Pre-clinical Characterization of a Novel Series of FGFR2 Selective Inhibitors with Potency Against Clinically Relevant Mutations

John Fischer, Karyn Bouhana, Mark J Chicarelli, Josh Dahlke, Brad Fell, Jennifer Fulton, Anna Guarnieri, Leyla Haygood, Ravi Jalluri, Amber Johnson, Brent Mclean, Macedonio Mejia, Rob Rieger, John Robinson, Mareli Rodriguez, Francis Sullivan, Yang Wang, Shannon Winski, Yeyun Zhou

Cogent Biosciences, Inc., Boulder, Colorado and Cambridge, Massachusetts USA

Abstract Number: 167

Background

Figure 1. FGFR2 and FGFR3 are Well-Established Oncogenic Drivers in Multiple Indications

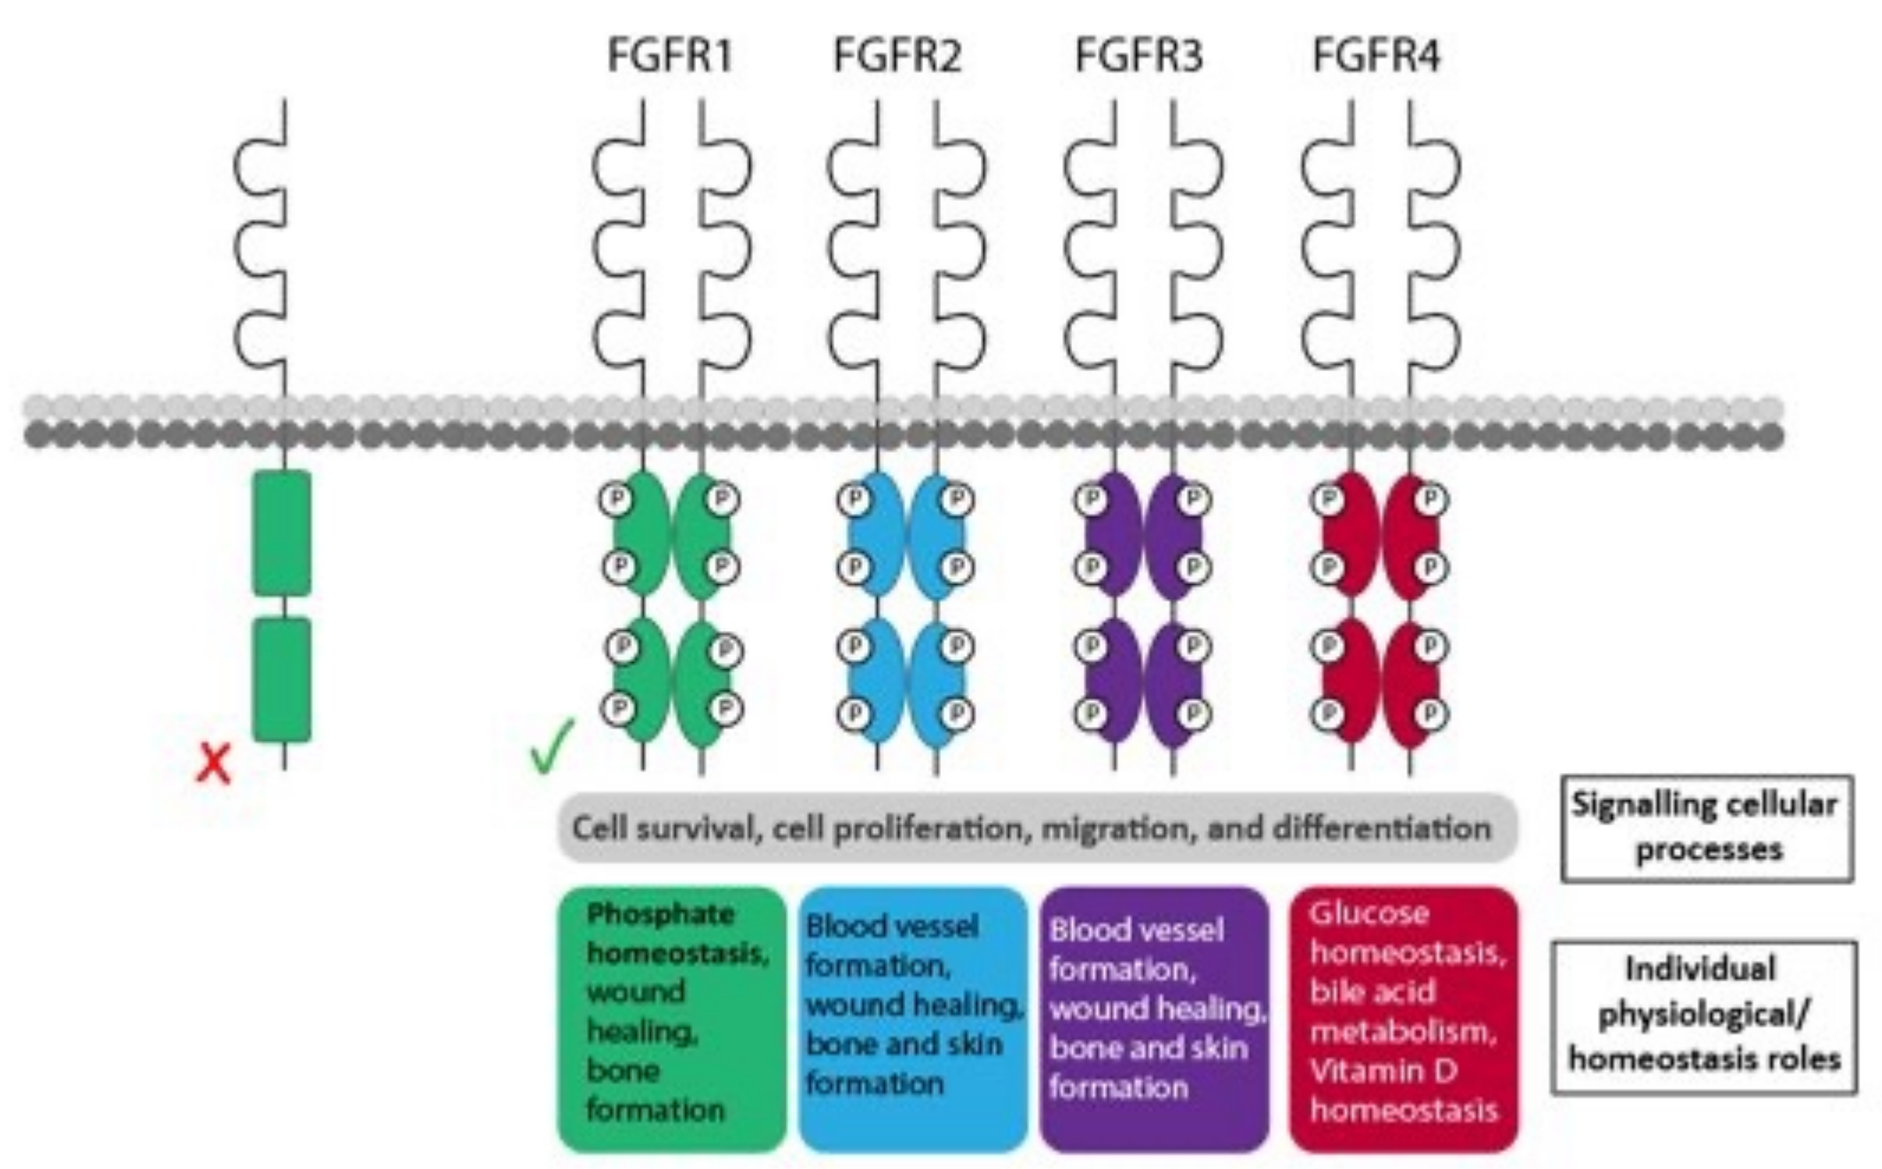
FGFR1
FGFR2
FGFR3
FGFR4
Cell survival, cell proliferation, migration, and differentiation
Signalling cellular processes
Phosphate homeostasis, wound healing, bone formation
Blood vessel formation, wound healing, bone and skin formation
Blood vessel formation, wound healing, bone and skin formation
Glucose homeostasis, bile acid metabolism, Vitamin D homeostasis
Individual physiological/ homeostasis roles

