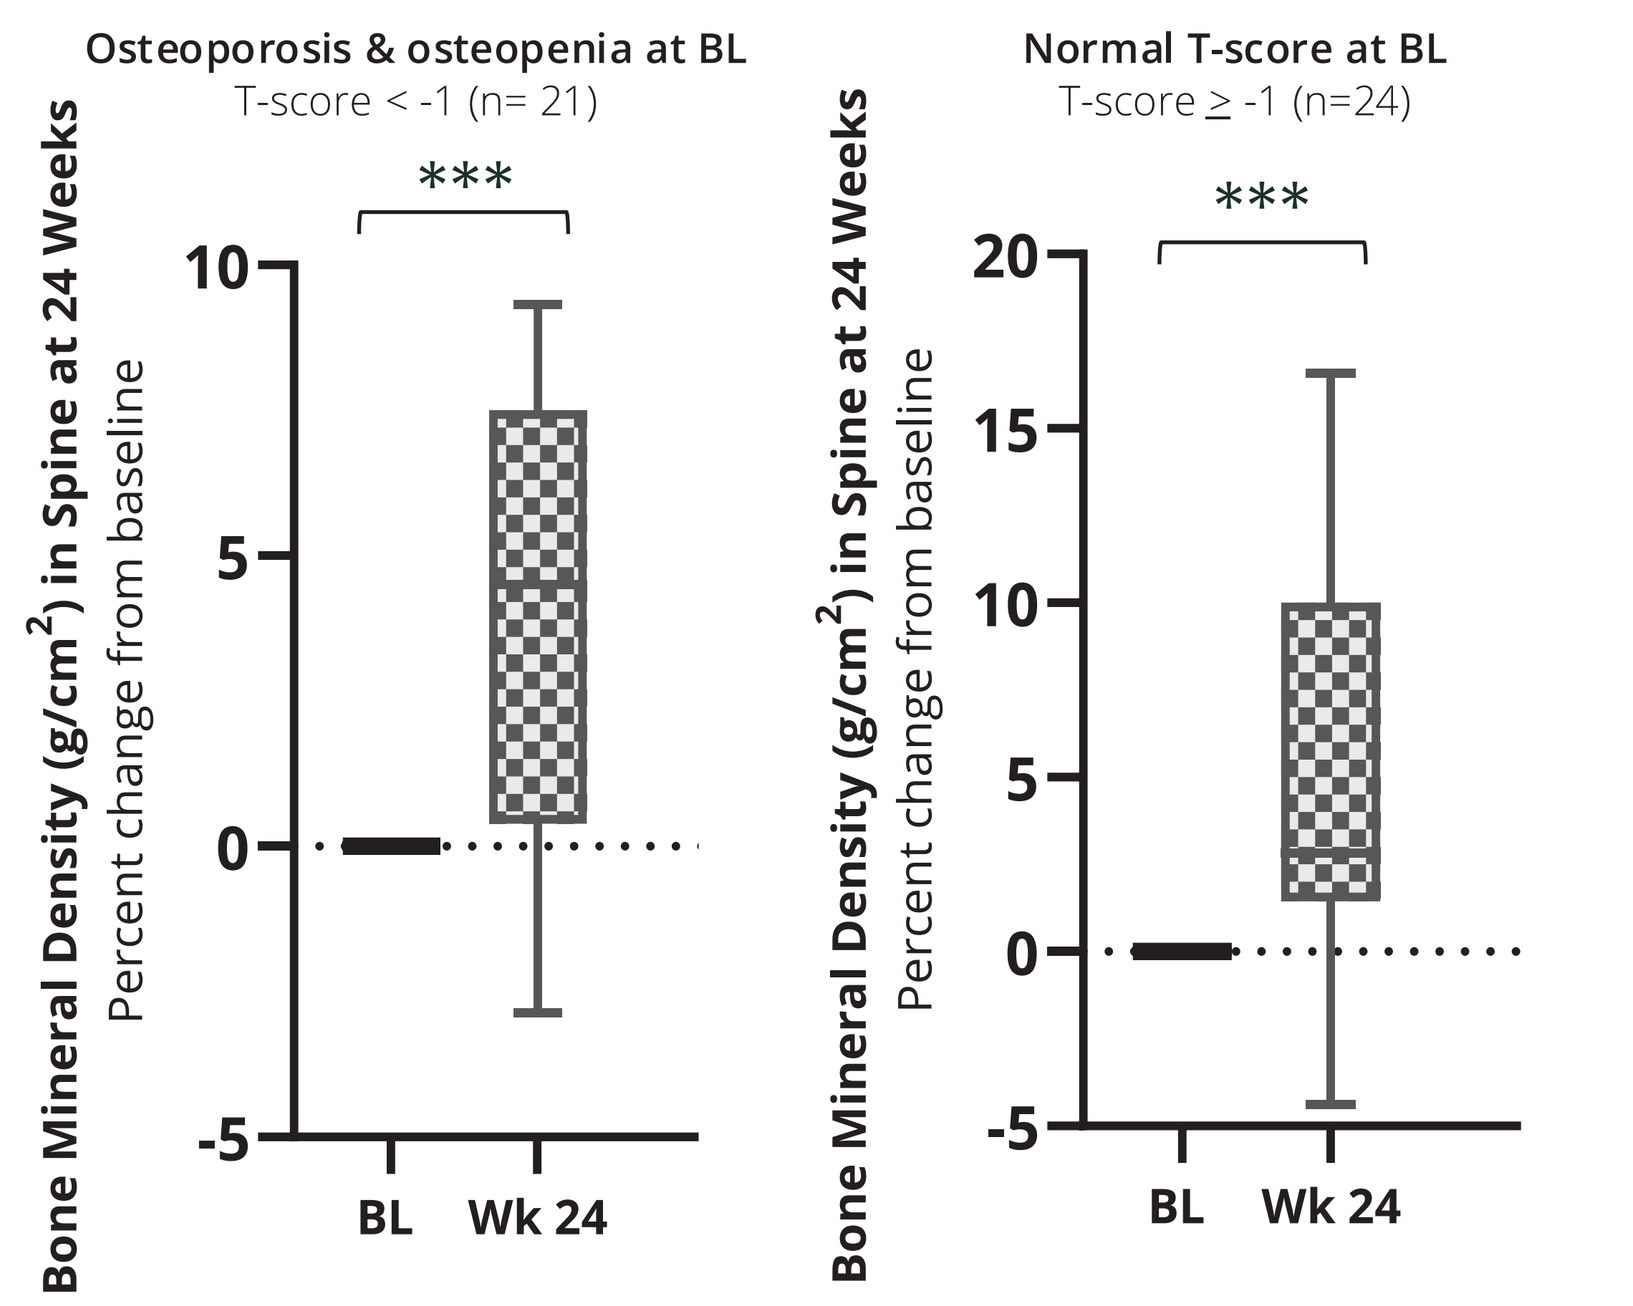
Osteoporosis & osteopenia at BL
T-score < -1 (n= 21)
Normal T-score at BL
T-score ≥ -1 (n=24)
Bone Mineral Density (g/cm²) in Spine at 24 Weeks
Percent change from baseline

