In Vivo Preclinical Characterization of a Novel FGFR2 Selective Inhibitor with Potency Against FGFR Activating Mutations

John Fischer, Karyn Bouhana, Mark J Chicarelli, Brad Fell, Jennifer Fulton, Anna Guarnieri, Leyla Haygood, Ravi Jalluri, Amber Johnson, Daniel Krischlunas, Cori Malinky, Macedonio Mejia, Rob Rieger, John Robinson, Mareli Rodriguez, Aaron C Smith, Francis Sullivan, Yang Wang, Shannon Winski, Yeyun Zhou | Cogent Biosciences, Inc., Boulder, Colorado and Cambridge, Massachusetts USA

Abstract Number: 227
Poster Board: PB107

Background

- Fibroblast growth factor receptors (FGFRs) are a family of transmembrane receptors consisting of isoforms FGFR1-FGFR4[1]
 - FGFR signaling impacts key cellular processes (**Fig. 1**)
 - Binding of a ligand to the inactive FGFR monomer leads to receptor dimerization, phosphorylation, and downstream signaling
 - Key processes include cell survival, proliferation, migration and differentiation
- FGFR2/3 alterations are well-established oncogenic drivers in multiple indications[2]
- FGFR2/3 alterations are present in 4.1% of all cancers, 80% of which are activating mutations
- FGFR2 is highly altered in cholangiocarcinoma and uterine endometroid tumors (**Fig. 2**)

Figure 1. Fibroblast Growth Factor Receptors (FGFRs)

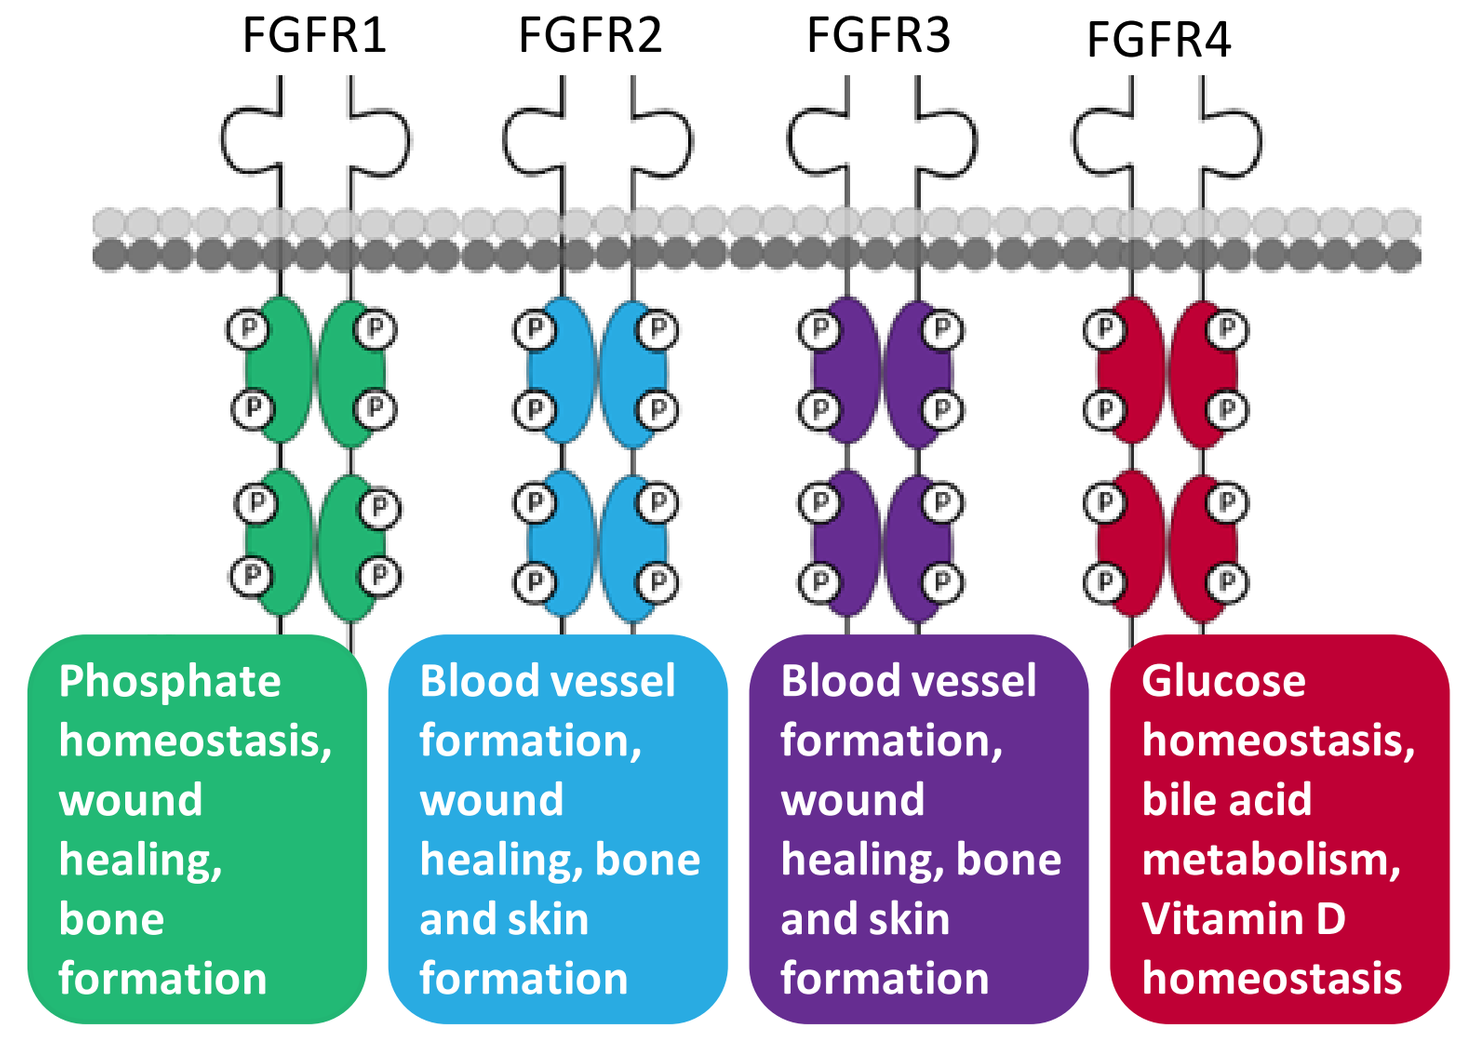
FGFR1
FGFR2
FGFR3
FGFR4
Phosphate homeostasis, wound healing, bone formation
Blood vessel formation, wound healing, bone and skin formation
Blood vessel formation, wound healing, bone and skin formation
Glucose homeostasis, bile acid metabolism, Vitamin D homeostasis

Figure 2. FGFR2 Alteration Frequency by Tumor Types[2]