- Fibroblast growth factor receptors (FGFRs) are a family of transmembrane receptors consisting of isoforms FGFR1-FGFR4
- Binding of a ligand to the inactive FGFR monomer leads to receptor dimerization, phosphorylation, and downstream signaling
- Receptor mutations, amplifications, and fusions can result in activation of FGFRs and are recognized as well-established oncogenic drivers in multiple indications[1,2]
- Inhibition of FGFR3 mutations in urothelial cancer and FGFR2 fusions in cholangiocarcinoma has led to improved clinical outcomes in defined patient populations[3,4]

Table 1. First Generation Pan-FGFR Inhibitors Have Dose Limiting Toxicities (DLT) and Poorly Serve the Landscape of FGFR2/FGFR3-Altered Cancers

Clinical Compound	Clinical Features					Approved Clinical Coverage						
	IC50 enzyme (nM) (FGFR1,2,3,4)	Dose Schedule	ORR	Hyperphos	Diarrhea	Adv/met ICC	Adv/met UC	FGFR2 fusions	FGFR3 fusions	FGFR2 Activating mutations	FGFR3 mutations (KD)	FGFR2/3 resistance mutations
Pemigatinib[3,5]	0.4, 0.5, 1, 30	2 wk on/ 1 wk off	36% (ICC)	92%	47%							
Infigratinib[6,7]	0.9, 1.4, 1, 60	3 wk on/ 1 wk off	23% (ICC)	82%	24%							
Erdafitinib[8]	1, 2.5, 3, 6	Daily (schedule based on phosphate levels)	32% (UC)	76%	47%							

UC, Urothelial Carcinoma; ICC, Intrahepatic Cholangiocarcinoma; KD, Kinase Domain

- FGFR1 mediated hyperphosphatemia was the most common DLT observed in patients taking pan-FGFR inhibitors
- Diarrhea is observed in up to 47% of patients taking pan-FGFR inhibitors
- Approved pan-FGFR inhibitors fail to capture the full landscape of FGFR2 and FGFR3 altered tumor types

Figure 2a. FGFR2 is Highly Altered Across Tumor Type[1]

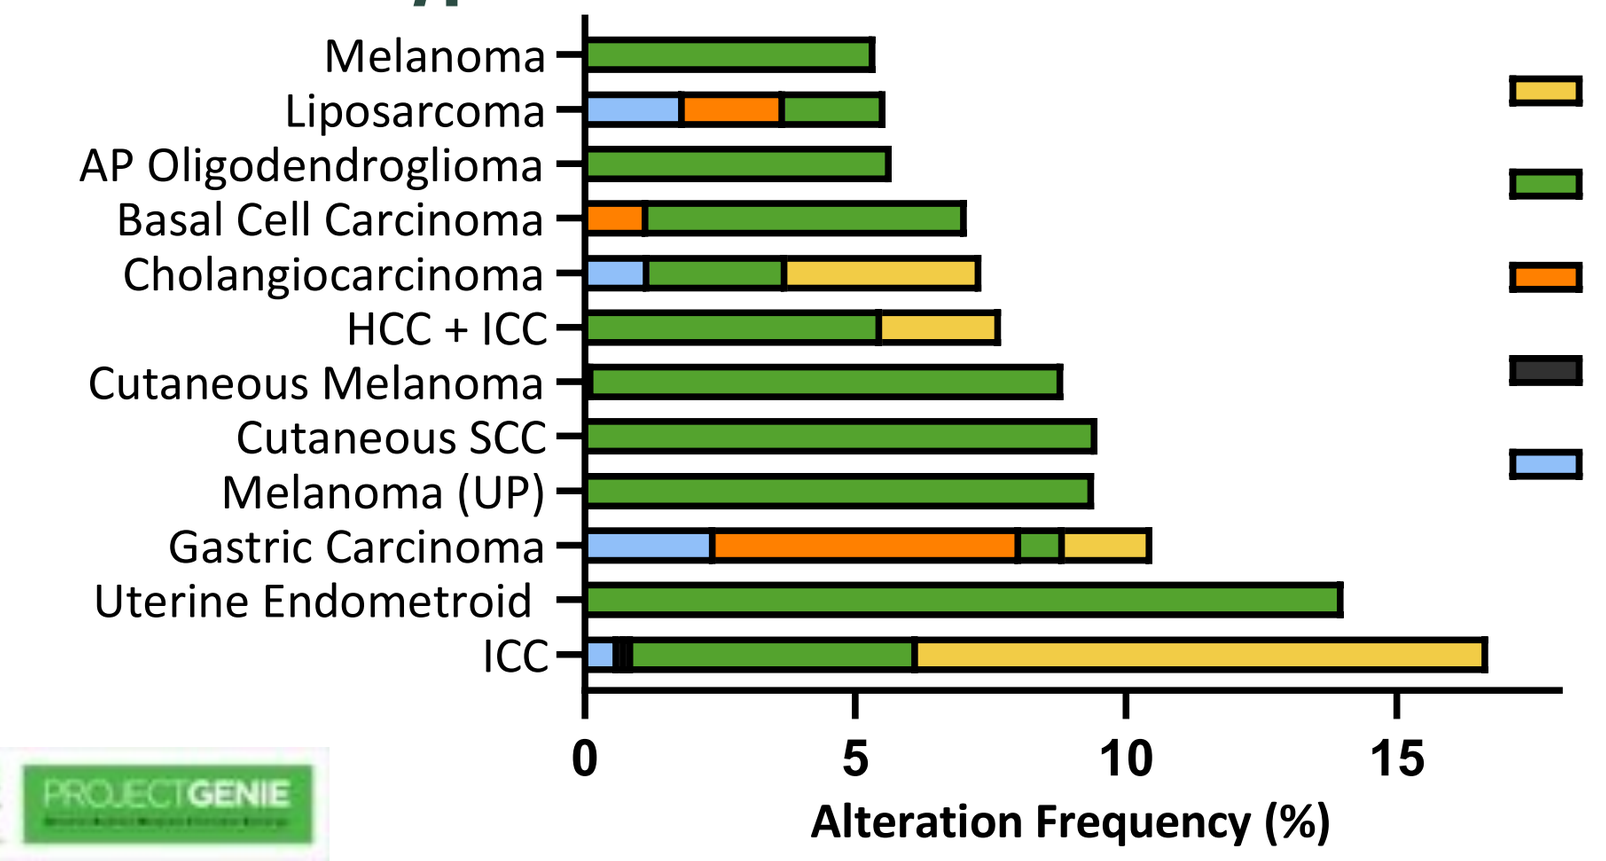
Melanoma
Liposarcoma
AP Oligodendroglioma
Basal Cell Carcinoma
Cholangiocarcinoma
HCC + ICC
Cutaneous Melanoma
Cutaneous SCC
Melanoma (UP)
Gastric Carcinoma
Uterine Endometroid
ICC
Fusion
Mutated
Amp
Homdel
Multiple
0
5
10
15
Alteration Frequency (%)
AACR
PROJECT GENIE

