Identification of a Novel Brain-Penetrant EGFR-Sparing ErbB2 Inhibitor with Activity Against Oncogenic ErbB2 Mutations

Mark J Chicarelli, Tanna Bettendorf, Abiezer Blandon, Karyn Bouhana, Richard K. Brizendine, LouAnn Cable, Michelle Crow, Brad Fell, John Fischer, Jennifer Fulton, Anna Guarnieri, Madison Hillman, Ravi Jalluri, Amber Johnson, Hailey Knox, Keith A. Koch, Vijay Kumar, Cori A. Malinky, Rob Rieger, John Robinson, Lee M Stunkard, Francis Sullivan, John I Trujillo, Roy Turton, Shannon Winski, and Yeyun Zhou; Cogent Biosciences, Inc., Boulder, CO and Waltham, MA

San Antonio Breast Cancer Symposium®
December 5-9, 2023
Presentation Number: PO3-26-02

Cogent ErbB2 Inhibitor Opportunity
Target Product Profile

EGFR Sparing	Avoids EGFR related toxicities
Potent on Mutations	Retains high potency against prevalent ErbB2 mutations
Covalent	Provides a prolonged pharmacodynamic effect for maximum efficacy
Brain Penetrant	Provides coverage in the CNS to treat brain metastases
Selective	Selective for ErbB2 across the kinome, receptors, channels, and hERG
Combinable	Low DDI risk based on in vitro data, potential to combine with Enhertu and other agents

Background

Figure 1. HER Receptor Signaling Pathway

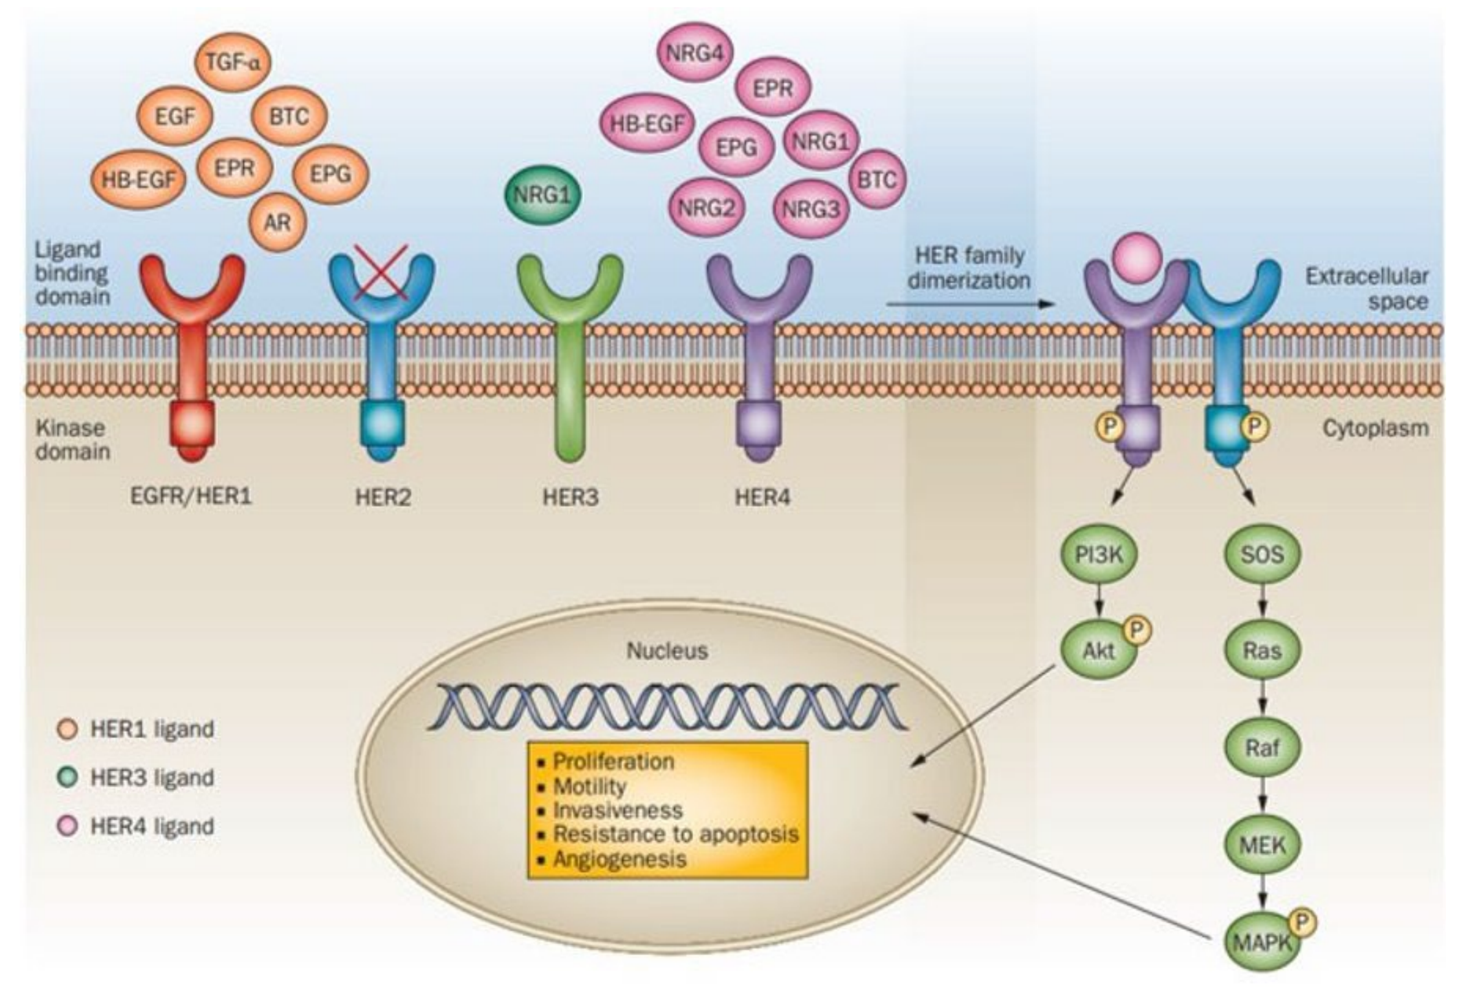
EGFR/HER1
HER2
HER3
HER4
HER family dimerization
Extracellular space
Cytoplasm
PI3K
Akt
SOS
Ras
Raf
MEK
MAPK
Nucleus
HER1 ligand
HER3 ligand
HER4 ligand

- ErbB2 is a receptor tyrosine kinase that belongs to a family of four receptors EGFR, ErbB2, ErbB3, and ErbB4, also known as HER1, 2, 3, 4[1]
- Receptor activation induces rapid dimerization with a marked preference for ErbB2 as a partner[2]
- Phosphorylation of the ErbB2 kinase domain activates PI3K/Akt and the Ras/Raf pathways which regulate cell growth, survival and differentiation[3]