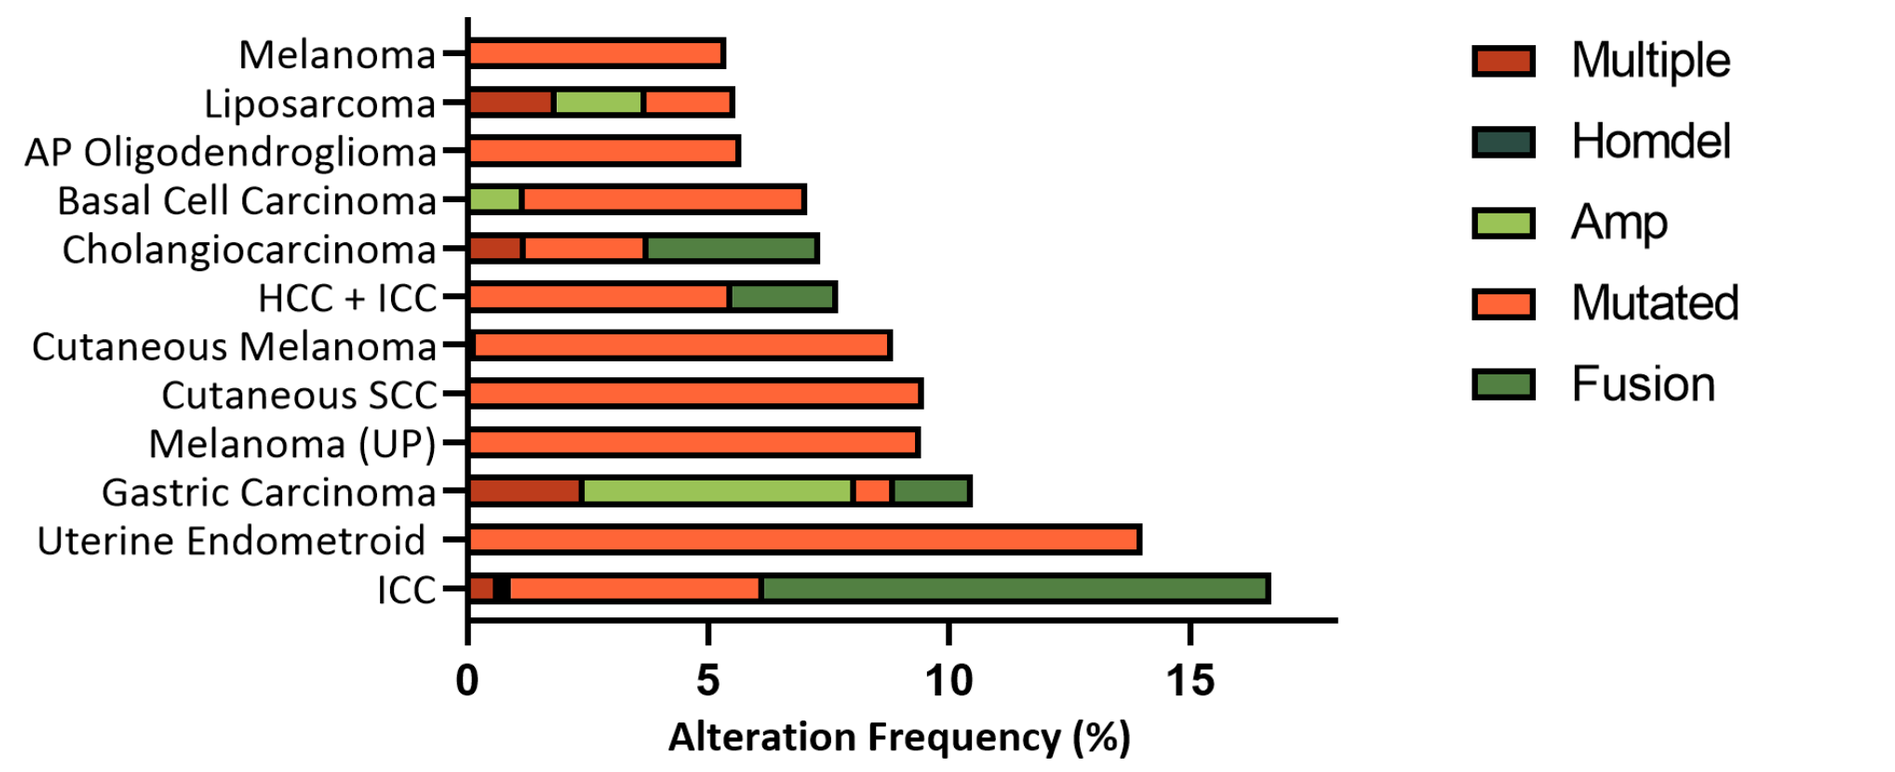
Melanoma
Liposarcoma
AP Oligodendroglioma
Basal Cell Carcinoma
Cholangiocarcinoma
HCC + ICC
Cutaneous Melanoma
Cutaneous SCC
Melanoma (UP)
Gastric Carcinoma
Uterine Endometroid
ICC
0
5
10
15
Alteration Frequency (%)
Multiple
Homdel
Amp
Mutated
Fusion

Table 1. Clinical Features and Clinical Coverage of Pan-FGFR Inhibitors

	Clinical Features				Clinical Coverage					
					Indication			FGFR alteration		
Clinical Compound	Dose Schedule	ORR	Hyperphos	Stomatitis	Adv/met ICC	Adv/met UC	Indications outside of ICC/UC	FGFR2 fusions	FGFR2 activating mutations	FGFR2 resistance mutations
Pemigatinib[3,5]	2 wk on/ 1 wk off	36% ICC	92%	35%						
Infigratinib[6,7]	3 wk on/ 1 wk off	23% ICC	82%	56%						
Futibatinib[8,9]	Monitor tolerability	42% ICC	85%	30%						

UC, Urothelial Carcinoma; ICC, Intrahepatic Cholangiocarcinoma; KD, Kinase Domain

Not Approved: Approved:

- FGFR1 mediated hyperphosphatemia was the most common DLT of approved pan-FGFR inhibitors
- Inhibition of FGFR2 fusions in cholangiocarcinoma and FGFR3 mutations in urothelial cancer has led to improved clinical outcomes in defined patient populations[3,4]
- There remains an unmet need for a potent FGFR2 inhibitor with broad mutational coverage and which avoids FGFR1 to enable increased response rates with longer durability (current drugs 5-9 months)[3, 10] and better tolerability than existing agents

Figure 3. Treatment of FGFR2 Acquired Resistance Mutations Represents an Unmet Medical Need[11-14]

- Despite clinical hyperphosphatemia, disease progression is frequently driven by acquired resistance mutations
- Gatekeeper V564X (43% of patients) and molecular brake N549K (48% of patients) mutations are the main mechanisms of resistance to existing therapies[11-14]
- This highlights the need for compounds with broader mutational coverage to maximize response rate and increase duration of response