Figure 2b. FGFR2 Resistance Mutations in Cholangiocarcinoma[9]

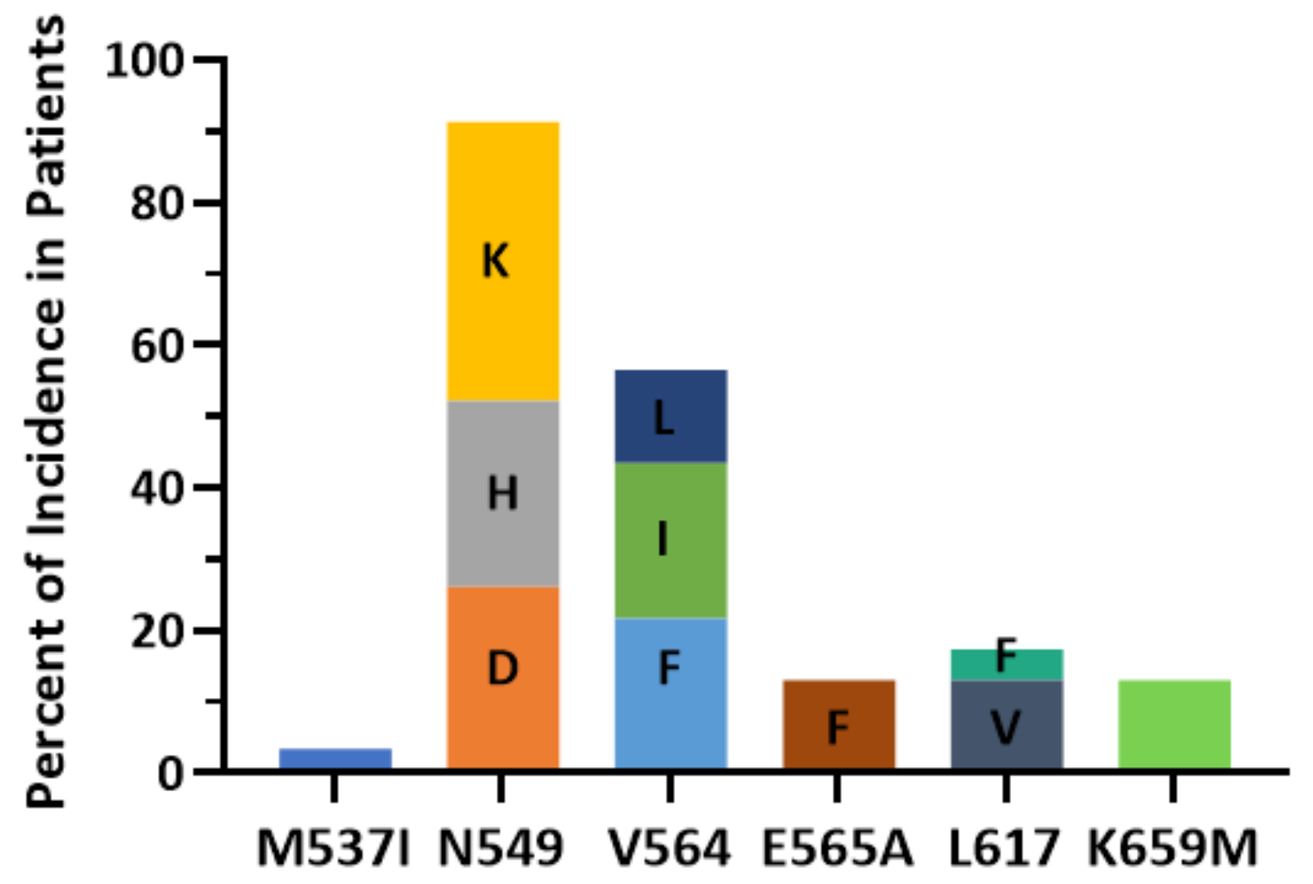
Percent of Incidence in Patients
100
80
60
40
20
0
K
H
D
L
I
F
F
F
V
M537I
N549
V564
E565A
L617
K659M