Figure 2. Prevalence of Oncogenic Mutations of ErbB2[4,5]

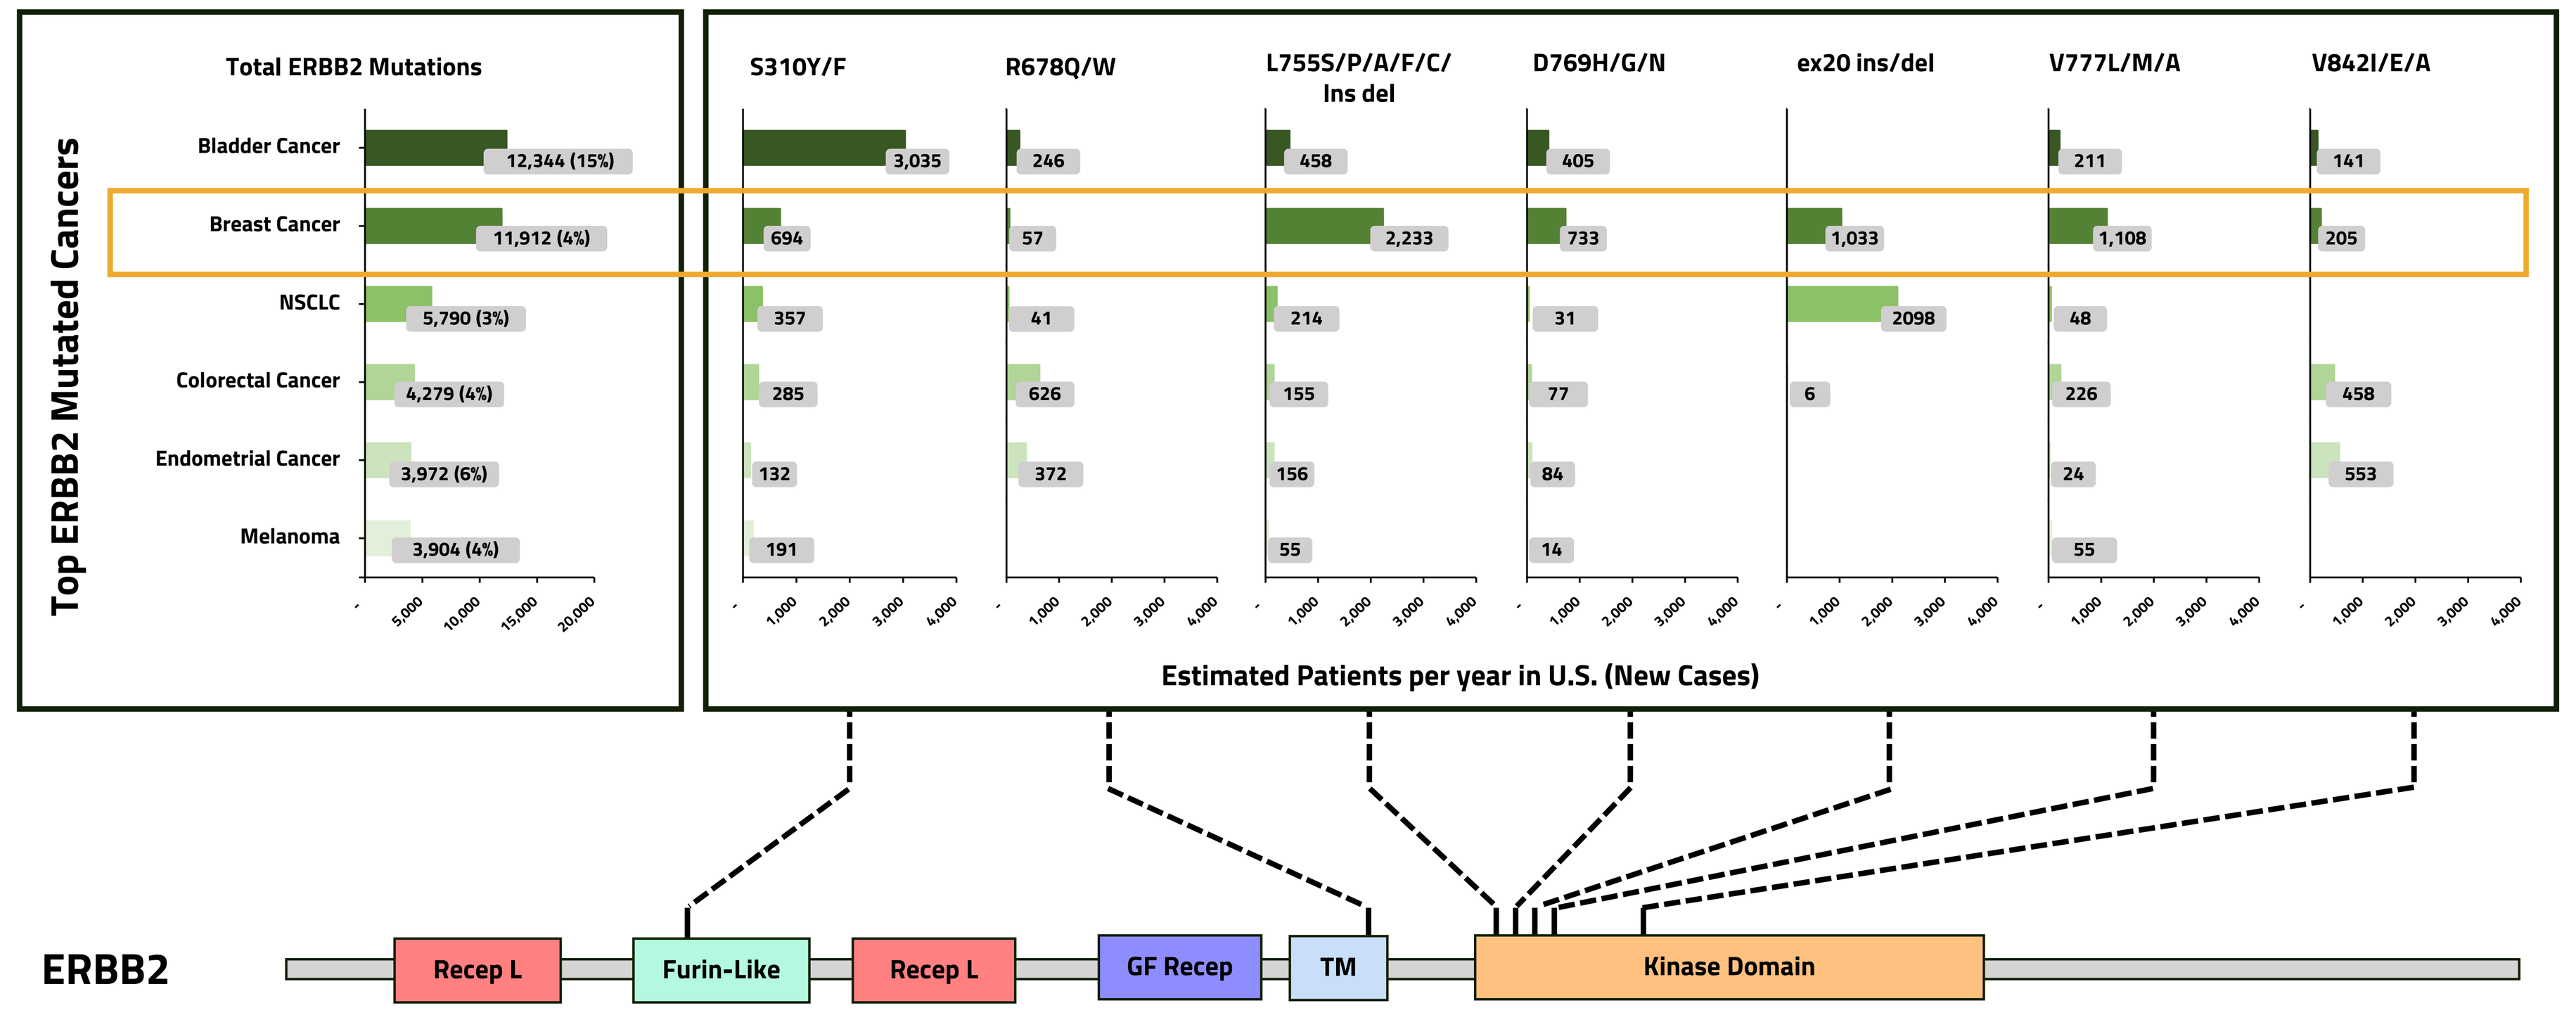
Total ERBB2 Mutations
Top ERBB2 Mutated Cancers
Bladder Cancer 12,344 (15%)
Breast Cancer 11,912 (4%)
NSCLC 5,790 (3%)
Colorectal Cancer 4,279 (4%)
Endometrial Cancer 3,972 (6%)
Melanoma 3,904 (4%)
S310Y/F
R678Q/W
L755S/P/A/F/C/ Ins del
D769H/G/N
ex20 ins/del
V777L/M/A
V842I/E/A
Estimated Patients per year in U.S. (New Cases)
ERBB2
Recep L
Furin-Like
Recep L
GF Recep
TM
Kinase Domain