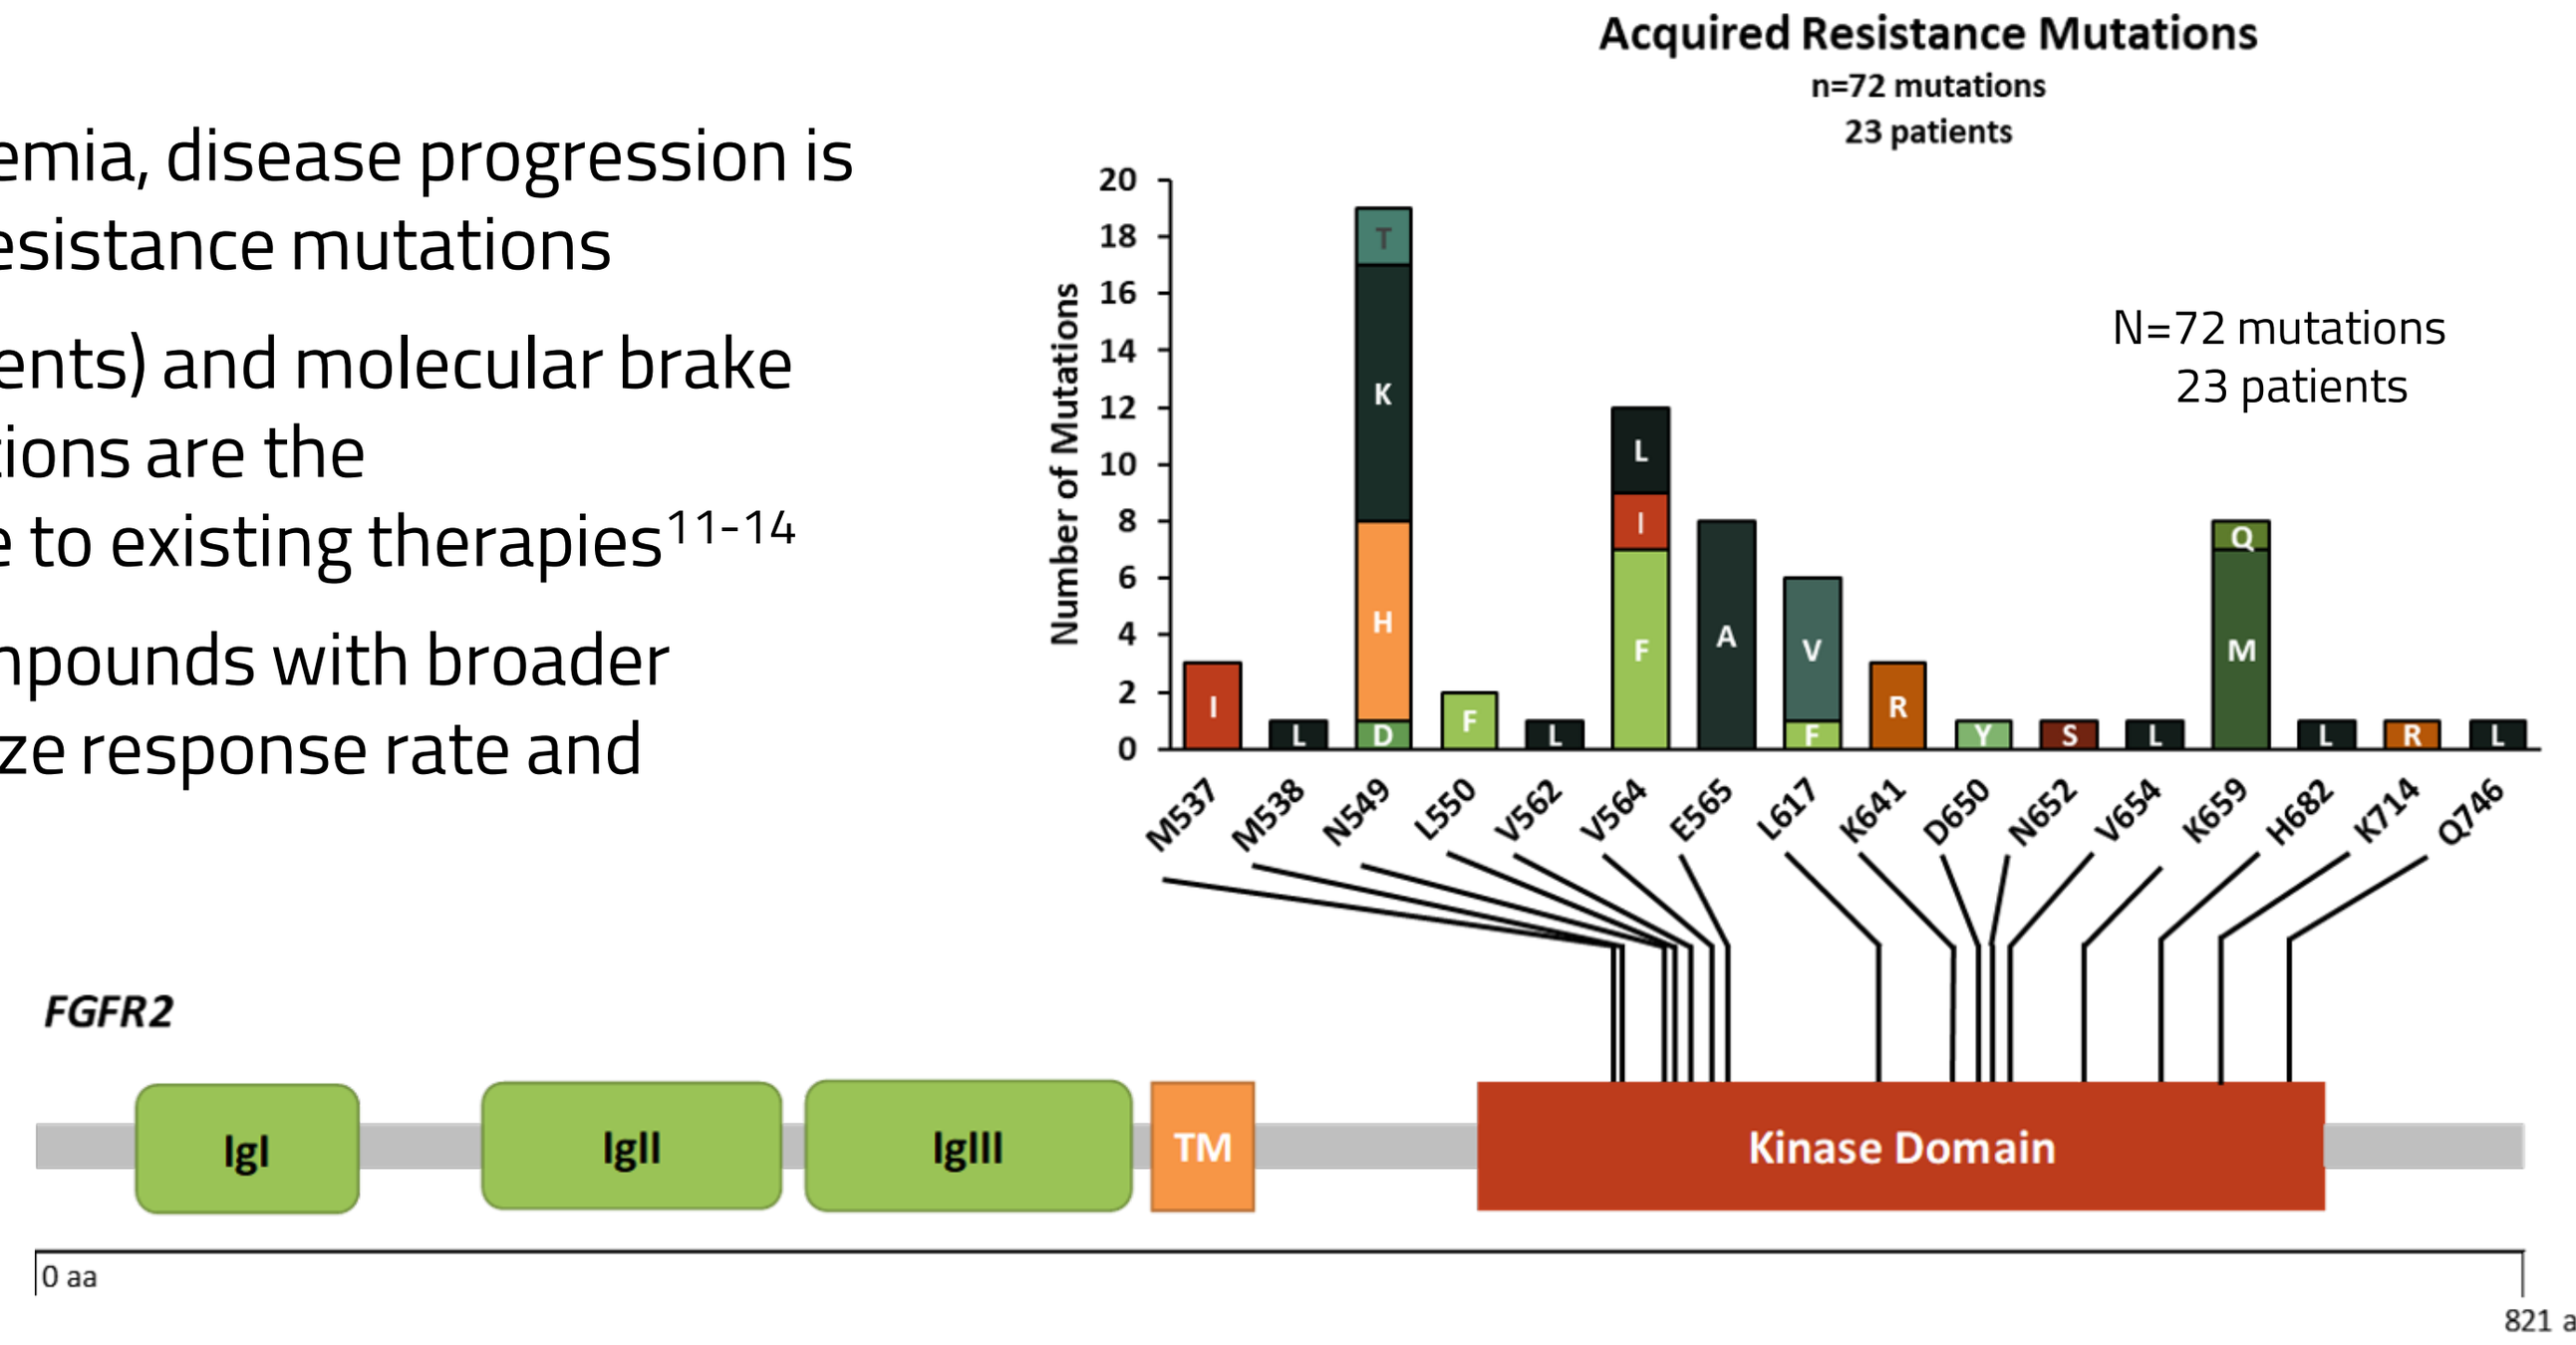
Acquired Resistance Mutations
n=72 mutations
23 patients
N=72 mutations
23 patients
Number of Mutations
M537
M538
N549
L550
V562
V564
E565
L617
K641
D650
N652
V654
K659
H682
K714
Q746
FGFR2
IgI
IgII
IgIII
TM
Kinase Domain
0 aa
821 aa