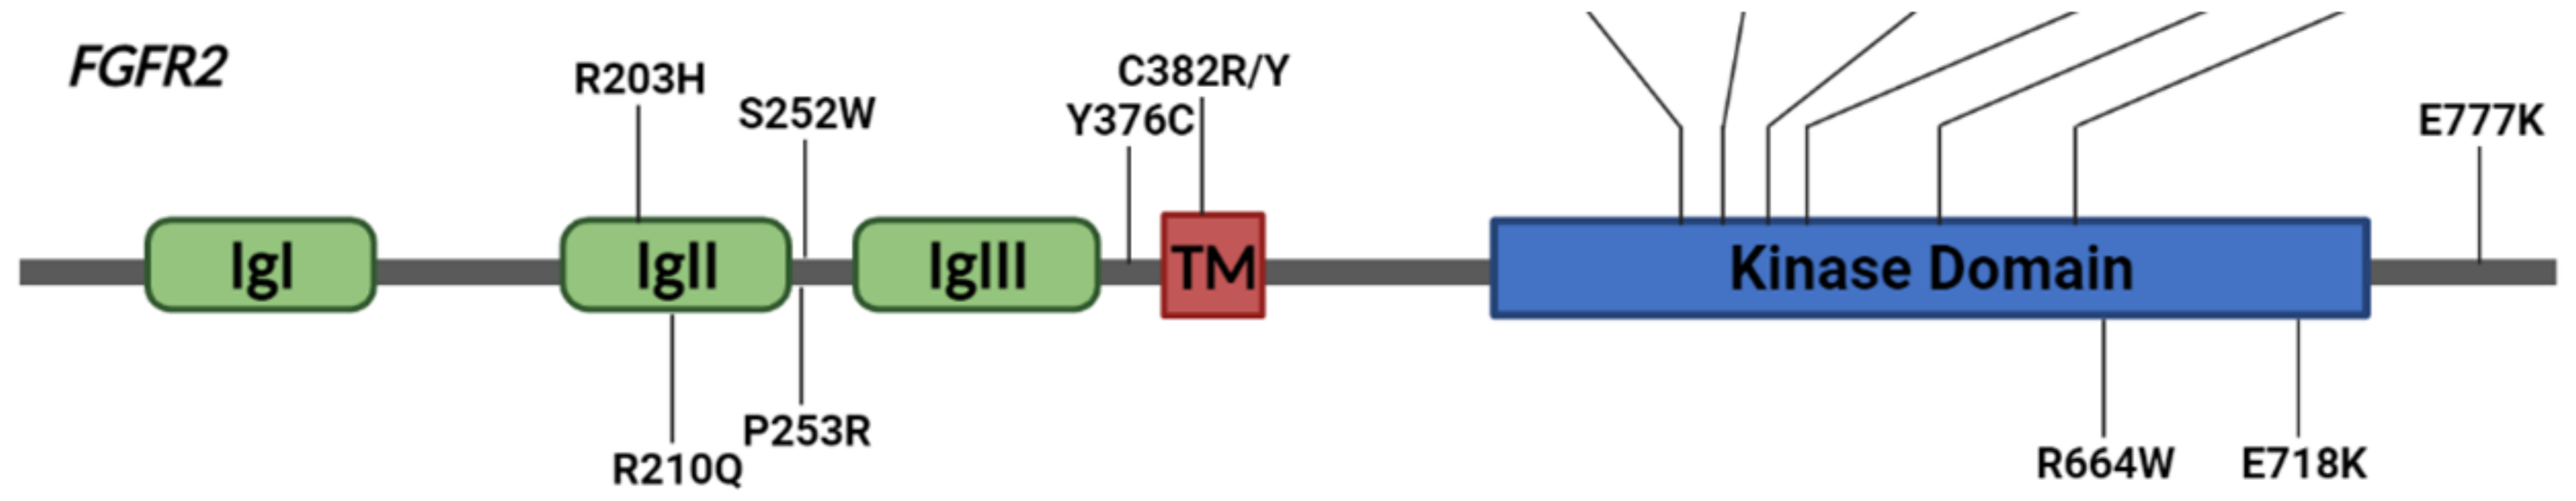
FGFR2
R203H
S252W
C382R/Y
Y376C
E777K
IgI
IgII
IgIII
TM
Kinase Domain
R210Q
P253R
R664W
E718K

Results

Figure 3. Majority of Lead Series Compounds are >30-Fold Selective for FGFR2 vs. FGFR1

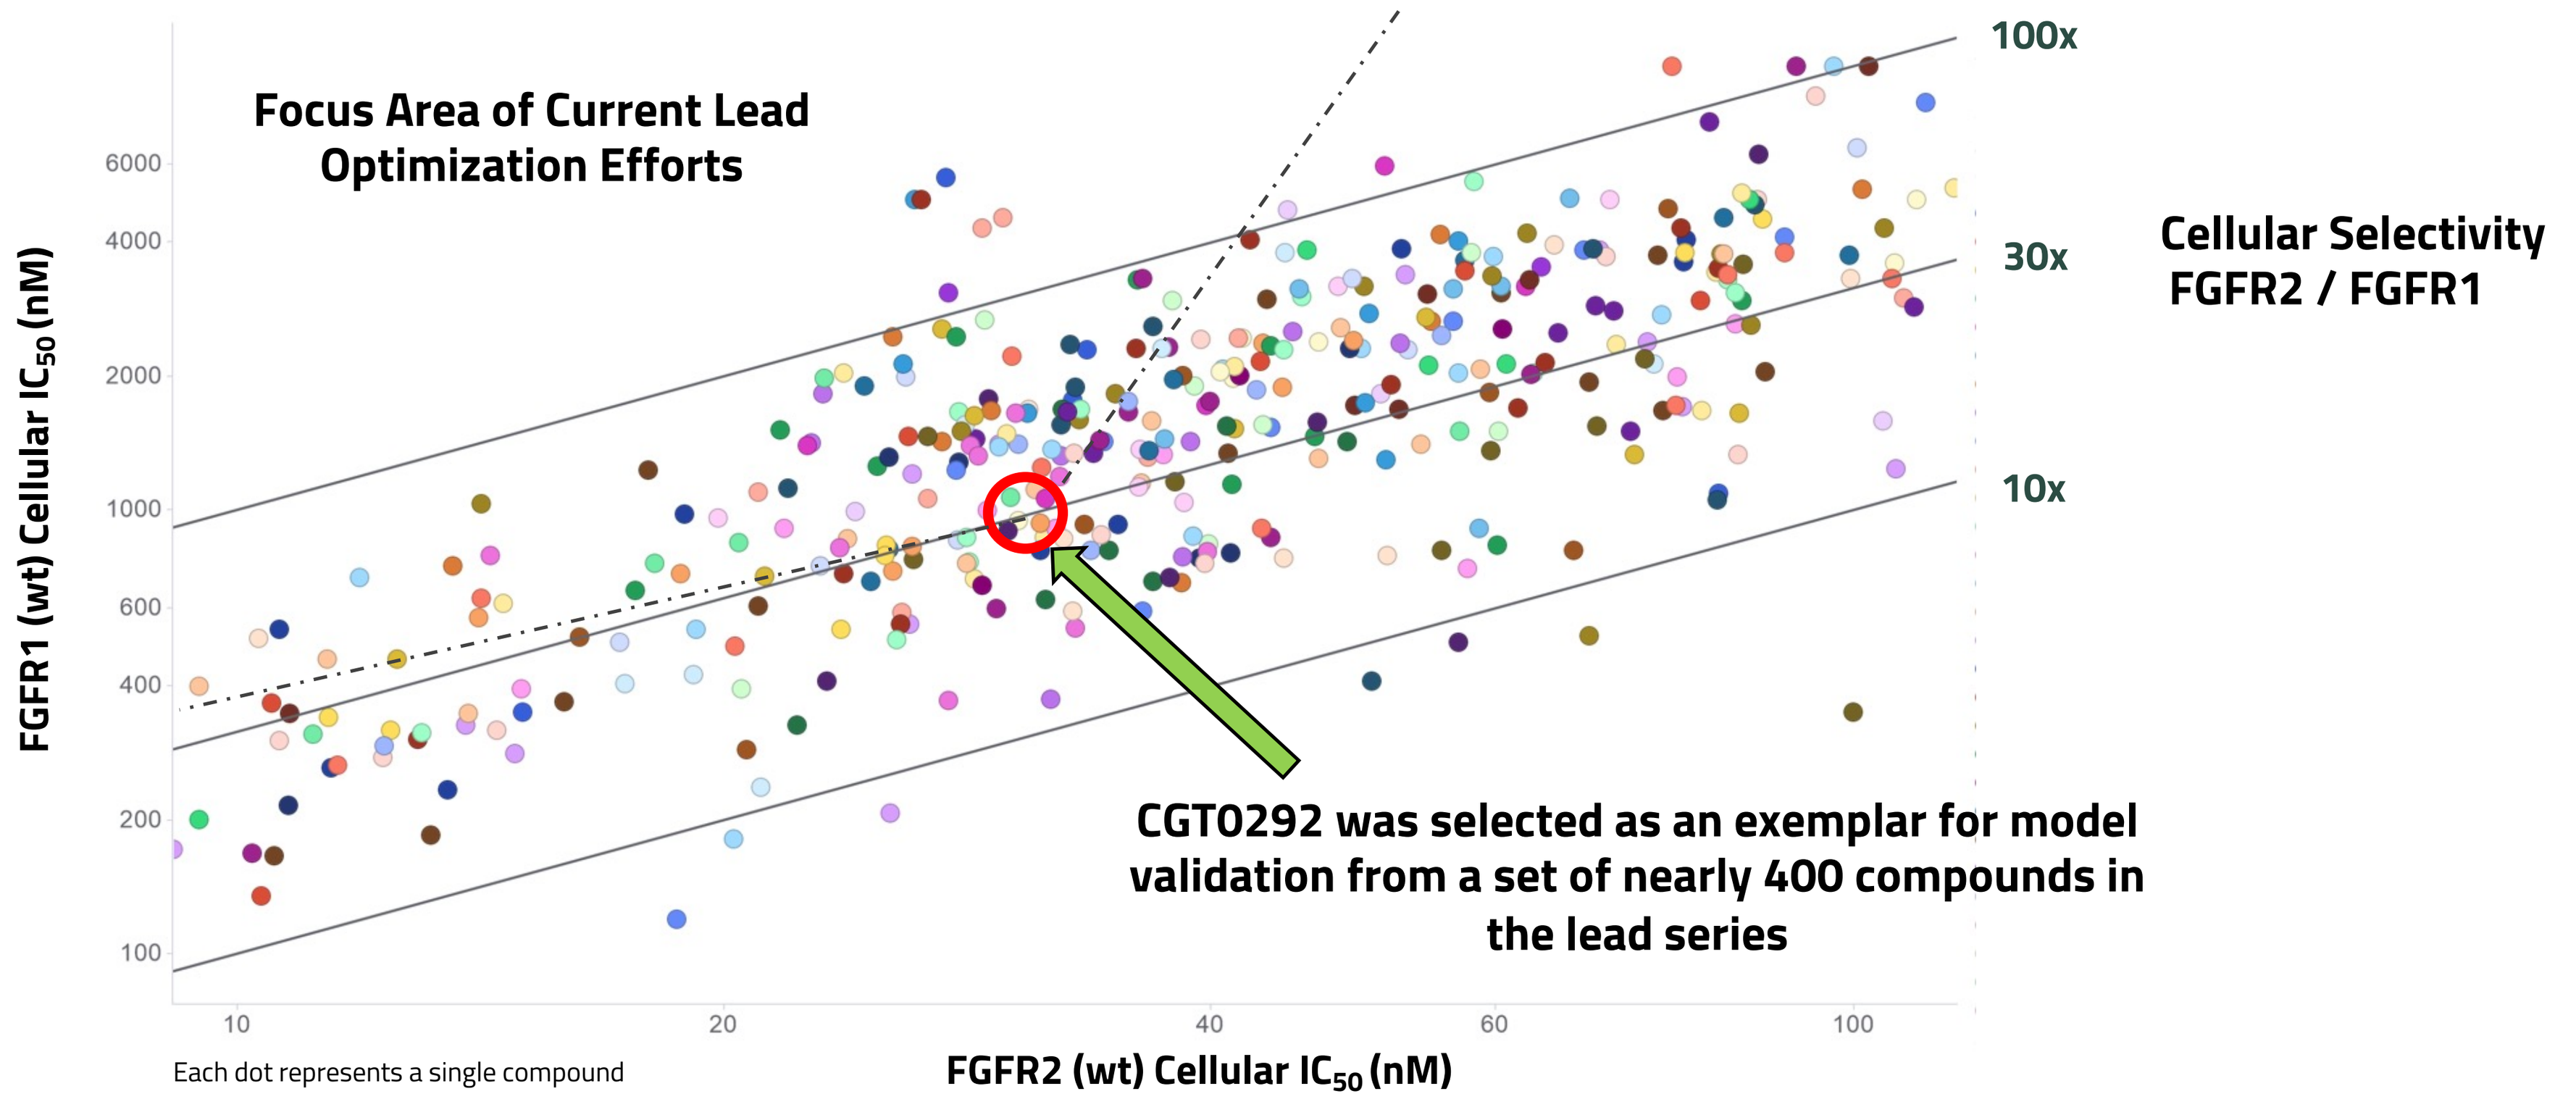
Focus Area of Current Lead Optimization Efforts
100x
30x
10x
Cellular Selectivity FGFR2 / FGFR1
FGFR1 (wt) Cellular IC50 (nM)
FGFR2 (wt) Cellular IC50 (nM)
CGT0292 was selected as an exemplar for model validation from a set of nearly 400 compounds in the lead series