- ErbB2 drives breast cancer growth through amplification or through genetic mutations leading to constitutive activation
- ErbB2 is mutated in roughly 4% of breast cancer patients with L755S being the most common alteration[5]
- Sequencing of tumors that have progressed post front-line breast cancer treatments have revealed ErbB2 mutations as mechanisms of resistance in metastatic breast cancer
- These mutations occur primarily in the furin-like and kinase domains

Results

Table 1. CGT4255 Shows Low nM Cellular Activity Against Oncogenic ErbB2 Mutations

	ErbB2 Cellular IC_{50} Inhibition of pErbB2				
	ErbB2 WT	L755S	YVMA	S310F	V842I
CGT4255	8 nM	9 nM	3 nM	7 nM	15 nM

Adjusted for FBS-binding

- Mechanistic cell assay in engineered lines measuring inhibition of pErbB2
- CGT4255 shows similar low nM potency across WT and mutant cell lines

Figure 3. CGT4255 is Selective Across the Kinome and 100X Selective Over EGFR

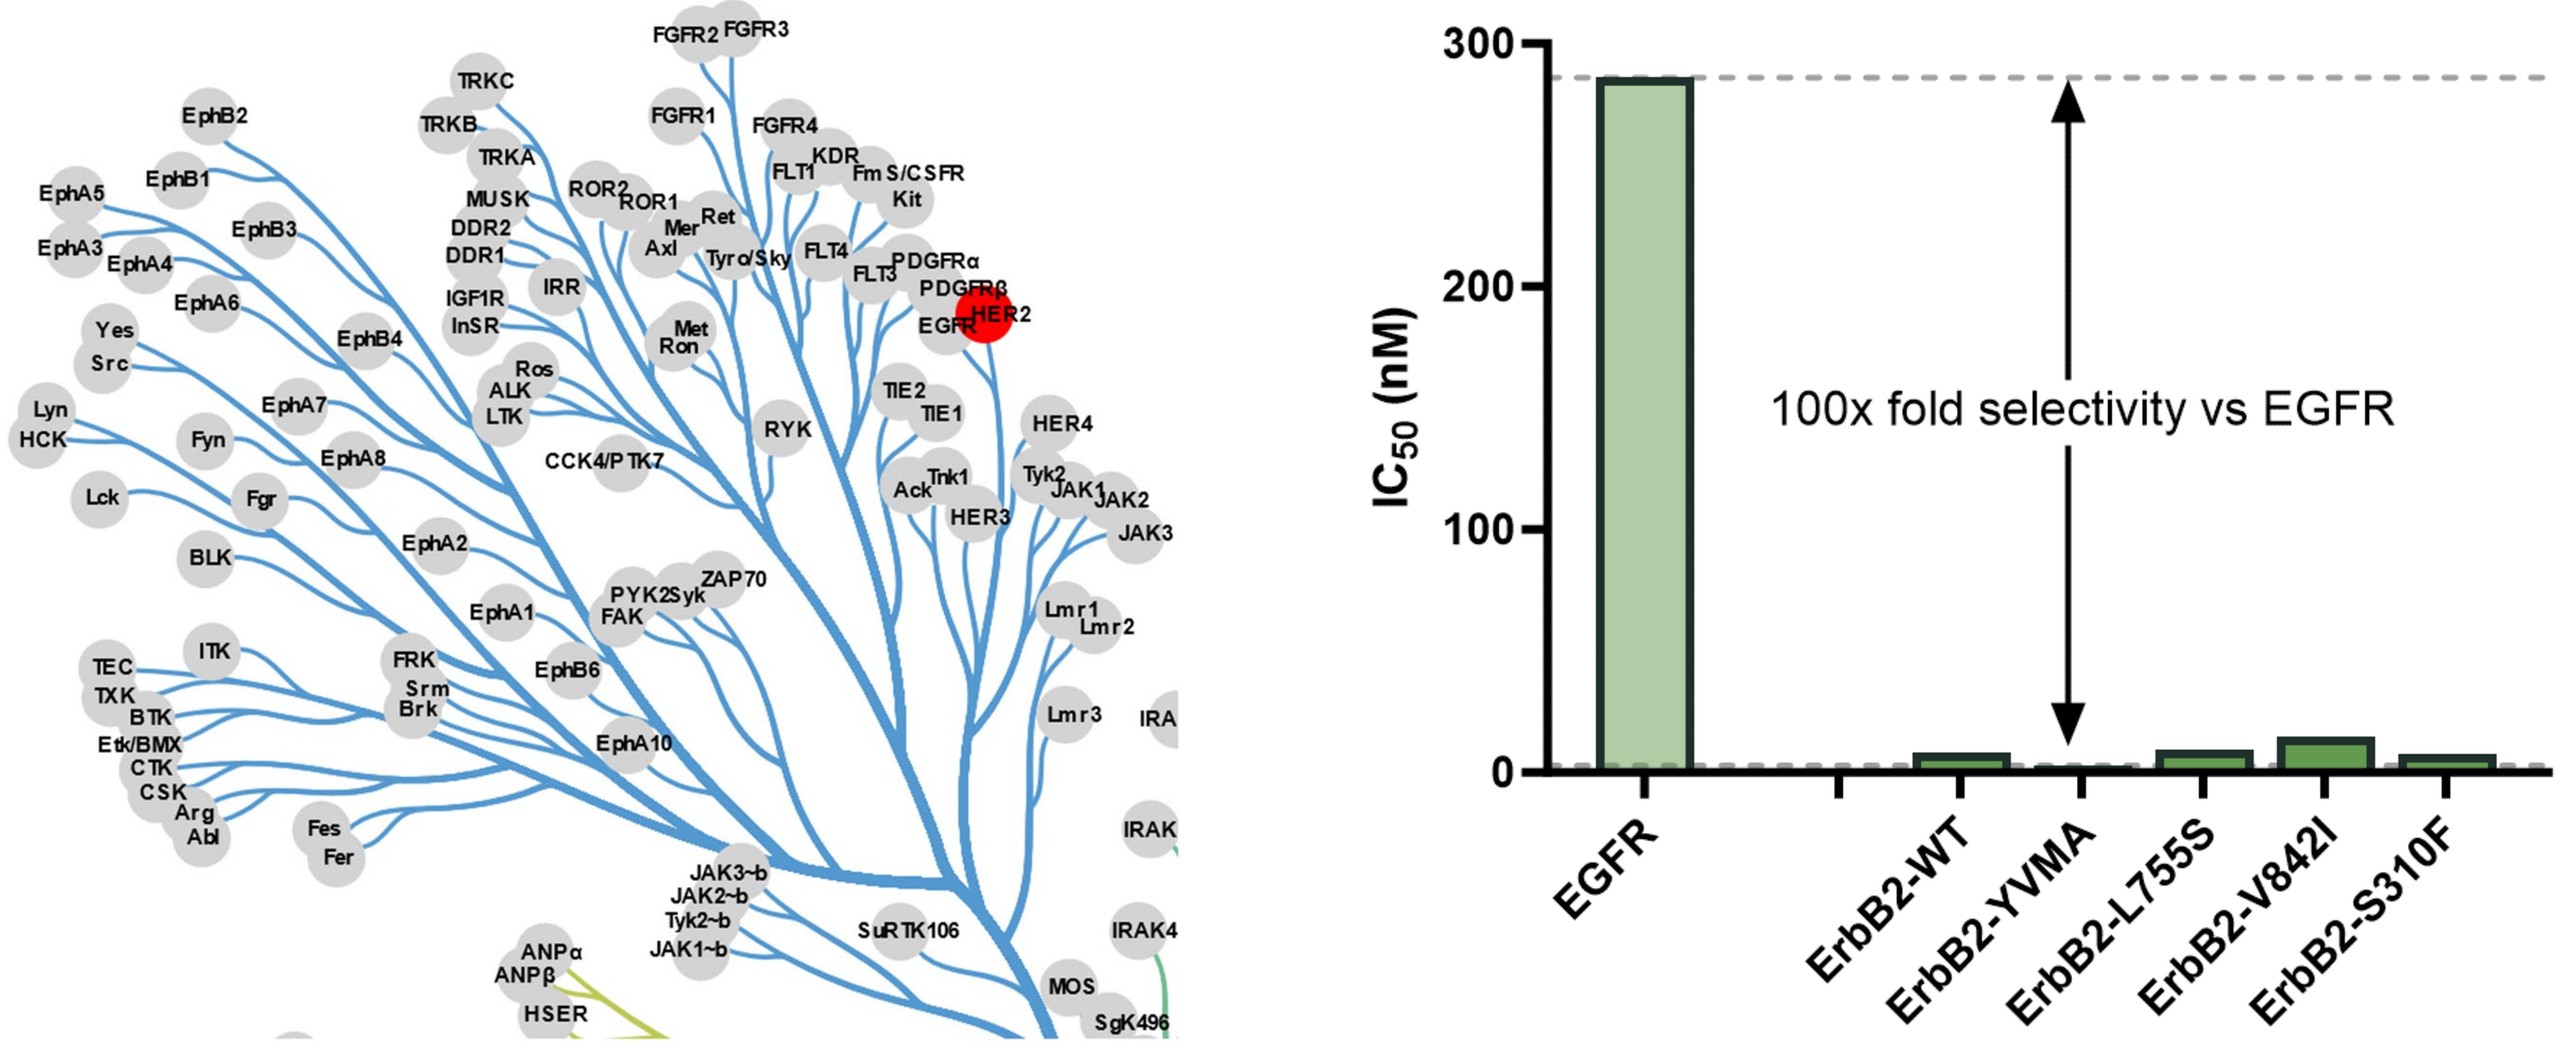
HER2
EGFR
100x fold selectivity vs EGFR
IC_{50} (nM)
EGFR
ErbB2-WT
ErbB2-YVMA
ErbB2-L755S
ErbB2-V842I
ErbB2-S310F

- CGT4255 was profiled at 10x the enzyme IC_{50} for WT ErbB2 against a panel of 371 kinases, ErbB2 was the only kinase that showed >30% target inhibition
- Mechanistic cellular assays show CGT4255 is 100-fold selective for ErbB2 YVMA over WT-EGFR

Table 2. CGT4255 has Long Half-Life in Whole Blood and Human Liver Cytosol Stability Assays

	Mouse	Rat	Dog	Cyno	Human
Whole Blood Stability, t1/2 min	637	903	1120	669	1089

	Poziotinib	Pyrotinib	ELVN-002[6]	CGT4255
Human Liver Cytosol Stability, t1/2 min	269	56	101	577

- CGT4255 shows greater than 600 min half-life across species in the whole blood stability assay, highlighting 18h half-life in human whole blood
- Compared to other covalent ErbB2i, CGT4255 has superior stability in a human liver cytosol stability assay

Figure 4. CGT4255 Shows Dose Escalating Mouse PK, With Best-in-Class Brain Exposure