Results

Table 2. CGT1672 Retains Potency Across Primary and Acquired Resistance Mutations

	Anti-Target (nM)	FGFR2 (nM)				
	FGFR1	WT	V564F	V564L	V564I	N549K
CGT1672	346	15	9	7	14	18
RLY4008	2063	8	1	5	88	28
Pemigatinib	10	2	>1000	819	58	81
Futibatinib	4	2	147	30	2	1

- Table 2 shows mechanistic cellular IC_{50}'s of FGFR1, FGFR2WT and the FGFR2 gatekeeper and molecular brake mutants
- Optimization of CGT0292 (AACR 2022 Abs 167) led to CGT1672 with improved potency vs. point mutation
- **Only CGT1672 is selective over FGFR1 and maintains potency across the acquired resistance mutations**
- Known clinical and commercial stage FGFR2 inhibitors have FGFR1 selectivity and/or target coverage issues which prevent optimal clinical efficacy
- Inhibition of the primary and acquired resistance mutations may provide deeper and more durable clinical responses

Figure 4. Crystal Structure of CGT1672 Bound to FGFR2 N549K Supported Structure Based Drug Design

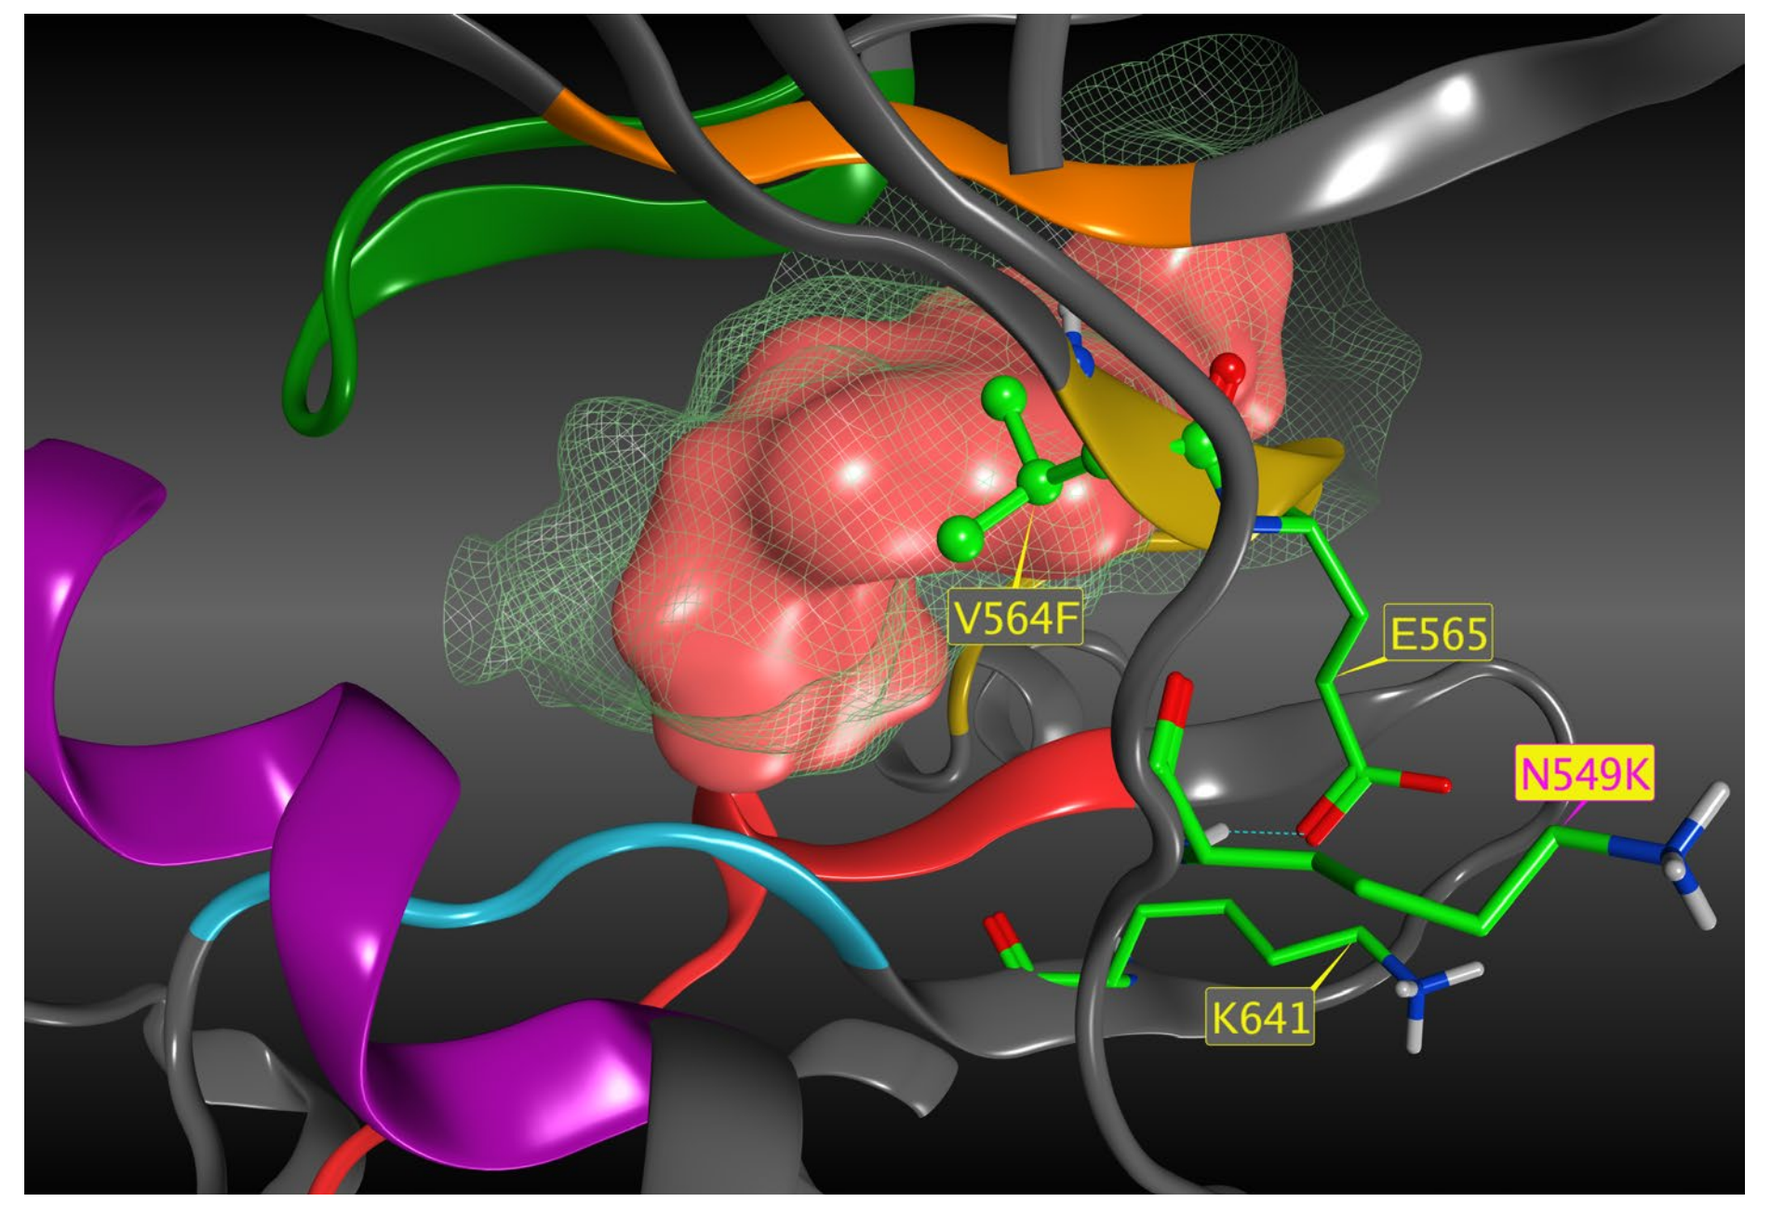
V564F
E565
N549K
K641

- 2.1Å Crystal structure of CGT1672 (shown as pink surface) bound to FGFR2 N549K mutant reinforces understanding for potency and selectivity
- CGT1672 has optimized binding in the ATP pocket and maintains potency for all of the prevalent FGFR2 mutations which are common modes of resistance to current drugs
- Structure based drug design has also been used to further optimize the inhibitors for selectivity over the general kinome, FGFR1, and FGFR4

Figure 5. Ongoing Optimization of CGT1672 has Led to Enhanced FGFR2/FGFR1 Selectivity