Each dot represents a single compound

Table 2. Drug-Like In Vitro Properties

Assay	CGT0292
tPSA / clogP / MW	~80 / ~2.0 / <400
Solubility (pH 1.2 / 6.5 / 7.4)	3250 / 454 / 335
Permeability A-B Efflux Ratio	16 (10^{-6} cm/sec) 9
Microsomal CL (% ER) M / R / H	20 / 13 / 11
Hepatocyte CL (% ER) M / R / H	61 / 14 / 21
Protein Binding (%) M / R / H	70.6 / 72.9 / 64.6
Reversibility Assay Enzyme IC_{50} with Preincubation	0 min = 2.1 nM 30 min = 2.1 nM 60 min = 2.2 nM

CGT0292:

- Has drug-like calculated physiochemical properties
- Exhibits BCS Class 1 properties with high thermodynamic solubility and high MDR1-MDCKII permeability
- Shows low CL across species in microsomes and hepatocytes
- Displays low plasma protein binding across species tested: favorable free fraction-adjusted target coverage observed
- Is a reversible inhibitor, IC_{50} does not change with preincubation
 - Avoids potential for time-dependent inhibition of FGFR1
 - Potential proteome selectivity advantage
 - Avoids non-traditional glutathione metabolism
- Provides a robust scaffold for further compound optimization

Table 3. CGT0292 Demonstrates Progress Toward Superior FGFR1 Selectivity and Mutant Target Coverage Compared to Late Stage / Approved Pan-FGFRi

Compound	Cell IC_{50} (nM)		Enzyme IC_{50} (nM)						
	FGFR1-WT	FGFR2-WT	FGFR1-WT	FGFR2-WT	FGFR2 N549H	FGFR2 V564I	FGFR2 V564F	FGFR3-WT	FGFR4-WT
CGT0292	949	30	27	2.1	0.93	5.6	3.0	5.8	213
Infigratinib	10.8	4.2	0.37	0.45	0.93	63	879	4.6	65
Pemigatinib	10.1	1.6	0.49	0.18	1.7	5.2	244	1.1	13
Erdafitinib	5.3	1.4	0.24	0.11	0.32	0.16	173	0.14	1.0
Futibatinib	4.2	2.1	0.55	0.20	0.59	0.70	54	0.32	1.0

- CGT0292 displays >30X selectivity between FGFR1 and FGFR2 in cellular assays
- Is potent against all tested FGFR2 mutations, enzyme IC_{50} < 10 nM
- Exhibits > 100x selectivity for FGFR2 over FGFR4 in the enzyme assay