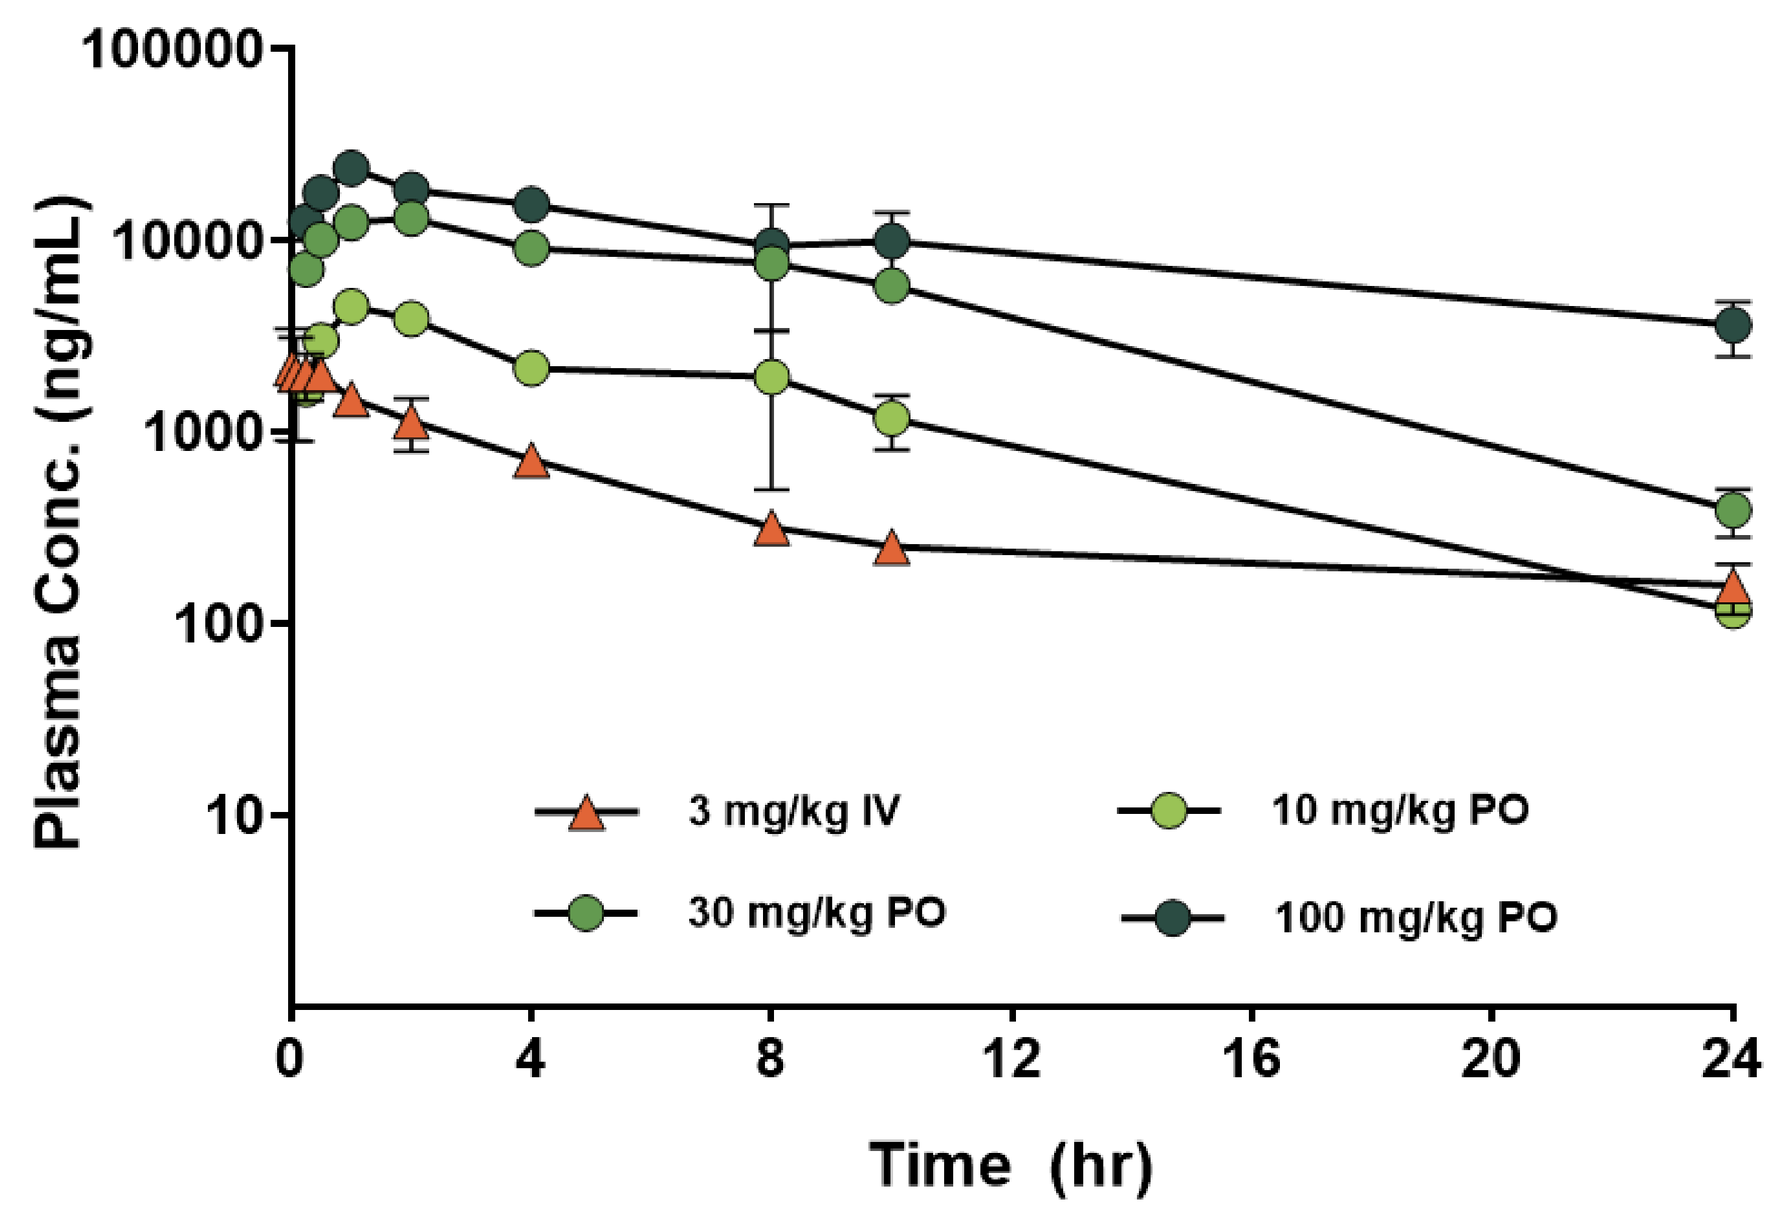
Plasma Conc. (ng/mL)
Time (hr)
3 mg/kg IV
10 mg/kg PO
30 mg/kg PO
100 mg/kg PO