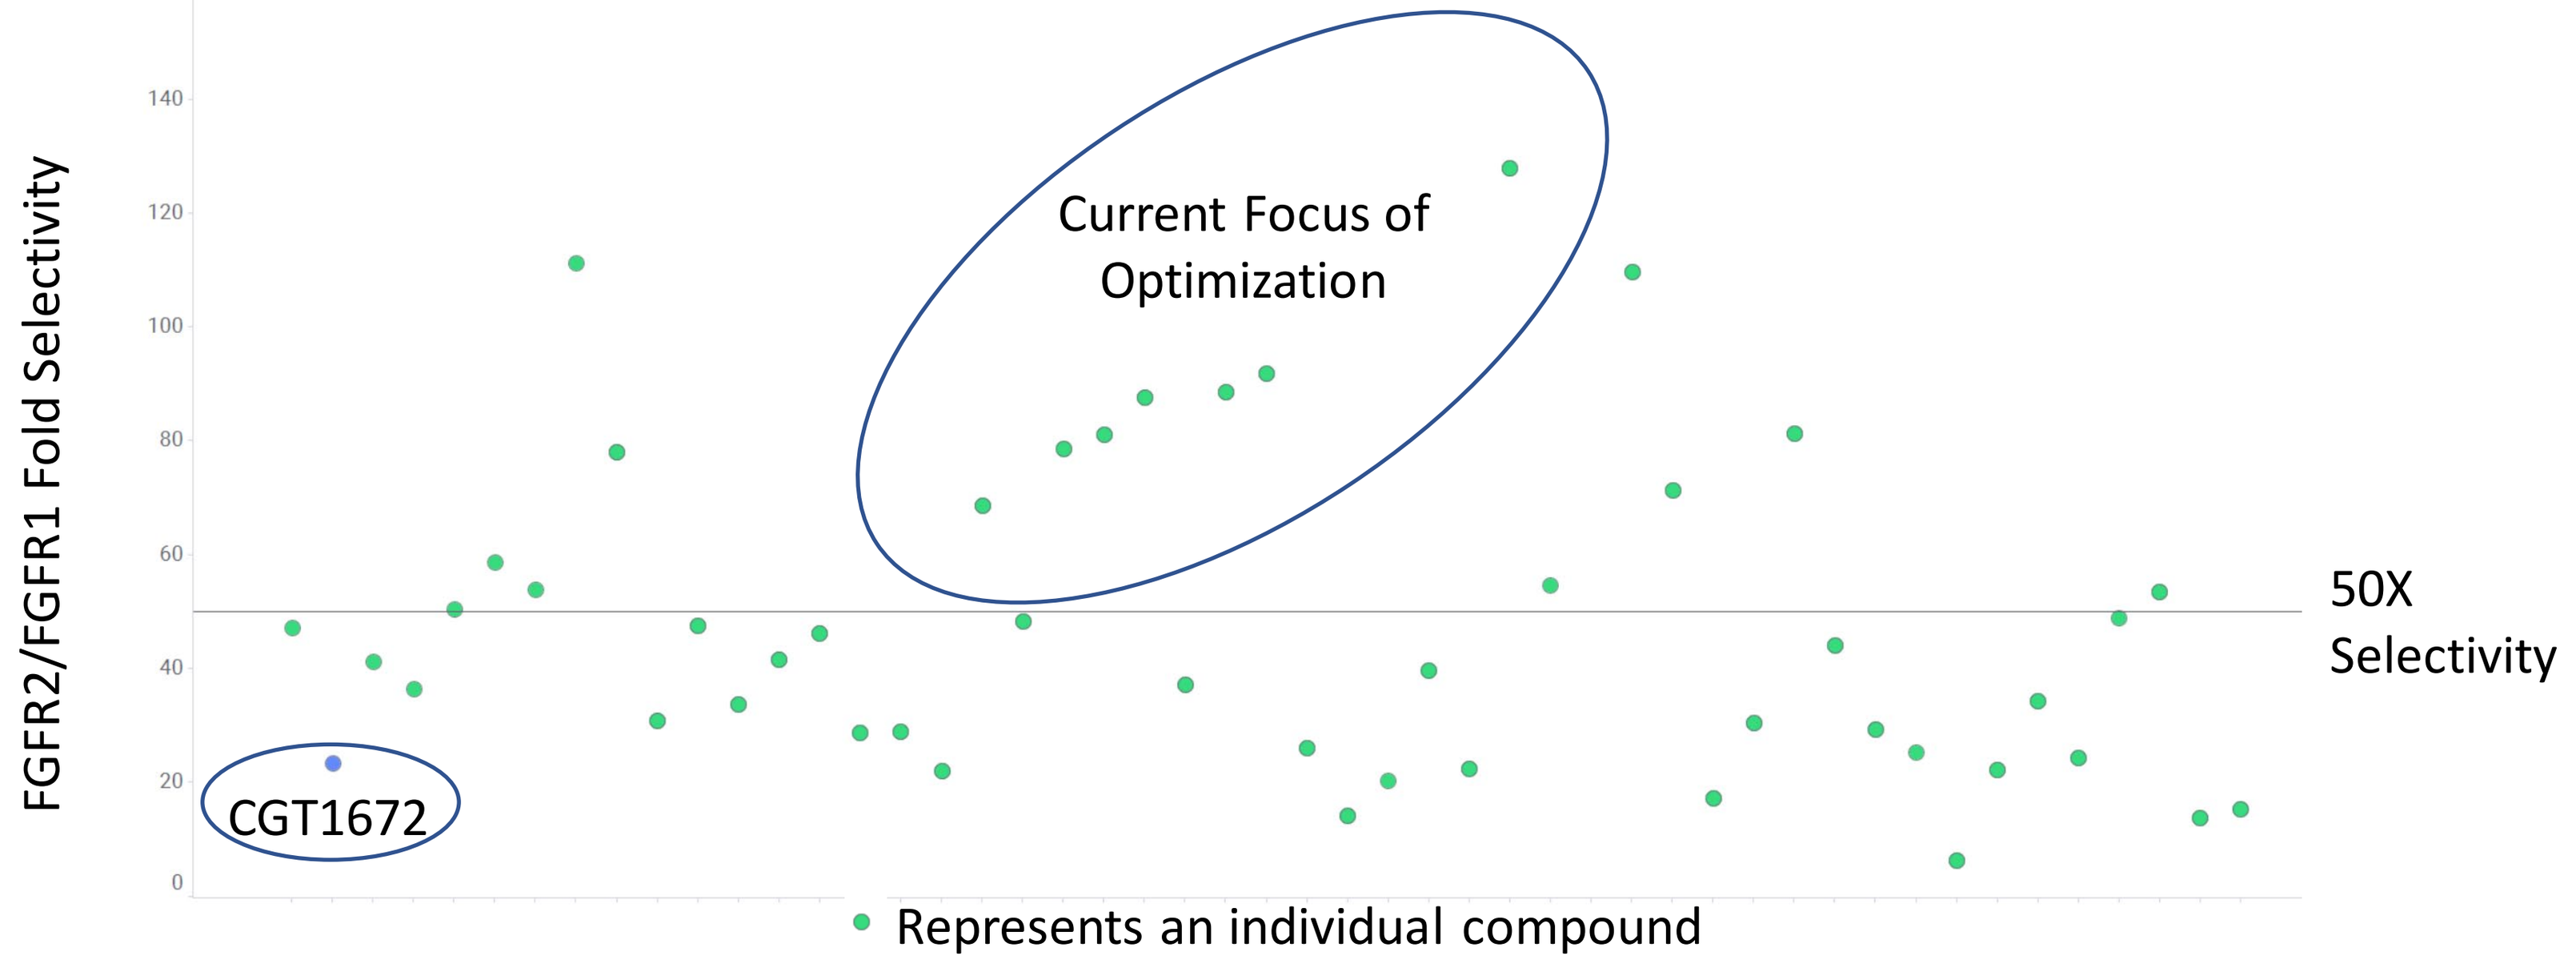
FGFR2/FGFR1 Fold Selectivity
Current Focus of Optimization
50X Selectivity
CGT1672
Represents an individual compound

- CGT1672 was selected as a tool compound to be used to complete validation of our in vivo models
- Optimization of CGT1672 has led to a set of analogs that retain high potency across all FGFR2 mutants, 30% of which are >50X selective for FGFR2 vs. FGFR1

Figure 6. CGT1672 Shows Favorable PK/PD in the Clinically Relevant AN3 CA (K310R/N549K) Model