BL
Wk 24

Significant improvement in spine T-scores at 24 weeks, demonstrating the clinical impact of bone remodeling after treatment initiation

Comparison to baseline in patients receiving bezuclastinibᵃ

- Improvements in mean BMD corroborated by improvements in mean spine T-score compared with baseline
- T-score improvements observed across baseline bone health categories, including:
 - — Patients with baseline osteopenia or osteoporosis (T-score < –1)
 - — Patients with normal baseline T-scores (≥ –1)

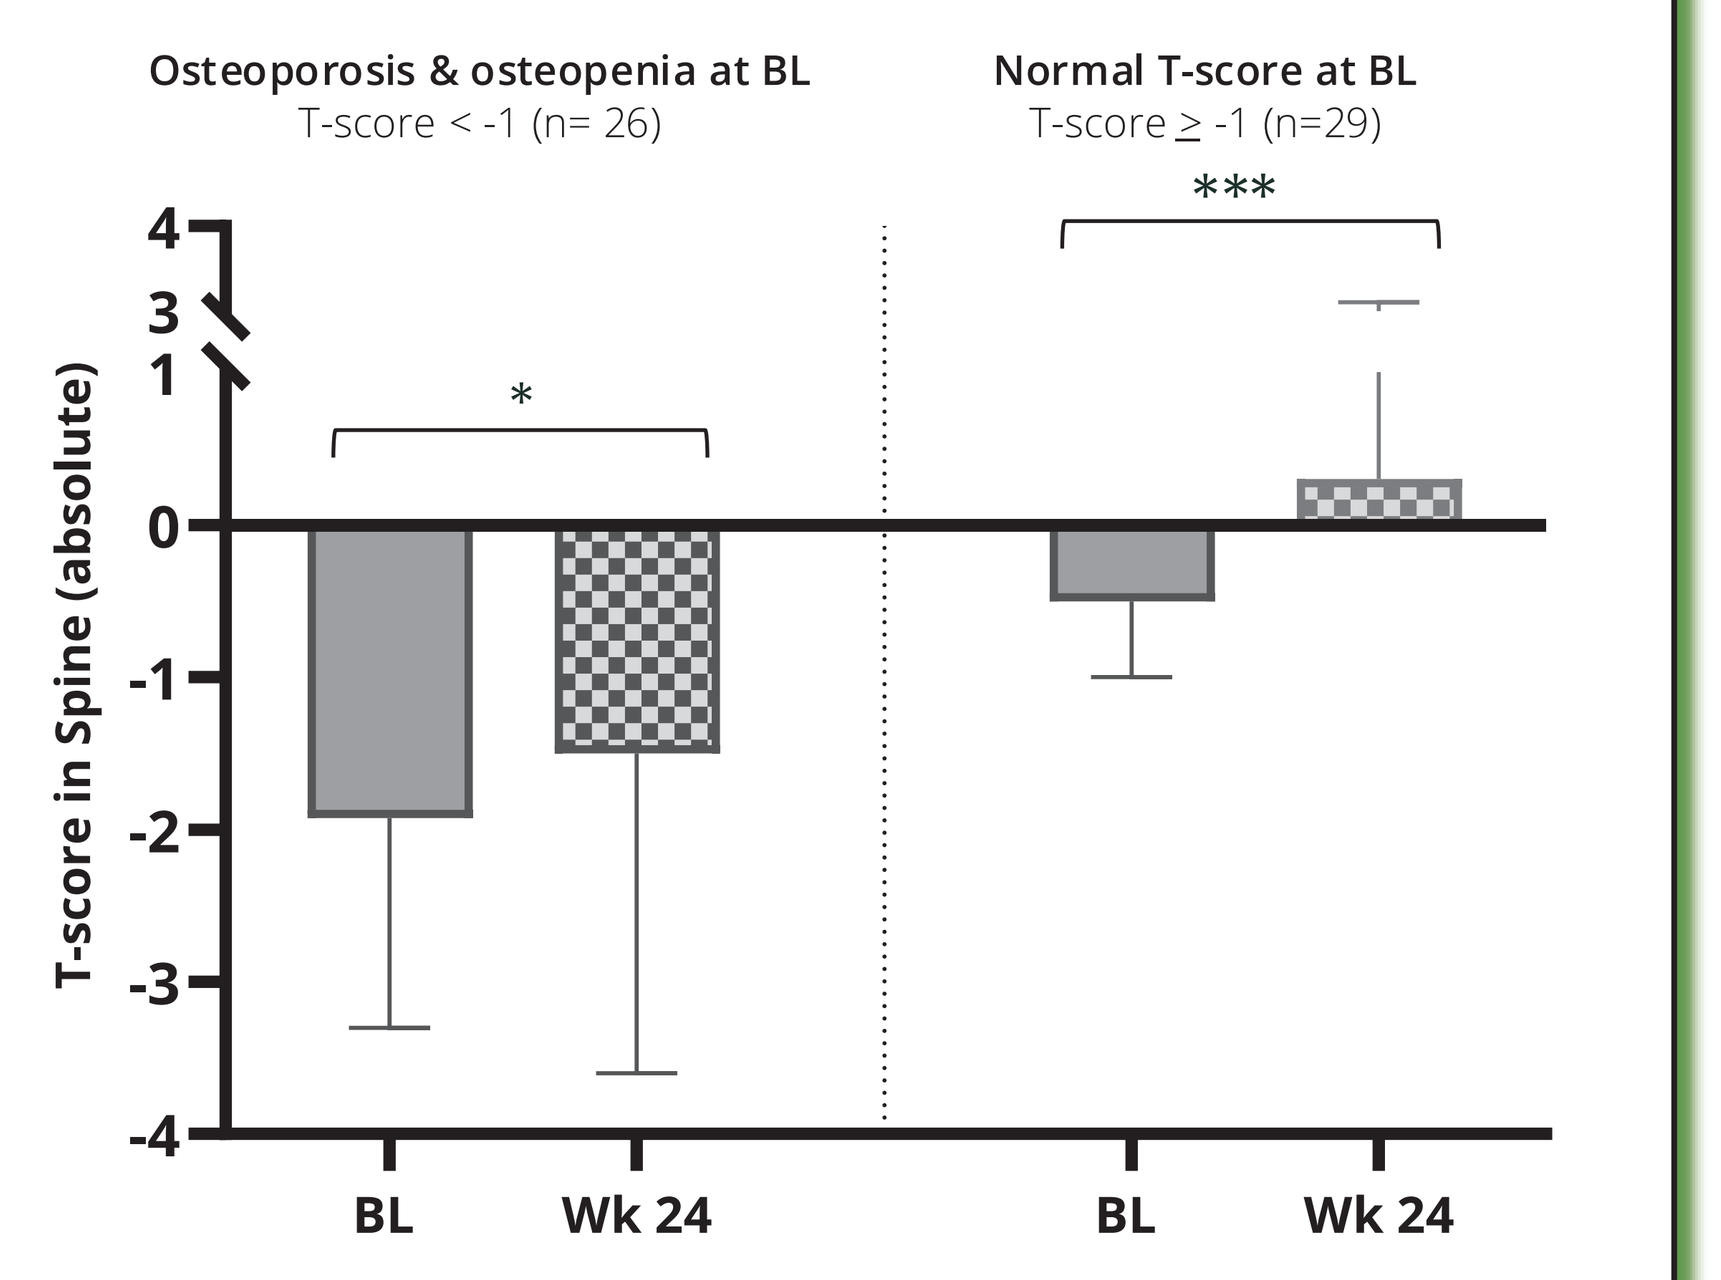
Osteoporosis & osteopenia at BL
T-score < -1 (n= 26)
Normal T-score at BL
T-score ≥ -1 (n=29)
T-score in Spine (absolute)
*