PK @ 3 mg/kg IV	Result
CL (mL/min/kg)	3.5
VDss (L/kg)	3.9
PK @ 100 mg/kg PO	**Result**
t1/2 (hr)	6.8
F (%)	73
Kpuu @1h	0.80

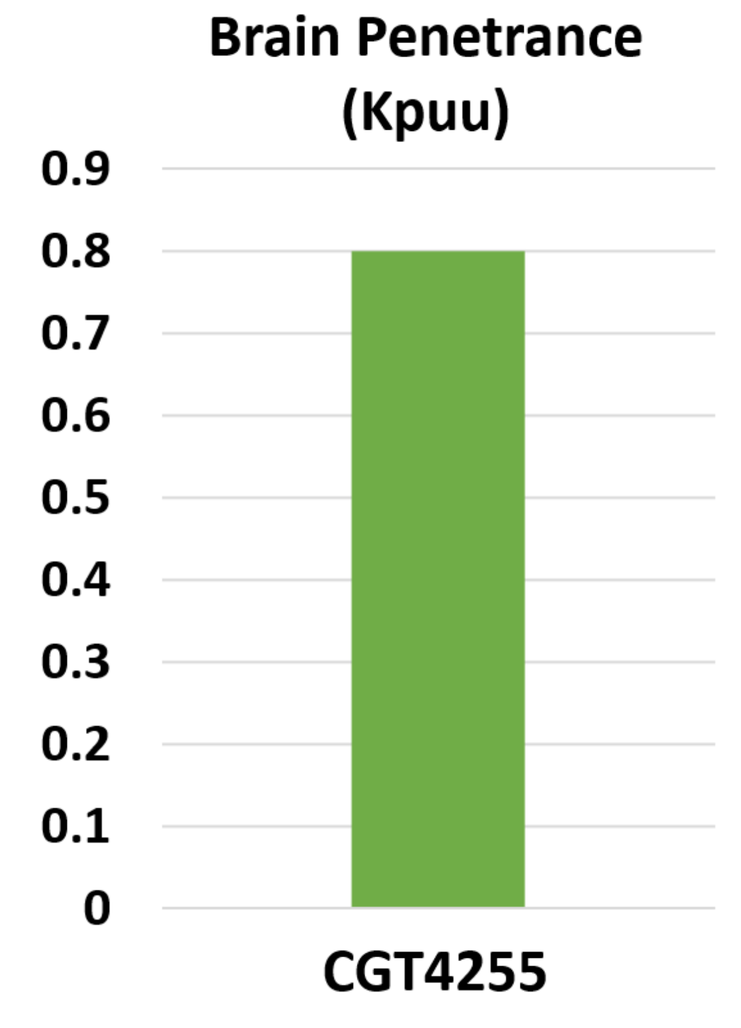
Brain Penetrance (Kpuu)
CGT4255

- Dose escalating oral exposure in mice
- Low clearance and high oral bioavailability seen at all doses
- CGT4255 Kpuu was 0.80 at 100 mg/kg at 1h in mice