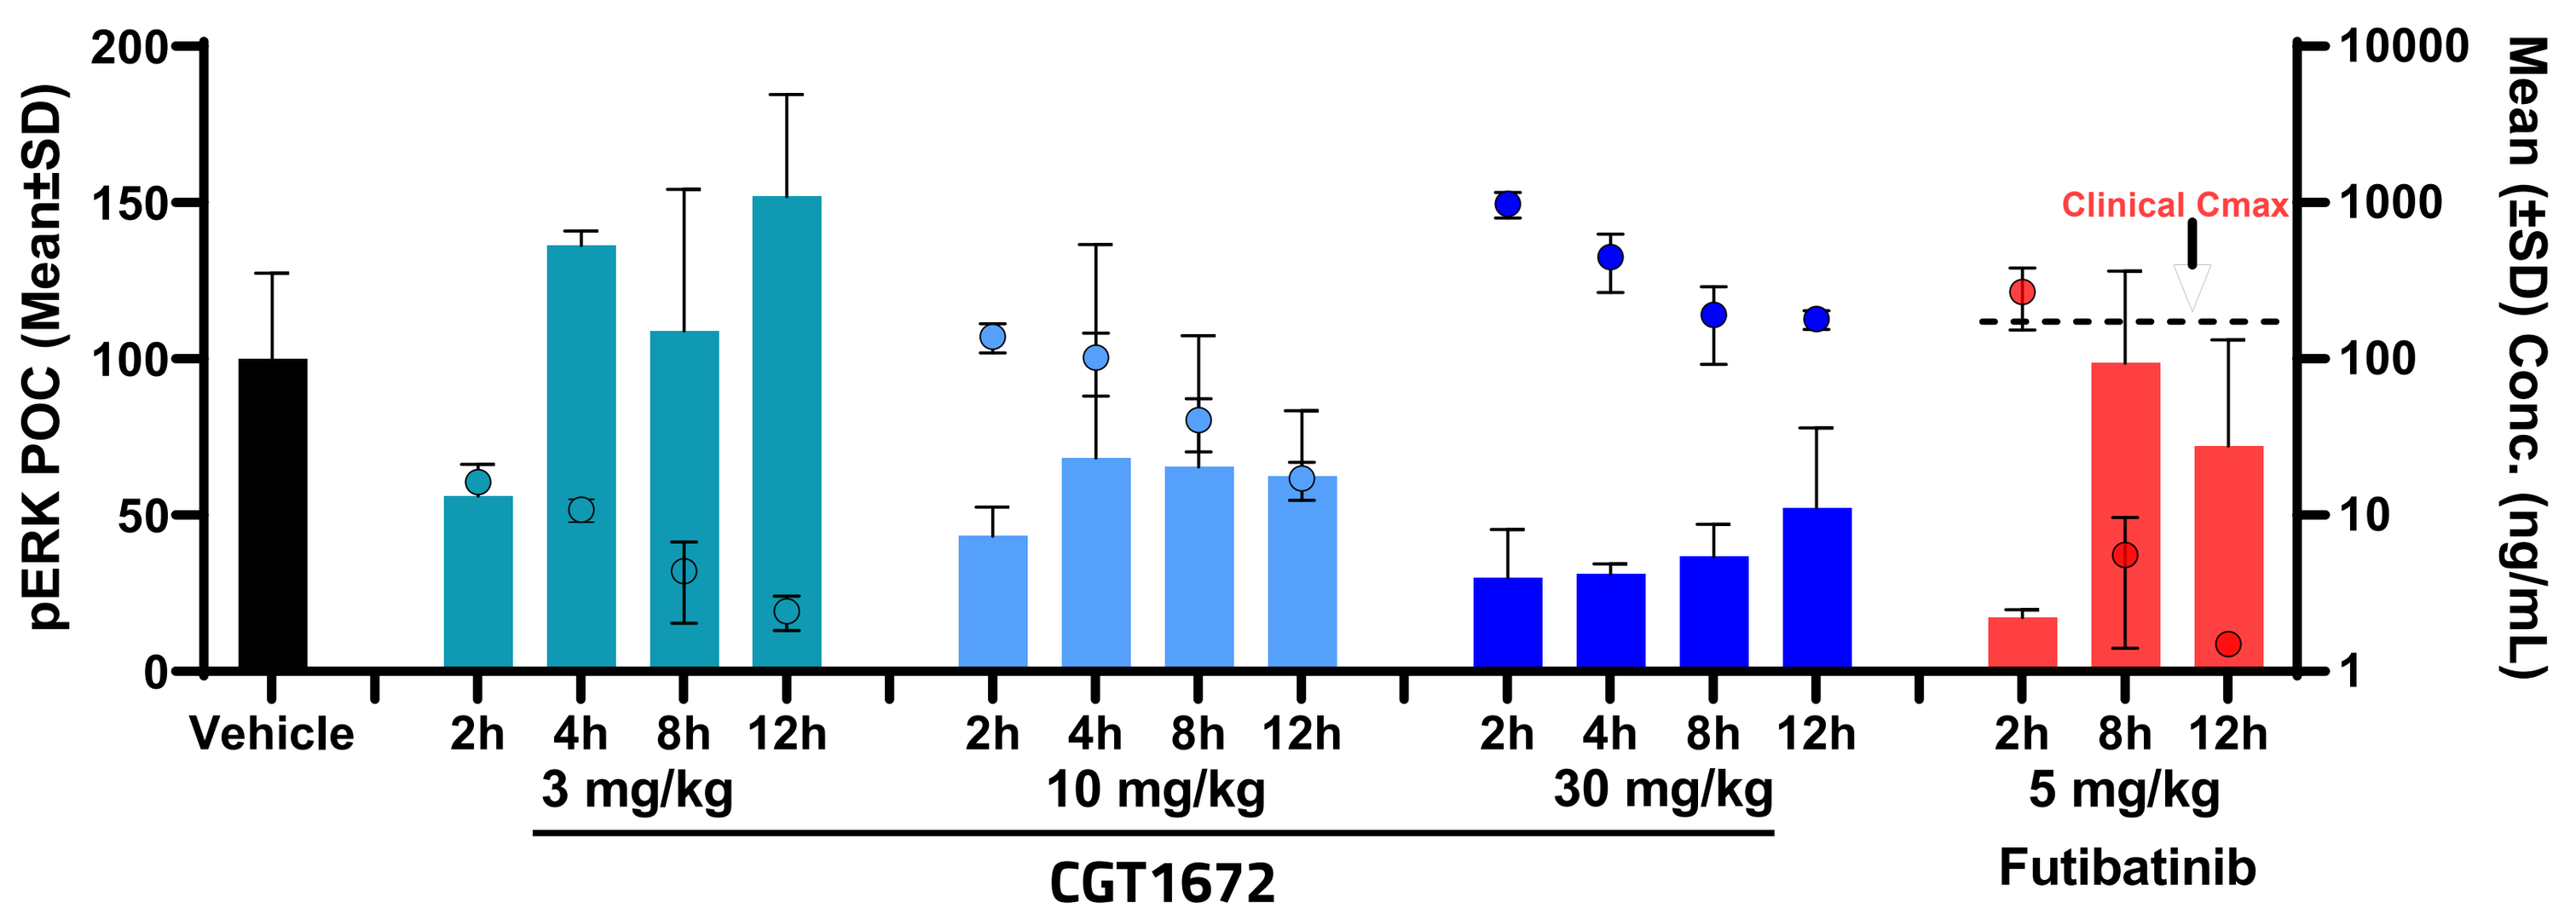
pERK POC (Mean±SD)
Mean (±SD) Conc. (ng/mL)
Clinical Cmax
Vehicle
2h 4h 8h 12h
3 mg/kg
10 mg/kg
30 mg/kg
CGT1672
2h 8h 12h
5 mg/kg
Futibatinib

- CGT1672 shows a dose dependent inhibition of pERK in the AN3 CA PK/PD model
- Futibatinib was used as the control compound for both the PD(**Fig. 6**) and TGI(**Fig. 7**) AN3 CA mouse tumor models but has limited clinical utility due to hyperphosphatemia

Figure 7. CGT1672 Demonstrates DE Mouse PK and Regressions in the AN3 CA (K310R/N549K) TGI Model