Figure 4. CGT0292 Shows Promising In Vivo Performance and Complete Suppression of pFGFR

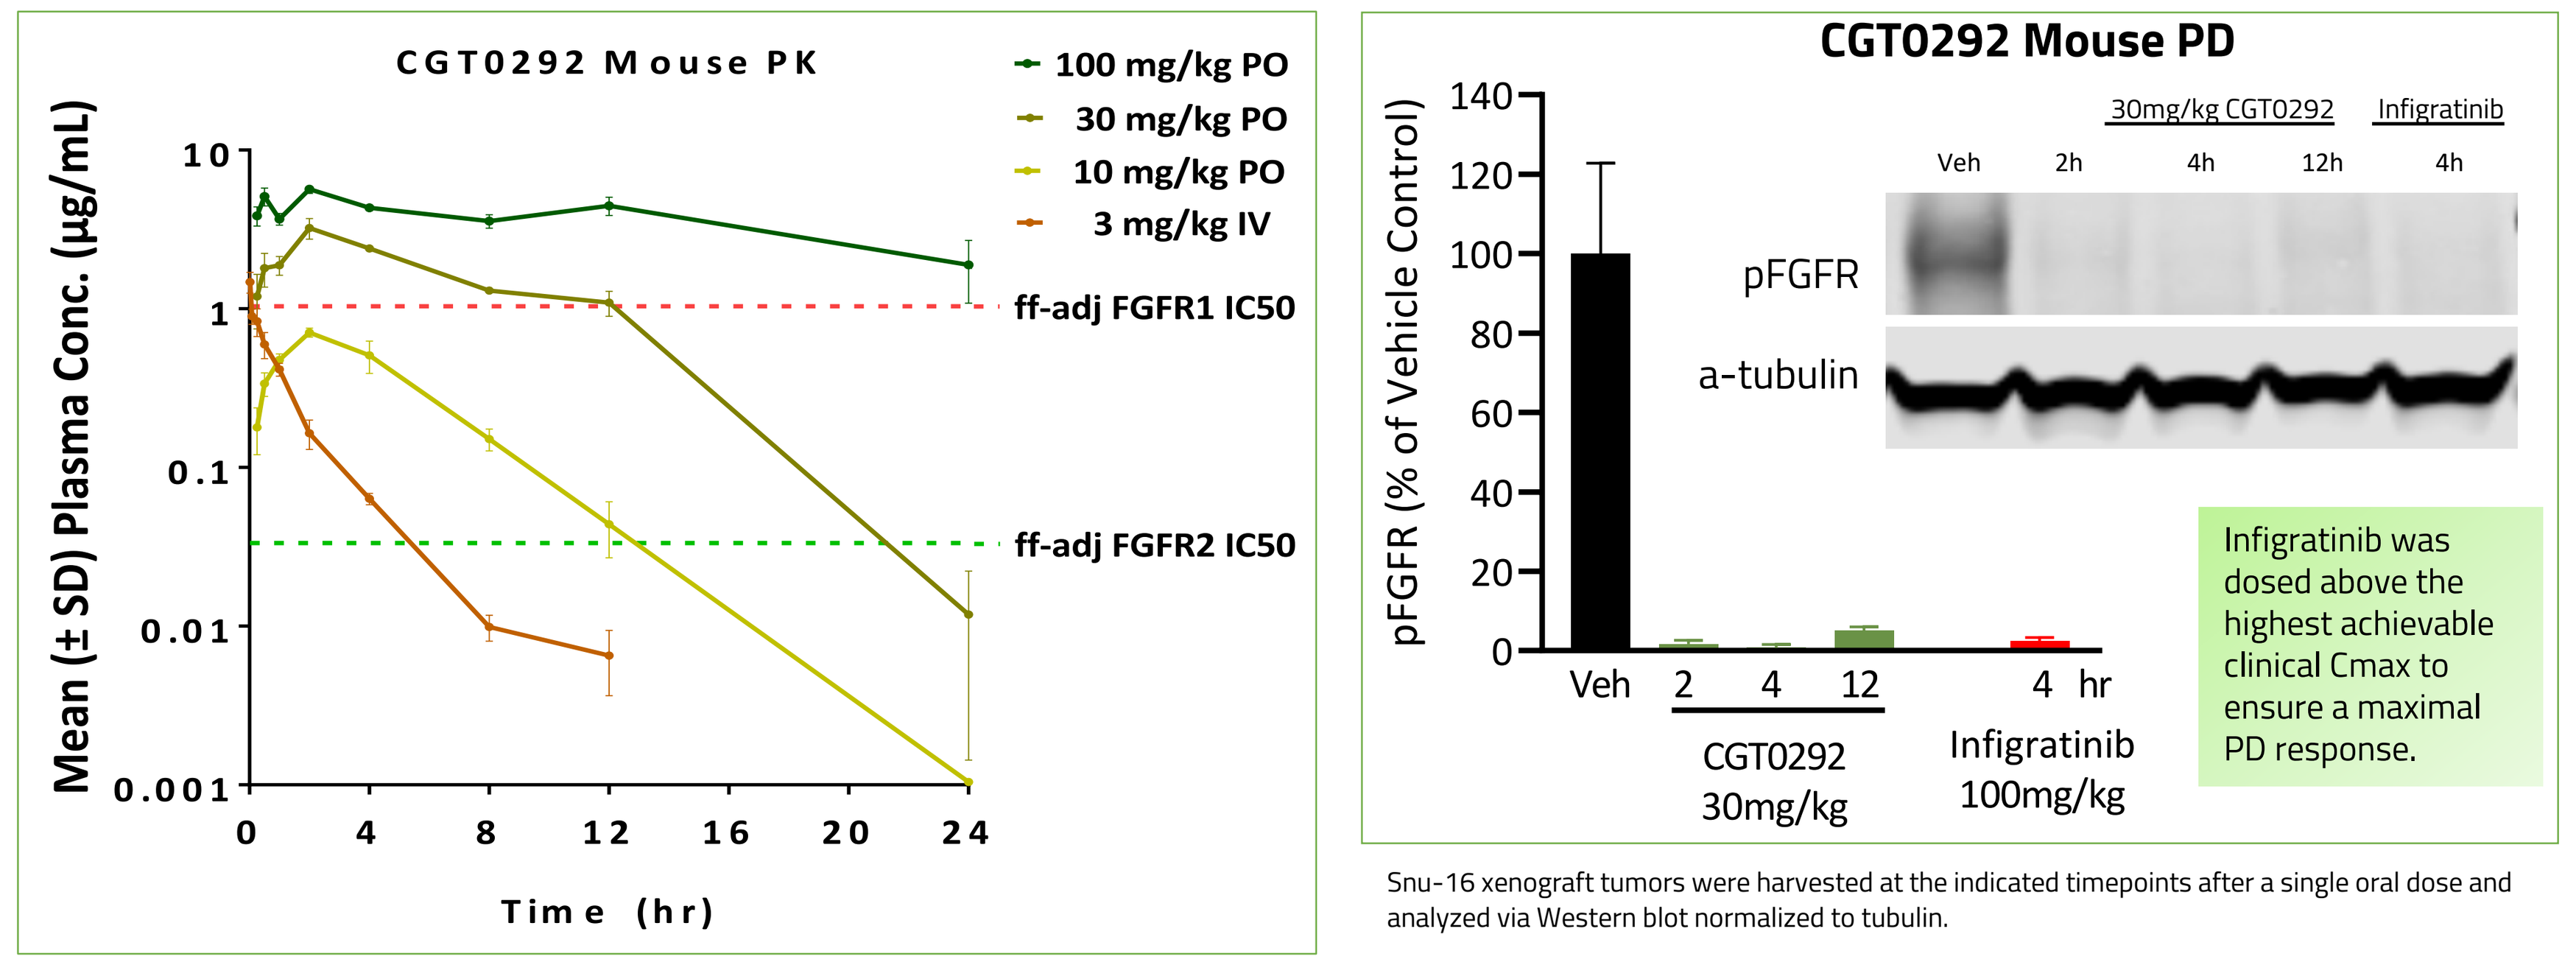
CGT0292 Mouse PK
100 mg/kg PO
30 mg/kg PO
10 mg/kg PO
3 mg/kg IV
ff-adj FGFR1 IC50
ff-adj FGFR2 IC50
Mean (± SD) Plasma Conc. (µg/mL)
Time (hr)
CGT0292 Mouse PD
pFGFR (% of Vehicle Control)
30mg/kg CGT0292
Infigratinib
Veh
2h
4h
12h
4h
pFGFR
a-tubulin
Veh
2
4
12
4 hr
CGT0292 30mg/kg
Infigratinib 100mg/kg
Infigratinib was dosed above the highest achievable clinical Cmax to ensure a maximal PD response.