BL
Wk 24

T-test: Mann-Whitney test, unpaired, nonparametric test, 2-tailed
* = p<0.01; *** = p<0.0001
ᵃAmong patients with available DXA assessments
T-score: Normal ≥ −1.0, Osteopenia −2.5 to < −1.0, Osteoporosis < −2.5

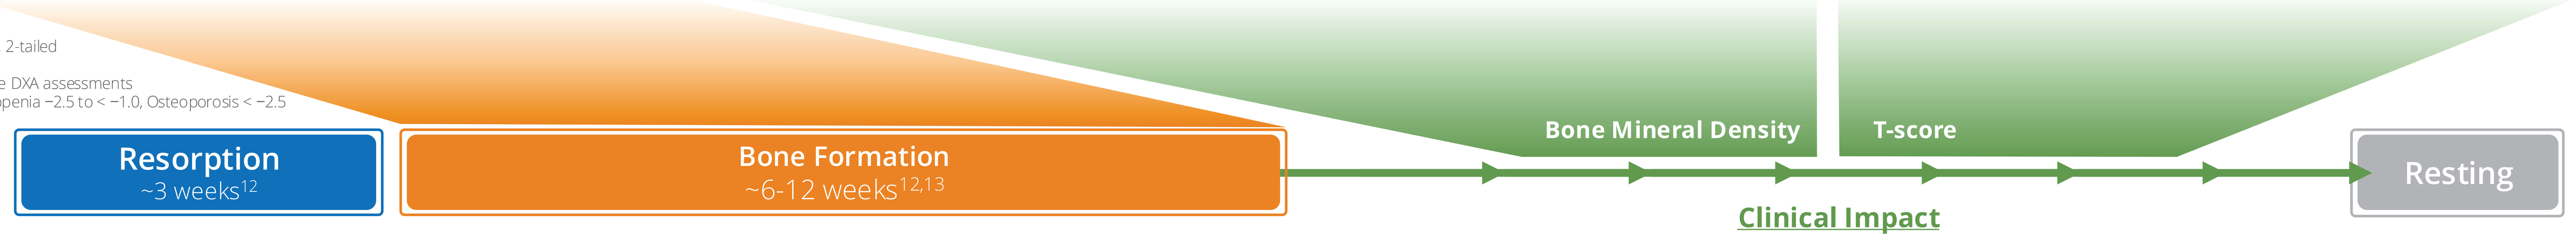
Resorption
~3 weeks[12]
Bone Formation
~6-12 weeks[12,13]
Bone Mineral Density
T-score
Resting
Clinical Impact