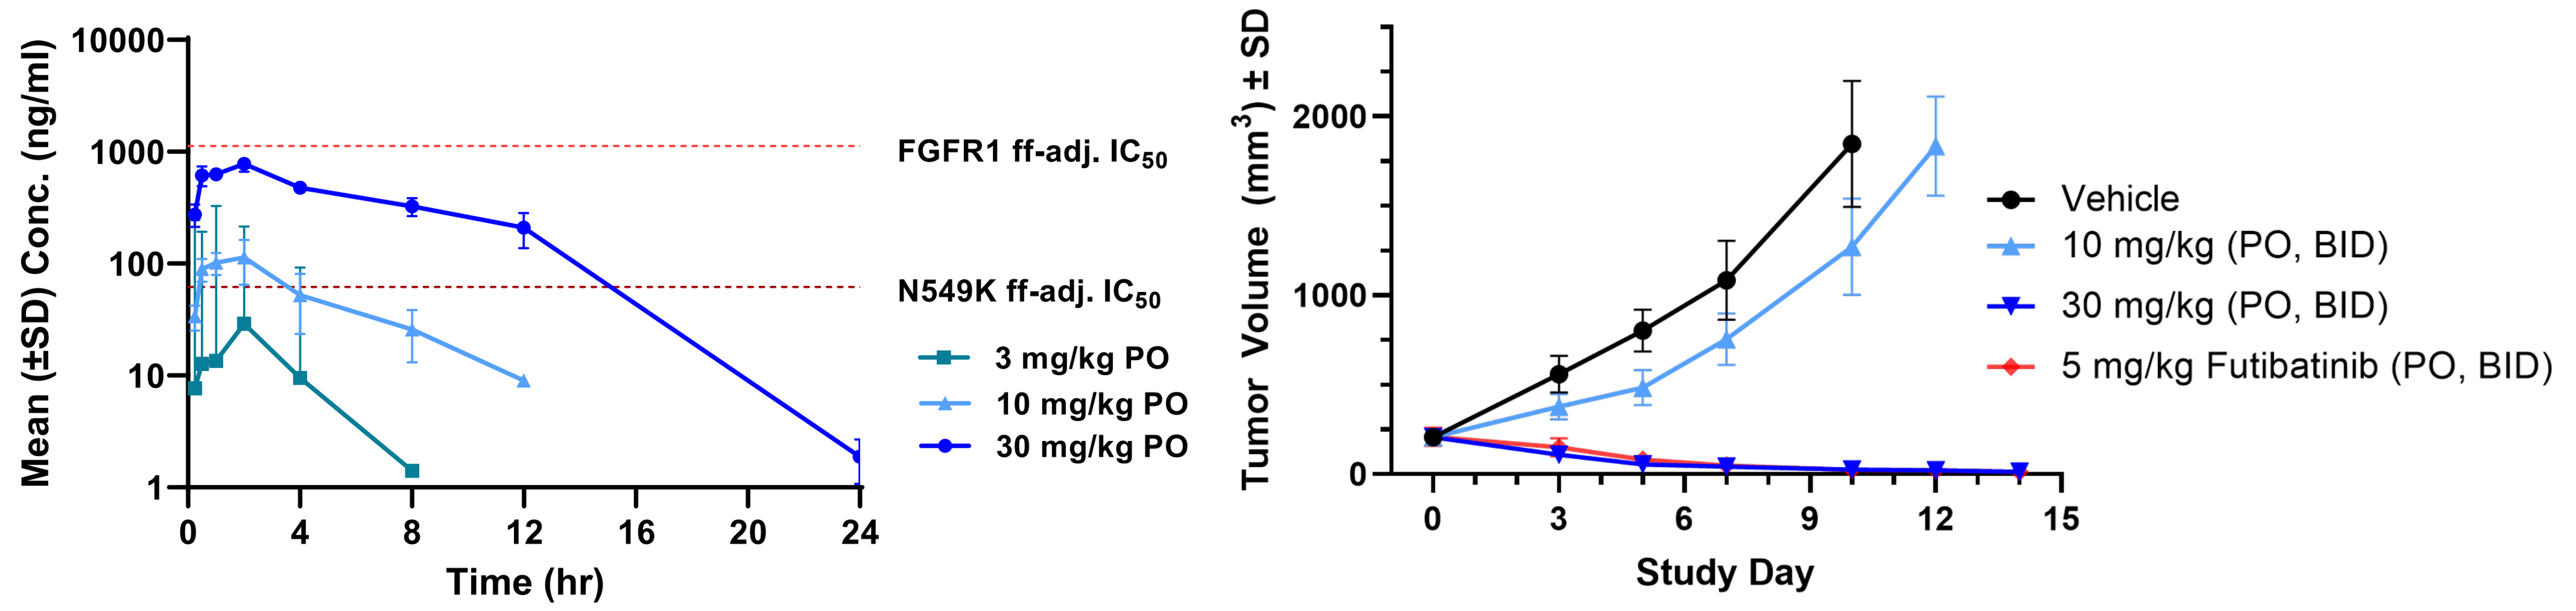
Mean (±SD) Conc. (ng/ml)
Time (hr)
FGFR1 ff-adj. IC50
N549K ff-adj. IC50
3 mg/kg PO
10 mg/kg PO
30 mg/kg PO
Tumor Volume (mm3) ± SD
Study Day
Vehicle
10 mg/kg (PO, BID)
30 mg/kg (PO, BID)
5 mg/kg Futibatinib (PO, BID)

- CGT1672 dose response PK shows IC_{50} target coverage of N549K mutant FGFR2 with a selectivity window for FGFR1 at 30 mg/kg
- CGT1672 at 30 mg/kg and futibatinib at 5 mg/kg gave regression with BID PO dosing
- Futibatinib is clinically limited due to inhibition of FGFR1, preventing treatment of the N549K molecular brake mutation

Conclusions

- FGFR2 alterations are well established oncogenic drivers in multiple indications
- Approved FGFR pan-inhibitors fail to capture the full landscape of FGFR2 altered tumor types
- **Gatekeeper (V564X) and molecular brake (N549K) mutations are untreated in the primary setting and have been identified as a common mechanisms of resistance to current therapies, which may explain their limited durability of response**
- CGT1672 and inhibitors in this series are potent across FGFR2 primary and required resistance mutations
- Optimization of the tool compound CGT1672, using structure-based drug design ,has led to a promising series of FGFR2 inhibitors that are >50x selective for FGFR2 over FGFR1
- Dose Response PK/PD is shown with CGT1672 in the AN3 CA (K310R/N549K) model
- CTGT1672 shows tumor regressions in an AN3 CA mouse xenograft model, which differentiates CGT1672 from other selective FGFR2 inhibitors that lack activity against the N549K molecular brake mutantion
- Further optimization of this series has led to compounds which are at least 50X selective versus FGFR1

Scan the QR code to access a PDF copy of this poster

References. 1. Helsten T, et al. *Clin Cancer Res.* 2016;22(1):259-267; 2. AACR Project GENIE Consortium. *Cancer Discov.* 2017;7(8):818-831; 3. Abou-Alfa GK, et al. *Lancet Oncol.* 2020;21(5):671-684; 4. Siefker-Radtke et al. *Lancet Oncol.* 2022;23(2):248-258; 5. PEMAZYRE (pemigatinib; prescribing information). Wilmington, DE: Inctye; Corporation; 2021; 6. Javle M, et al. *J Clin Oncol.* 2018;36(3):276-282. *2018;* 7. TRUSELTIQ (infigratinib; prescribing information). Brisbane, CA: QED Therapeutics: 2021; 8. Goyal L, et al. *Cancer Discov.* 2017; 7(3): 252–263; 9. LYTGOBI (futibatinib, prescribing information). Princeton, NJ: Taiho Pharmaceutical Co., Ltd.; 2022; 10. Goyal L, et al. *Cancer Research.* 2021;81(13_Suppl) Abstract CT010; 11. Goyal L, et al. *Cancer Discov.* 2017; 7(3): 252–263; 12. Goyal L, et al. *Cancer Discov.* 2019;9(8):1064-1079; 13. Varghese AM, et al. *JCO Precis Oncol.* 2021;5:PO.20.00178; 14. Silverman IM, et al. *Cancer Discov.* 2021;11(2):326-339. **Disclosures.** All authors are employees of Cogent Biosciences

34th EORTC/NCI/AACR (ENA) Symposium 2022 | 26-28 October 2022 | Barcelona, Spain