Figure 5. CGT4255 Shows Robust Inhibition of pErbB2 in ErbB2 Mutant PD Models

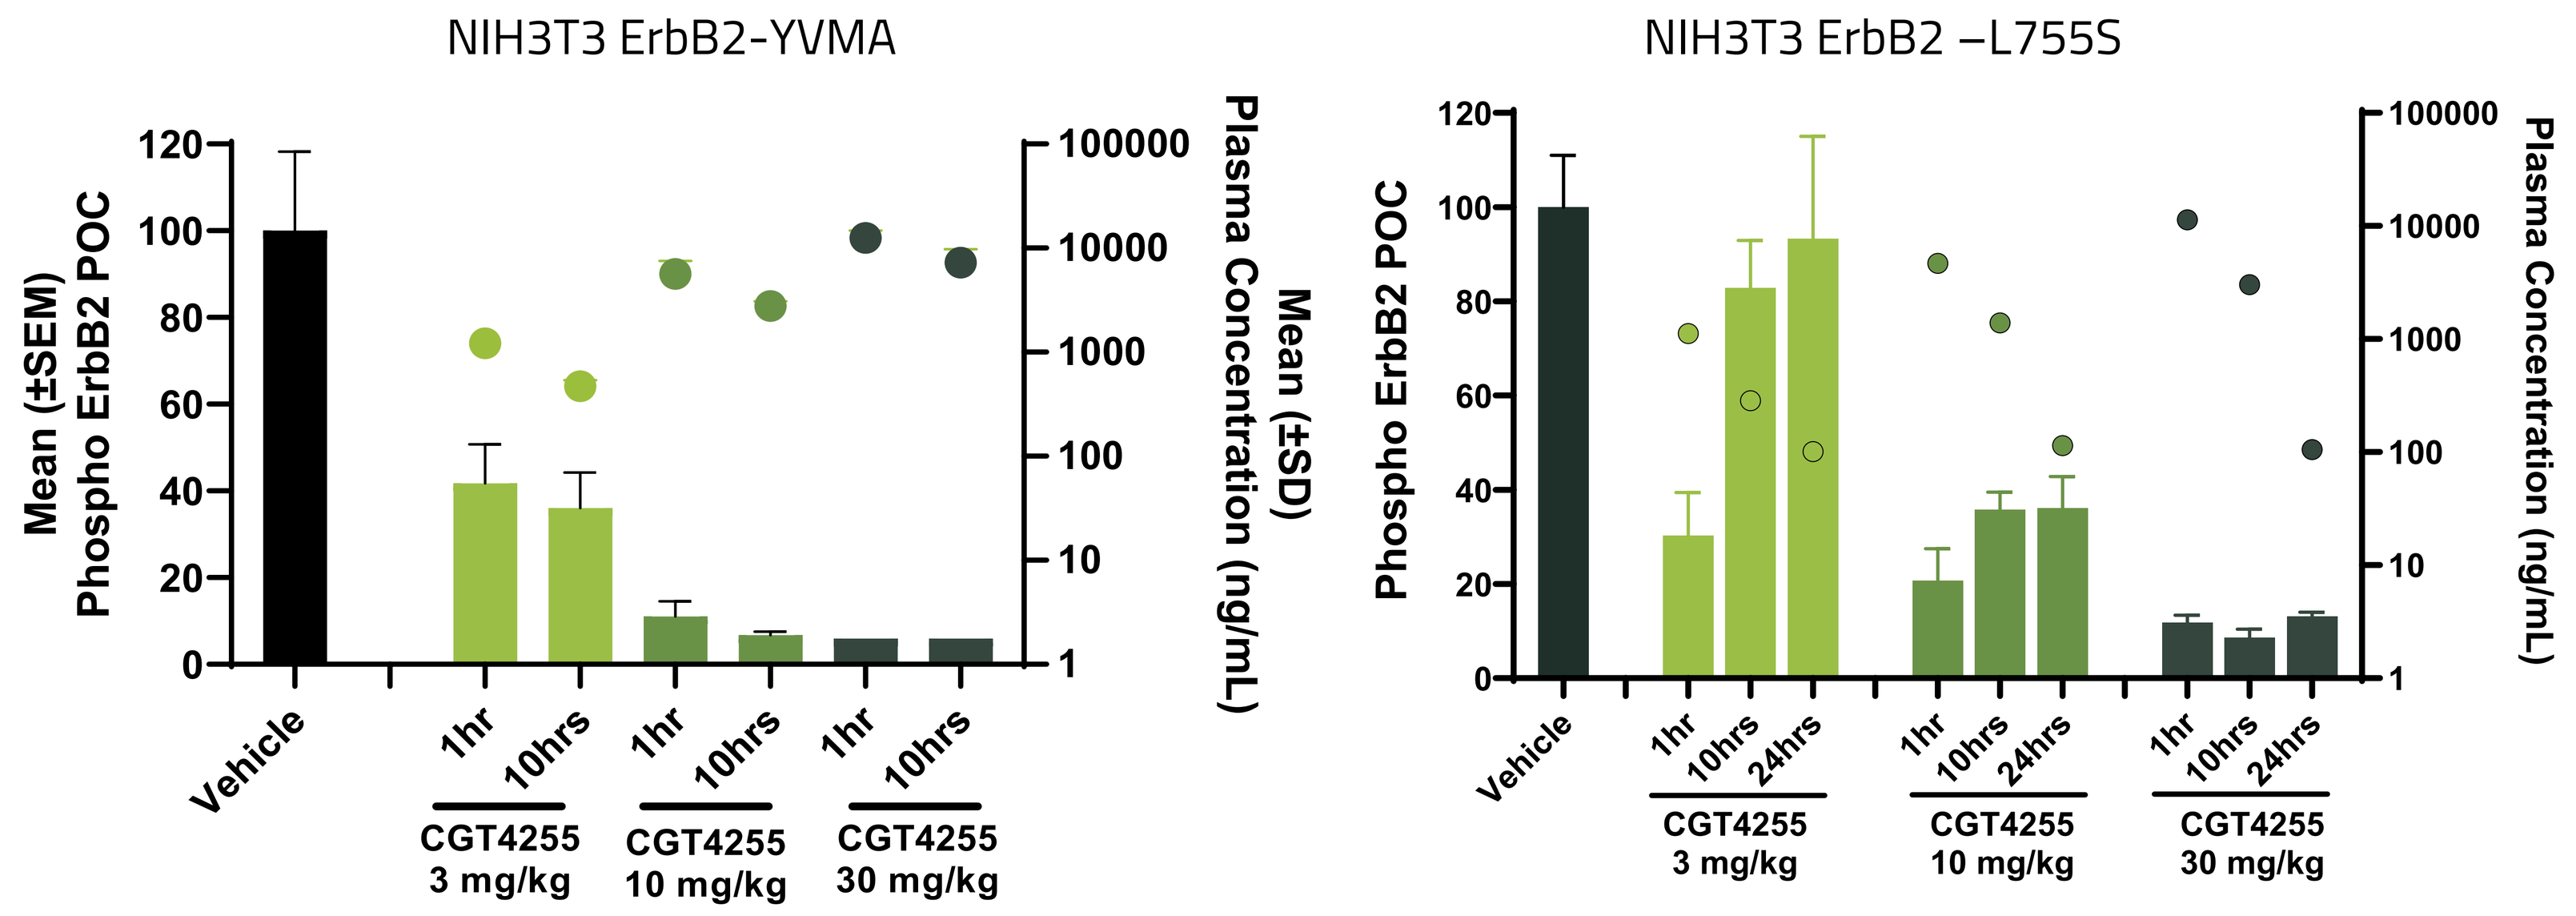
NIH3T3 ErbB2-YVMA
NIH3T3 ErbB2 –L755S
Mean (±SEM) Phospho ErbB2 POC
Mean (±SD) Plasma Concentration (ng/mL)
Phospho ErbB2 POC
Plasma Concentration (ng/mL)
Vehicle
1hr
10hrs
24hrs
CGT4255 3 mg/kg
CGT4255 10 mg/kg
CGT4255 30 mg/kg

- CGT4255 shows dose dependent inhibition of pErbB2 in NIH3T3 YVMA and L755S PD models
- Inhibition >90% seen up to 10 hr at 10 mg/kg PO in the YVMA model
- In the L755S model, p-ErbB2 inhibition of >90% was observed for 24hr at 30 mg/kg PO

Figure 6. CGT4255 Shows Regressions and is Well-Tolerated in an NIH3T3 ErbB2 – L755S TGI Study