CONCLUSIONS

Evidence of disease modification with bezuclastinib based on improved bone formation markers, BMD, and T-scores by week 24

- A substantial proportion of Summit patients with DXA assessments showed baseline bone involvement, with 39% presenting with osteopenia and 11% with osteoporosis
 - — This distribution reflects abnormal bone loss commonly observed in patients with NonAdvSM and highlights the relevance of evaluating bone density and bone remodeling outcomes in this population
- Bezuclastinib treatment was associated with early activation of bone remodeling, reflected by increases in bone formation markers (osteocalcin, P1NP, ALP) at 8 weeks, peaking during the expected bone formation phase (6–12 weeks)
- Clinically meaningful improvements in BMD were observed at Week 24, particularly at the lumbar spine, a key fracture-risk site in patients with NonAdvSM
 - — Patients receiving bezuclastinib demonstrated increased spine BMD at Week 24, while those receiving placebo showed no improvement or a decrease
 - — BMD gains were seen regardless of baseline bone health, including patients with normal T-scores (≥ –1) and those with baseline osteopenia or osteoporosis (T-score < –1)
- Case summaries across age groups (<50 and >50 years) reinforced a consistent pattern, suggesting an emerging anabolic effect on bone with bezuclastinib treatment in patients with NonAdvSM:
 - — Early increases in bone formation markers,
 - — Measurable improvements in spine BMD
 - — Favorable shifts in T- and Z-scores

1. DeAngelo DJ, et al. Hemasphere. 2022; 6(suppl). 2. Guarnieri A, et al. Poster presented at: AACR 2022 Annual Meeting; April 8-13, 2022; Orleans, LA. 3. Rein LAM, et al. Oral presentation at: ASH 2025 Annual Meeting; December 6-7, 2025; Orlando, FL. 4. Ragipoglu D, et al. Front Immunol. 2020;11:163. 5. Letizia Mauro G, et al. Medicina (Kaunas). 2024;60(11):1752. 6. Degboé Y, et al. Curr Osteoporos Rep. 2025;23(1):10. 7. Rossini M, et al. Bone. 2011;49(4):880-885. 8. Rossini M, et al. Immunol Allergy Clin North Am. 2014;34(2):383-396. 9. Jankovski L, et al. Int J Mol Sci. 2025;26(12):5649. 10. Farmer I, Radia DH. Curr Hematol Malig Rep. 2024;19(5):197-207. 11. Das A, et al. *Crit Rev Oncol Hematol.* 2021;157:103186. 12. Agerbaek MO, et al. Bone Miner. 1991;12(2):101-112. 13. Eriksen EF, et al. *Metab Bone Dis Relat Res*. 1984;5(5):243-252.

ALP, alkaline phosphatase; BL, baseline; BM, bone marrow; BMD, bone mineral density; BMM, bone marrow mastocytosis; BSC, best supportive care; H1RA, histamine receptor type 1 antagonist; HR2A, histamine receptor type 2 antagonist; ISM, indolent SM; LTRA, leukotriene receptor antagonists; MC, mast cell; MS2D2, mastocytosis symptom severity daily diary; NonAdvSM, non-advanced systemic mastocytosis; OCN, osteocalcin; P1NP, procollagen type I N-terminal propeptide; PPI, proton pump inhibitors; QD, once daily; R, randomized; SSM, smoldering SM; TSS, total symptom score; VAF, variant allele frequency; Wk, week; WHO, World Health Organization.

American Academy of Allergy, Asthma & Immunology (AAAAI) Annual Meeting
February 27 – March 2, 2026 | Philadelphia, PA
For questions, please contact MedicalAffairs@cogentbio.com

The authors would like to thank the patients participating in this study, as well as their families, the investigators, patient advocates, research and operations staff. This study is sponsored by Cogent Biosciences.

The opinions and assertions expressed herein are those of the author(s) and do not reflect the official policy or position of the Uniformed Services University of the Health Sciences, the Department of War, or the Department of Health and Human Services.

To obtain a digital copy of this poster, please scan the Quick Response (QR) code. Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from AAAAI or the authors of this poster.