Snu-16 xenograft tumors were harvested at the indicated timepoints after a single oral dose and analyzed via Western blot normalized to tubulin.

- CGT0292 showed increased exposure with increased dose in mice
- IV CL: 35.9 mL/min/kg, F: 87.9% (10 mg/kg PO dose)
- Comparable to Infigratinib, CGT0292 showed complete inhibition of pFGFR2 in the SNU-16 PK/PD model

Figure 5. CGT0292 Does Not Show Serum Phosphorus Increase at a 5 mg/kg Dose in the SD Rat Model of Hyperphosphatemia

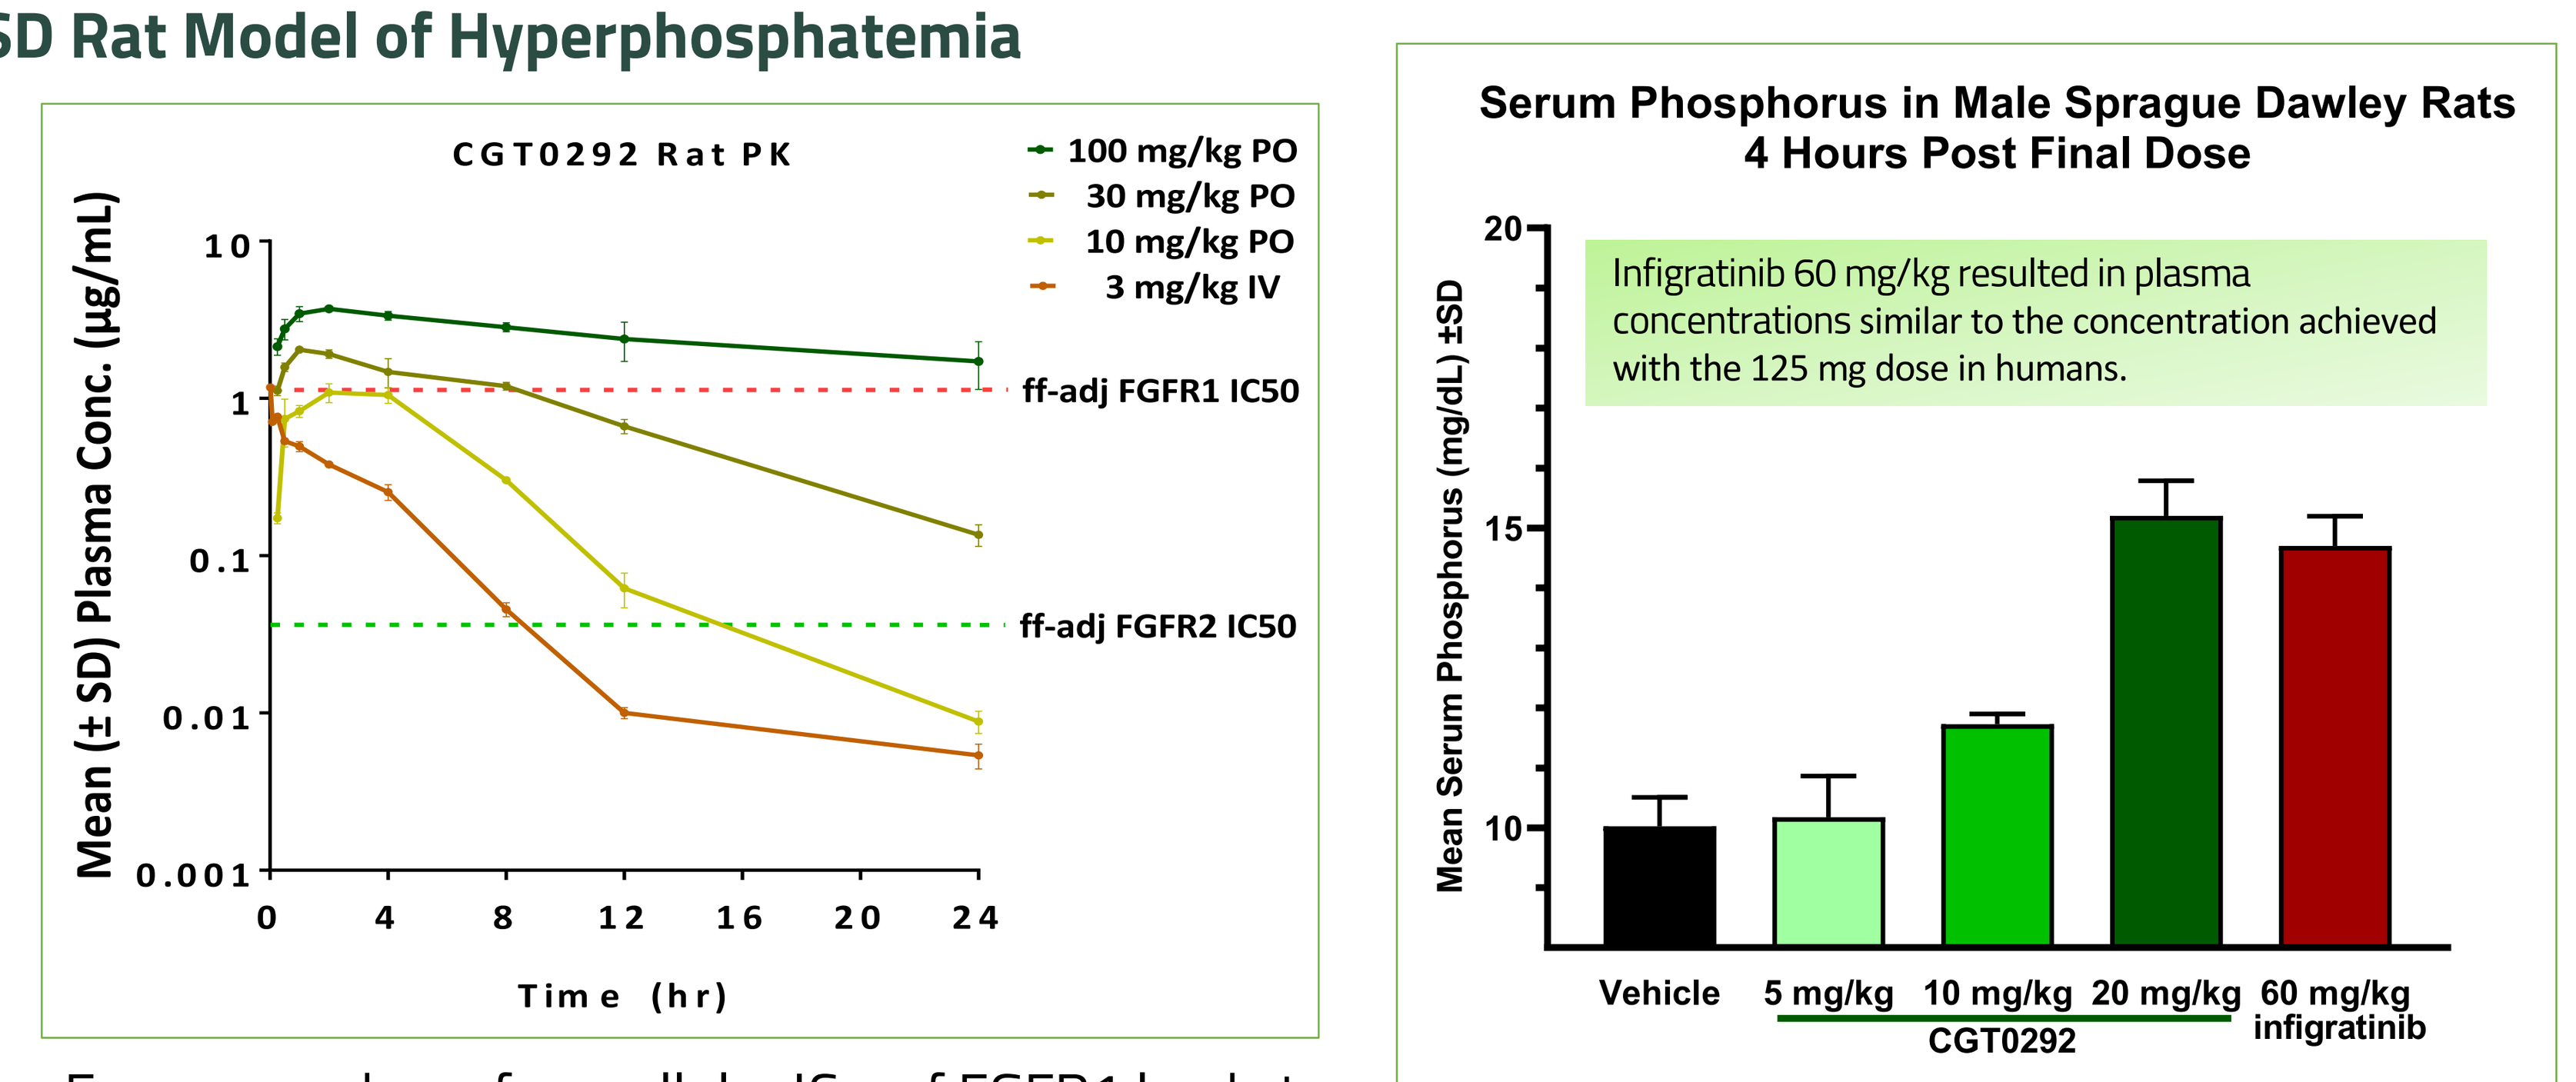
CGT0292 Rat PK
100 mg/kg PO
30 mg/kg PO
10 mg/kg PO
3 mg/kg IV
ff-adj FGFR1 IC50
ff-adj FGFR2 IC50
Mean (± SD) Plasma Conc. (µg/mL)
Time (hr)
Serum Phosphorus in Male Sprague Dawley Rats 4 Hours Post Final Dose
Mean Serum Phosphorus (mg/dL) ±SD
Infigratinib 60 mg/kg resulted in plasma concentrations similar to the concentration achieved with the 125 mg dose in humans.
Vehicle
5 mg/kg
10 mg/kg
20 mg/kg
CGT0292
60 mg/kg infigratinib