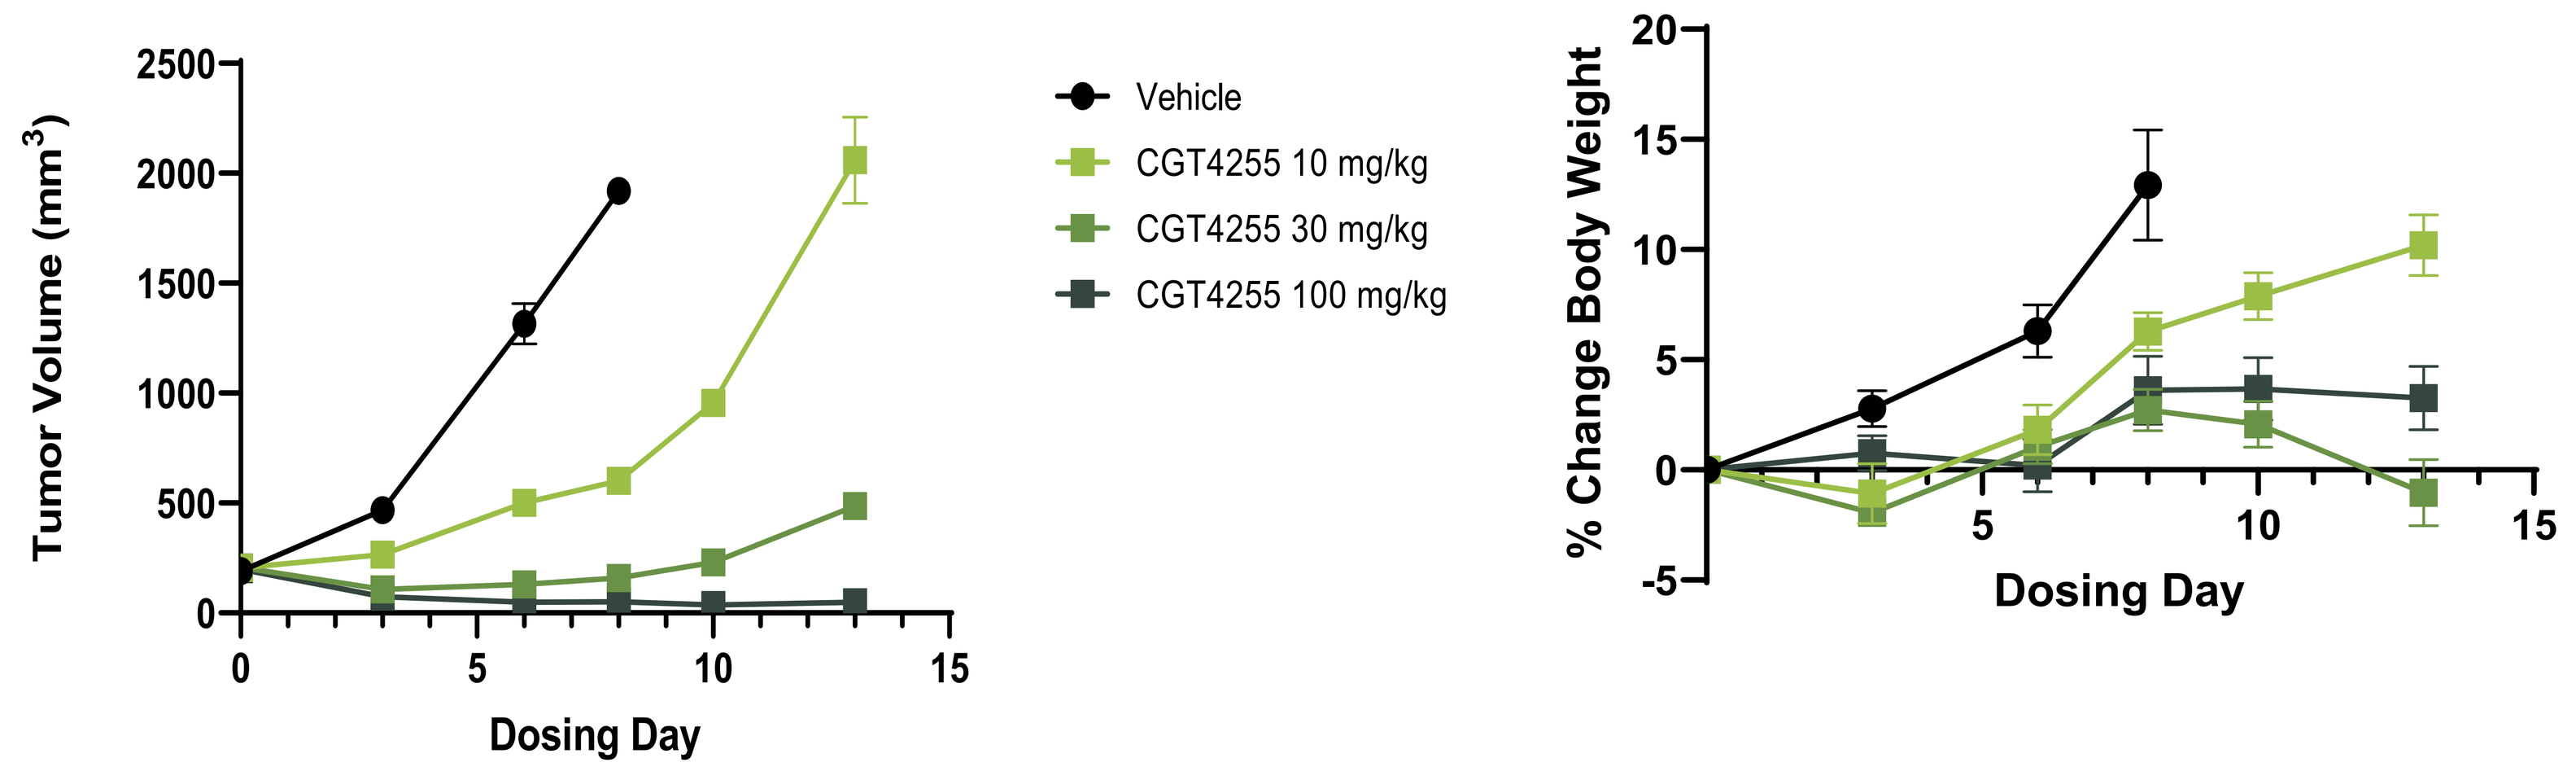
Tumor Volume (mm^3)
Dosing Day
Vehicle
CGT4255 10 mg/kg
CGT4255 30 mg/kg
CGT4255 100 mg/kg
% Change Body Weight
Dosing Day

- CGT4255 was dosed PO BID at 10, 30, or 100 mg/kg for 13 days in an engineered NIH3T3 ErbB2-755S tumor growth inhibition mouse model
- CGT4255 showed dose responsive TGI with full regressions observed at the 100 mg/kg dose
- CGT4255 was well tolerated at all doses with no body weight loss or deaths observed in any treatment group

Conclusions

- **CGT4255 –Advanced Lead**
 - **Low nM potency against ErbB2 WT and oncogenic ErbB2 mutations**
 - **100-fold selectivity over WT-EGFR, highly selective across the kinome, Cyps, receptors, channels, and hERG**
 - **Exceptional stability in human whole blood and liver cytosol fractions**
 - **Dose ascending PK in mice with low clearance and high oral bioavailability**
 - **Best-in class 80% brain penetrance at 100 mg/kg in mice**
- **Maximum inhibition of pErbB2 observed at a 30 mg/kg PO dose in both NIH/3T3 ErbB2-YVMA and ErbB2-L755S tumor models**
- **Complete regressions were seen at 100 mg/kg PO BID in the NIH3T3 ErbB2-L755S TGI model and was well tolerated**

REFERENCES: 1. Arteaga et al. 2012. Nat Rev Clin Oncol. 9:16-32. 2. Tai et al. 2010. J Control Release.146(3):264–275. 3. Yarden and Pines. 2012. Nat Rev Cancer.12:553–563. 4. American Cancer Society. Cancer Facts & Figures 2023. Atlanta: American Cancer Society; 2023. 5. The AACR Project GENIE Consortium. AACR Project GENIE: Powering Precision Medicine Through An International Consortium, Cancer Discovery 2017. 6. Monette Aujay et al. AACR 2023 Poster # 4019. 7. Kristin L. Andrews et al EORTC 2022, Poster 212.

For questions, please contact mark.chicarelli@cogentbio.com.

DISCLOSURES: All authors are employees of Cogent Biosciences
ACKNOWLEDGEMENTS: Zhang Peichuan, Qiaoqiao Xu, Huijuan Lv, Ning Han, and the Enzymology & *In Vitro* Novel Drug Screening Team, WuXi Biology. Dr. Binglin Wang, Dr. Le Chang, Dr. Ke Zhang and their WuXi AppTec Chemistry team.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster

This presentation is the intellectual property of Cogent Biosciences, Inc.
Contact evan.kearns@cogentbio.com for permission to reprint and/or distribute.