Male Sprague Dawley rats (n= 4/group) were dosed with 5, 10, or 20mg/kg CGT0292 (PO, BID) or 60mg/kg infigratinib (PO, QD) for a total of 3 days followed by serum collections 4 hours after the last dose for serum phosphorus levels.

- Exposures above free cellular IC_{50} of FGFR1 leads to increase in serum phosphorus preclinically

Conclusions

- FGFR1 mediated hyperphosphatemia is the most common dose limiting toxicity with pan-FGFR inhibitors
- Approved inhibitors fail to capture the full landscape of FGFR2 and FGFR3 altered tumor types
- PK/PD and hyperphosphatemia models have been established to interrogate and enlarge the therapeutic window during further optimization of CGT0292 and related compounds in the series

•CGT0292:

- Provides >30X selectivity window for FGFR2 vs. FGFR1 in wt p-FGFR cell assay
- Shows potency against a panel of FGFR2 gatekeeper and molecular brake mutations
- Has drug-like physiochemical properties with desirable solubility, permeability and plasma protein binding
- Displays dose ascending PK with low to moderate IV clearance and high oral bioavailability in mice
- Has equivalent inhibition of pFGFR2 compared to Infigratinib in the SNU-16 tumor PK/PD model
- Exhibits dose related increases in serum phosphorus levels

References : 1. AACR Project GENIE Consortium. *Cancer Discov.* 2017;7(8):818-831; 2. Helsten T, et al. *Clin Cancer Res.* 2016;22(1):259-267; 3. Abou-Alfa GK, et al. *Lancet Oncol.* 2020;21(5):671-684. 4. Siefker-Radtke et al. *Lancet Oncol.* 2022;23(2):248-258; 5. PEMAZYRE (pemigatinib; prescribing information). Wilmington, DE: Inctye Corporation; 2021; 6. Javle M, et al. *J Clin Oncol.* 2018;36(3):276-282. *2018*; 7. TRUSELTIQ (infigratinib; prescribing information). Brisbane, CA: QED Therapeutics: 2021; 8. BALVERSA® (erdafitinib, prescribing information). Horsham, PA: Janssen Products; 2020; 9. Goyal L, et al. *Cancer Discov.* 2017; 7(3): 252–263

*All animal studies were conducted in accordance with The Guide for the Care & Use of Laboratory Animals and IACUC approved policies and procedures at the facilities where they were performed.

Scan the QR code to obtain a PDF copy of this poster

AACR Annual Meeting 2022| Sunday, April 10th 2022 | New Orleans, LA